138

Exposure-Response Analyses of Efficacy and Safety of Trastuzumab Deruxtecan to Inform Dosing Recommendations in *HER2*-Mutant Non–Small Cell Lung Cancer

Amit Khatri,[1] Daniel Polhamus,[2] Ramon Garcia,[2] Todd Yoder,[2] Malaz Abutarif,[1] Tushar Garimella[1]

[1]Quantitative Clinical Pharmacology, Daiichi Sankyo, Inc, Basking Ridge, NJ, USA
[2]Metrum Research Group, Tariffville, CT, USA

Objectives

- To evaluate the relationship between trastuzumab deruxtecan (T-DXd) pharmacokinetic (PK) exposure modeling and efficacy endpoints in patients with human epidermal growth factor receptor 2 (*HER2*)-mutant non–small cell lung cancer (NSCLC) and safety endpoints across other tumor types and *HER2*-mutant NSCLC

Conclusions

- The exposure-response (E-R) analyses for efficacy and safety endpoints supported the dosing recommendation of T-DXd 5.4 mg/kg in patients with *HER2*-mutant NSCLC
 - No clinically meaningful difference in efficacy (objective response rate [ORR]) was estimated between T-DXd 5.4 and 6.4 mg/kg
 - The incidence of safety events was lower with T-DXd 5.4 mg/kg compared with T-DXd 6.4 mg/kg
- These findings were confirmed based on more mature data from the DESTINY-Lung02 primary analysis and support the utility of E-R analyses in dosing recommendations

Supplementary content available
Copies of materials obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ACCP and the author of this poster.

This information is the intellectual property of the authors/presenters.
Please contact Amit Khatri at amit.khatri@daiichisankyo.com for permission to reprint and/or redistribute.

Presented at the 2024 American College of Clinical Pharmacology (ACCP) Annual Meeting; September 8-10, 2024; Bethesda, Maryland

Background

- T-DXd is an antibody-drug conjugate made up of 3 components: a humanized anti-HER2 immunoglobulin G1 monoclonal antibody, a topoisomerase I inhibitor payload, and a tetrapeptide-based cleavable linker[1,2]
 - Based on phase 2 or 3 trials, T-DXd is approved for the treatment of adult patients with unresectable or metastatic HER2-positive or HER2-low breast cancer (BC), *HER2*-mutant NSCLC, HER2-positive gastric cancer (GC) or gastroesophageal junction (GEJ) adenocarcinoma, and HER2-positive (immunohistochemistry 3+) solid tumors with no other treatment options[3]
- Previously, E-R analyses for key efficacy and safety endpoints supported the recommended dose of T-DXd 5.4 mg/kg once every 3 weeks (Q3W) in patients with HER2-positive BC,[4,5] and an additional update to these analyses supported T-DXd 6.4 mg/kg Q3W in patients with HER2-positive GC or GEJ adenocarcinoma[6]
- The phase 2 DESTINY-Lung01 (T-DXd 6.4 mg/kg Q3W) and DESTINY-Lung02 (T-DXd 5.4 and 6.4 mg/kg Q3W) trials were conducted to investigate T-DXd in patients with *HER2*-mutant NSCLC[7,8]; based on results from these studies, T-DXd 5.4 mg/kg was approved for the treatment of patients with *HER2*-mutant NSCLC
- In the present study we conducted E-R analyses of key efficacy and safety endpoints using data from patients with *HER2*-mutant NSCLC to support the dosing recommendation of T-DXd in this patient population

Methods

Exposure-Efficacy Analyses

- Exposure-efficacy analyses for ORR included 181 patients with *HER2*-mutant NSCLC who received T-DXd 5.4 mg/kg Q3W or 6.4 mg/kg Q3W in DESTINY-Lung01 (NCT03505710) and the DESTINY-Lung02 (NCT04644237) interim analysis, as well as a small cohort of patients from a phase 1 trial (NCT02564900); the E-R model for ORR was later updated to include DESTINY-Lung02 primary analysis data, resulting in a total of 253 patients (**Figure 1**)

Figure 1. Exposure-Efficacy Analysis Plan

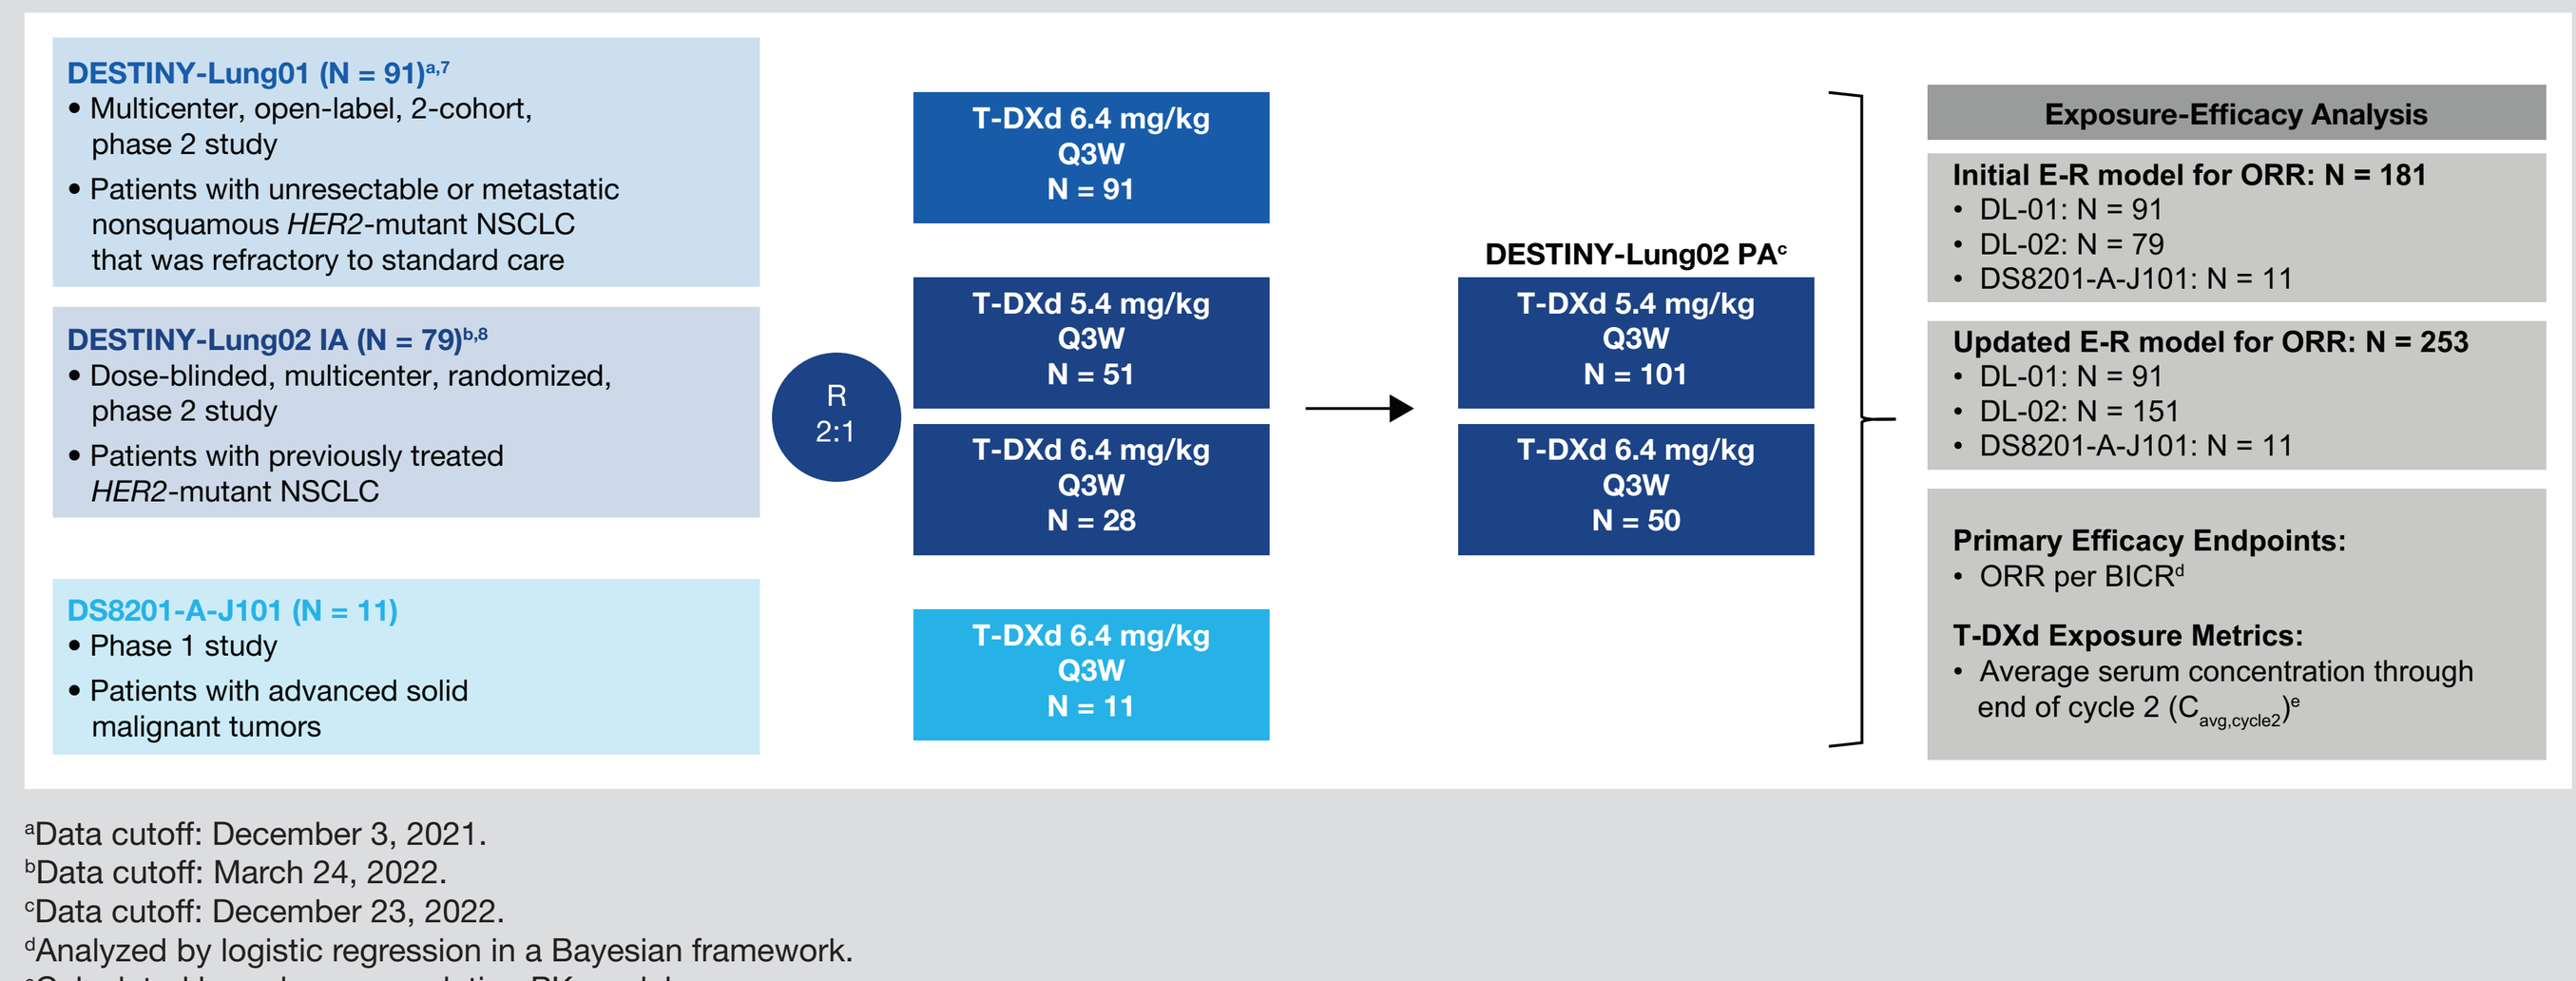

DESTINY-Lung01 (N = 91)a,7
• Multicenter, open-label, 2-cohort, phase 2 study
• Patients with unresectable or metastatic nonsquamous HER2-mutant NSCLC that was refractory to standard care
DESTINY-Lung02 IA (N = 79)b,8
• Dose-blinded, multicenter, randomized, phase 2 study
• Patients with previously treated HER2-mutant NSCLC
DS8201-A-J101 (N = 11)
• Phase 1 study
• Patients with advanced solid malignant tumors
T-DXd 6.4 mg/kg Q3W N = 91
R 2:1
T-DXd 5.4 mg/kg Q3W N = 51
T-DXd 6.4 mg/kg Q3W N = 28
T-DXd 6.4 mg/kg Q3W N = 11
DESTINY-Lung02 PAc
T-DXd 5.4 mg/kg Q3W N = 101
T-DXd 6.4 mg/kg Q3W N = 50
Exposure-Efficacy Analysis
Initial E-R model for ORR: N = 181
• DL-01: N = 91
• DL-02: N = 79
• DS8201-A-J101: N = 11
Updated E-R model for ORR: N = 253
• DL-01: N = 91
• DL-02: N = 151
• DS8201-A-J101: N = 11
Primary Efficacy Endpoints:
• ORR per BICRd
T-DXd Exposure Metrics:
• Average serum concentration through end of cycle 2 (Cavg,cycle2)e

[a]Data cutoff: December 3, 2021.
[b]Data cutoff: March 24, 2022.
[c]Data cutoff: December 23, 2022.
[d]Analyzed by logistic regression in a Bayesian framework.
[e]Calculated based on a population PK model.

- Using only exposure as a predictor, 4 E-R relationships were considered: linear, log-linear, maximum effect (E_{max}), and sigmoidal E_{max}; the best fitting model among these 4 relationships was used in subsequent model development
- Further analyses were conducted to identify baseline characteristics and demographics that were significant in the E-R relationships (**Supplementary Methods**)

Exposure-Safety Analyses

- Exposure-safety analyses were conducted using an integrated dataset across 11 phase 1 to 3 clinical trials (N = 1822), including data from patients with *HER2*-mutant NSCLC and other tumor types (HER2-overexpessing NSCLC, BC, GC or GEJ adenocarcinoma, others)
- Safety endpoints evaluated included dose reduction due to adverse events (AEs), grade ≥3 AEs, serious AEs, grade ≥3 anemia, grade ≥3 neutropenia, grade ≥3 thrombocytopenia, and any-grade and grade ≥3 adjudicated drug-related interstitial lung disease (ILD)
- For all safety endpoints, 4 E-R relationship models were considered, similar to ORR
- T-DXd exposure metrics, including maximum serum concentration and area under the serum concentration-time curve (AUC) at steady-state (AUC_{ss}), as well as released payload (DXd) average serum concentration to the time of safety event ($C_{avg\text{-}TOE}$) identified as significant in previous exposure-safety analyses, were tested for correlation with safety endpoints and were calculated based on a population PK model
 - Please see the population PK model poster, also being presented at the annual meeting of the American College of Clinical Pharmacology, for further information (poster 075)
- E-R relationships for safety endpoints assessed patient-specific covariates such as demographics and disease and patient characteristics, as were included in efficacy analyses, and baseline levels of platelets (for thrombocytopenia), hemoglobin (for anemia), neutrophils (for neutropenia), and oxygen saturation

Results

Exposure-Efficacy Analyses

- 177 patients (n = 51 for T-DXd 5.4 mg/kg Q3W and n = 126 for T-DXd 6.4 mg/kg Q3W) with *HER2*-mutant NSCLC with available PK exposure data were included in the initial exposure-efficacy analysis
 - Most patients were female (66.7%) and Asian (52.5%) or White (30.5%), with a median age of 60 years (range, 23 to 88 years) and a median body weight of 62 kg (range, 39.7 to 111 kg)
- $C_{avg,cycle\,2}$ was highly correlated (correlations >0.89) with exposure metrics such as AUC in cycle 1 or T-DXd C_{avg} up to time of ORR
 - Approximately 50% of patients had an objective response rate assessment by the end of cycle 2; because $C_{avg,cycle\,2}$ takes into account any dose reductions in cycle 2 (unlike cycle 1 or AUC_{ss}), it was used for the E-R analysis of ORR
- T-DXd exposure had a positive but shallow relationship with ORR (**Figure 2**)
 - For a typical patient at the 5th and 95th percentiles of $C_{avg,cycle\,2}$, predicted ORR varied from 57% (95% credible interval [CrI], 36%-77%) to 65% (95% CrI, 41%-84%)

Figure 2. Estimated E-R Relationship for ORR in Patients With *HER2*-Mutant NSCLC

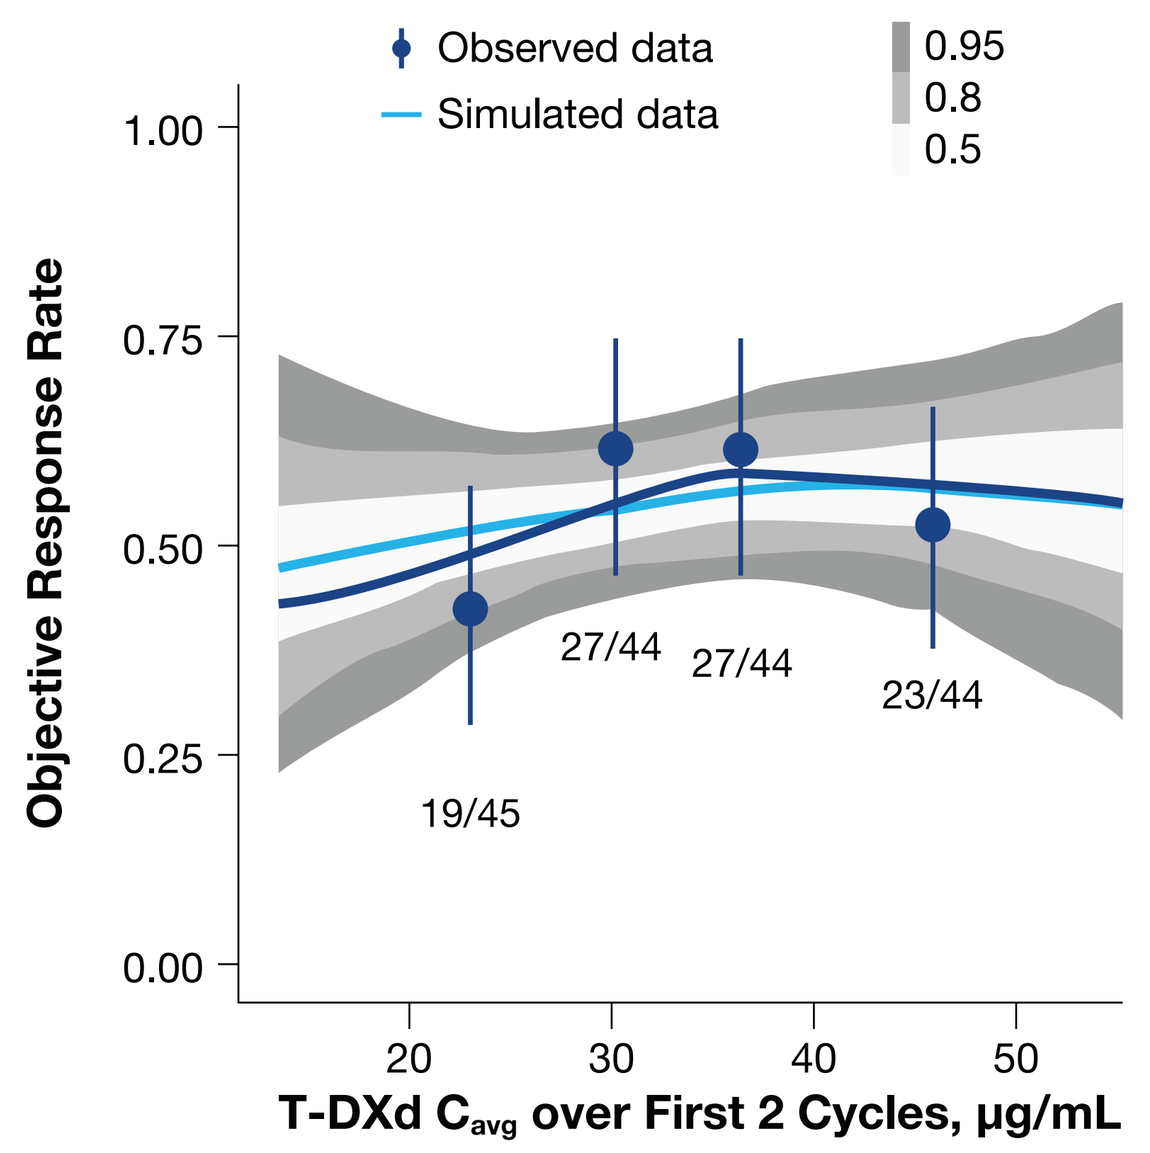

Observed data
Simulated data
0.95
0.8
0.5
Objective Response Rate
1.00
0.75
0.50
0.25
0.00
19/45
27/44
27/44
23/44
20
30
40
50
T-DXd Cavg over First 2 Cycles, µg/mL

The solid dark blue line represents a smooth (univariate generalized additive model) of the original data. The solid light blue line (simulation median) and shaded regions (50%, 80%, and 95% prediction intervals) represent the distribution of smooths fit to replicate data sets simulated using the estimated model. Points and vertical bars represent observed proportion and 95% CIs at quartiles of covariate. Numbers represent responders and total patients within each quartile.

- For a typical patient, predicted ORR varied from 43% (95% CrI, 28%-58%) to 61% (95% CrI, 46%-75%) to 69% (95% CrI, 49%-85%) for non-Asians, Asians from Japan, and Asians not from Japan, respectively
- For a typical patient at the 5th and 95th percentiles of baseline target tumor size, predicted ORR varied from 70% (95% CrI, 50%-84%) to 55% (95% CrI, 35%-74%)
- No clinically meaningful difference in ORR was observed across the T-DXd 5.4-mg/kg Q3W and T-DXd 6.4-mg/kg Q3W dosing regimens in the initial (ORR, 53% [95% CrI, 44%-60%] vs 55% [95% CrI, 47%-62%]) or updated E-R analyses (ORR, 50% [95% CrI, 44%-57%] vs 56% [95% CrI, 50%-62%])
 - These simulation results were consistent with those observed with T-DXd 5.4 and 6.4 mg/kg in the DESTINY-Lung02 primary analysis (data cutoff: December 23, 2022)[8]

Exposure-Safety Analyses

- The exposure-safety dataset consisted of 1822 patients across 11 clinical trials in which T-DXd doses ranged from 0.8 to 8 mg/kg
 - 1425 (78.2%) female and 397 (21.8%) male patients were included
 - 621 (34.1%) patients were Asians from Japan, 377 (20.7%) were Asians from countries other than Japan, and 824 (45.2%) were non-Asians
 - Most patients had BC (61.9%), GC (16.1%), or NSCLC (19.0%; *HER2*-mutant NSCLC, 13.5%, and HER2-overexpressing NSCLC, 5.5%)
- Exposure-safety analyses showed statistically significant relationships between T-DXd exposure and ILD, and between DXd exposure (average concentration to the event time) and all other safety endpoints (**Figures S1 and S2**)
- Covariate effects for the safety endpoints included in the analyses are shown in **Figure 3**

Figure 3. Forest Plots Showing Covariate Effects for (A) Dose Reduction Due to AE, (B) Grade ≥3 AE, (C) SAE, (D) Grade ≥3 Anemia, (E) Grade ≥3 Neutropenia, (F) Grade ≥3 Thrombocytopenia, (G) Any-Grade ILD, and (H) Grade ≥3 ILD Events

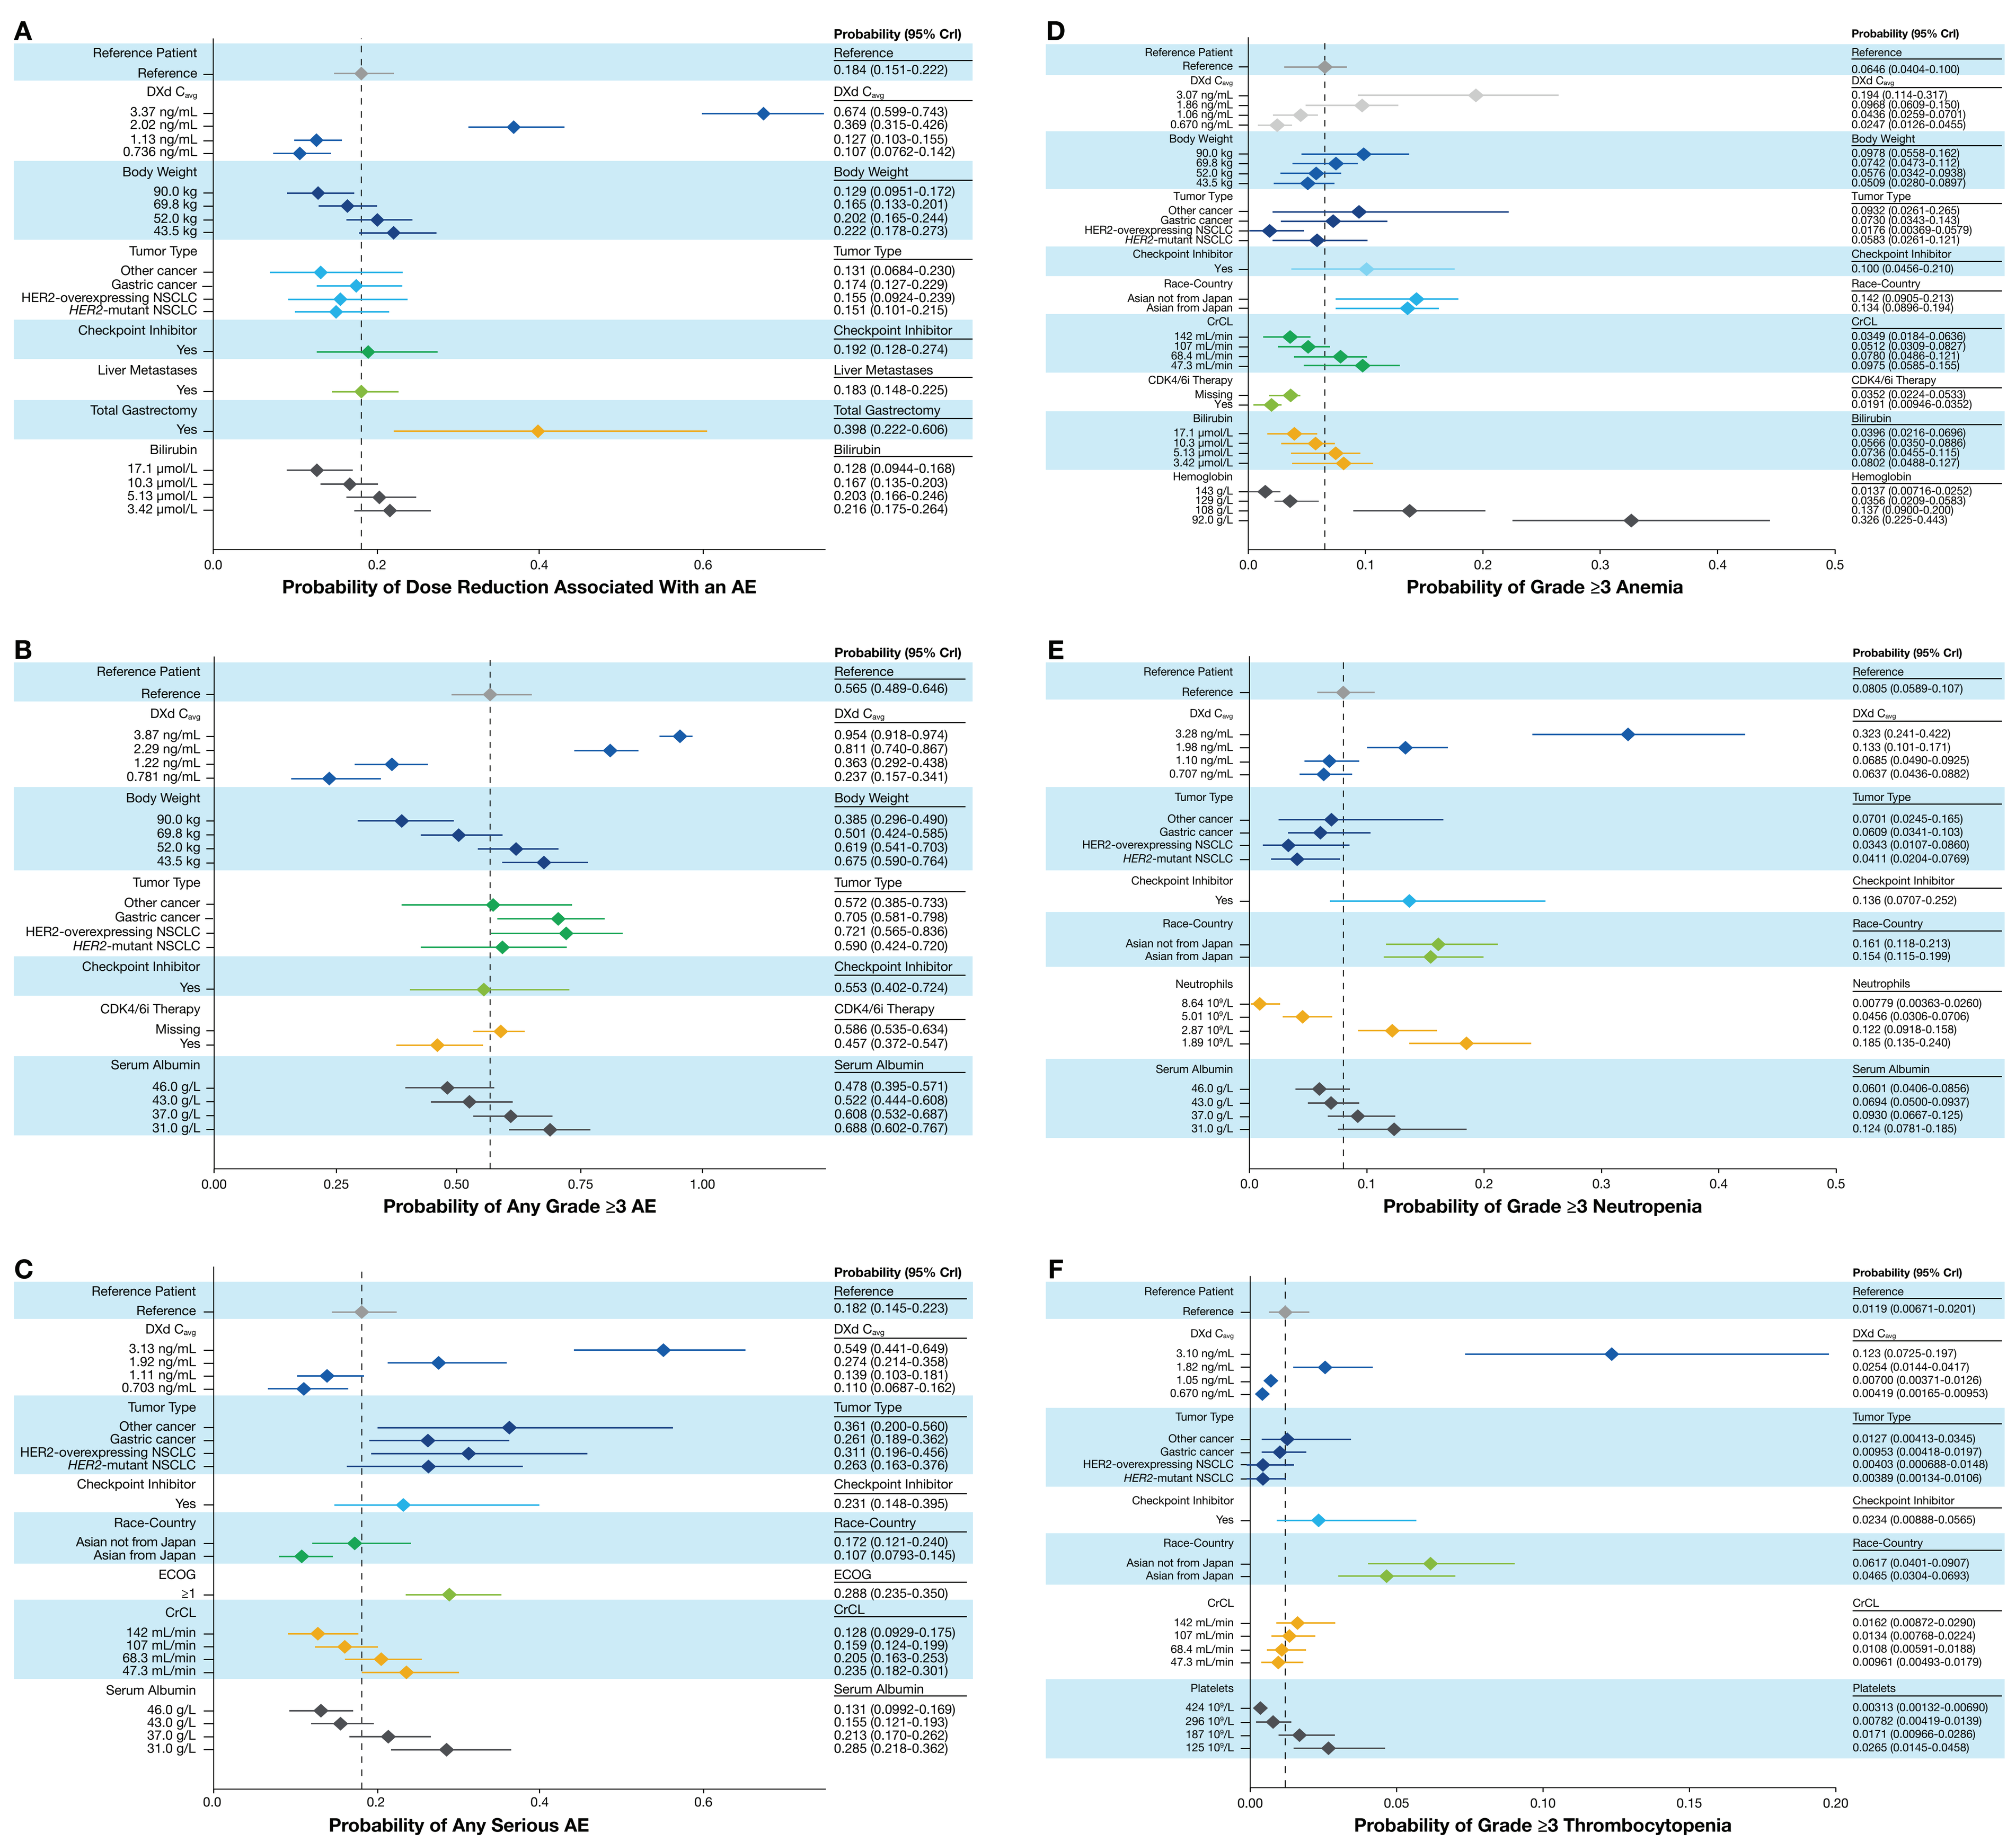

A
Probability of Dose Reduction Associated With an AE
B
Probability of Any Grade ≥3 AE
C
Probability of Any Serious AE
D
Probability of Grade ≥3 Anemia
E
Probability of Grade ≥3 Neutropenia
F
Probability of Grade ≥3 Thrombocytopenia

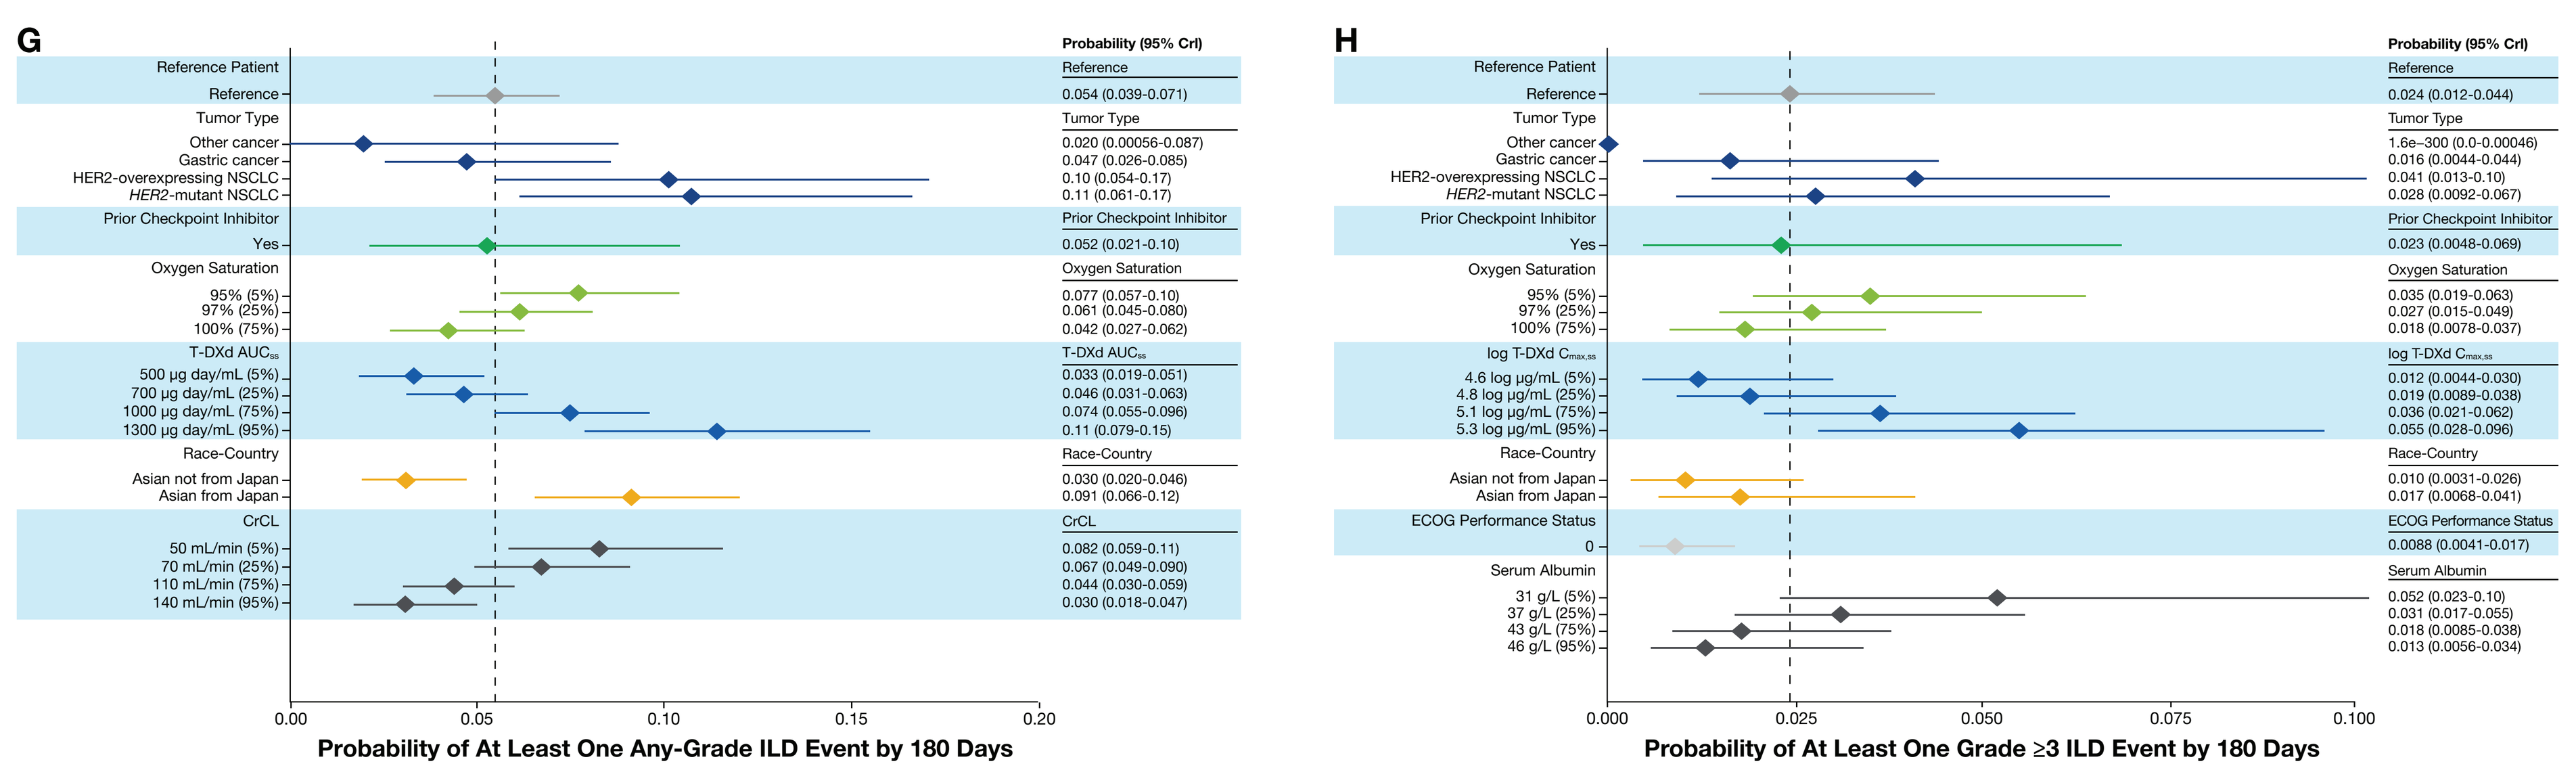

G
Probability of At Least One Any-Grade ILD Event by 180 Days
H
Probability of At Least One Grade ≥3 ILD Event by 180 Days

Covariate effects were expressed as predicted responses comparing a "reference individual" (all covariates set to reference values) and a "perturbed reference individual" (differing from the reference individual only for the covariate of interest). Responses were summarized in terms of the posterior median and 95% CrI, based on 1000 posterior samples.

Median and 95% CrI are shown to the right of each plot. Perturbations in calculations of typical value predictions were considered for all categorical covariate levels and at the approximate 5th, 25th, 75th, and 95th percentiles of exposure and continuous covariate values, with the approximate 50th percentile and mode of the categorical covariates representing the reference subject. ILD endpoints were analyzed by accelerated failure time survival models, of which the residual distribution models tested were exponential, Weibull, or log-normal TTE parametric distribution; all other endpoints were analyzed by logistic regression in a Bayesian framework.

- The model-predicted rates of safety parameters by tumor type based on E-R analyses are shown in **Table 1**

Table 1. Model-Predicted E-R Relationships for Safety Parameters

Endpoint	Model-Predicted Rate Estimate, % (95% CI)					
	BC	GC	*HER2*-mutant NSCLC		HER2-overexpressing NSCLC	
	5.4 mg/kg Q3W	6.4 mg/kg Q3W	5.4 mg/kg Q3W	6.4 mg/kg Q3W	5.4 mg/kg Q3W	6.4 mg/kg Q3W
Dose reduction associated with AE	22.9 (20.3-25.6)	29.5 (24.7-34.5)	20.8 (16.7-25.6)	26.3 (21.6-31.9)	20.8 (13.8-28.8)	26.1 (17.8-35)
Grade ≥3 AE	55.2 (52.1-58.3)	72.8 (68-77.7)	52 (46.8-57.8)	59.5 (53.9-65.3)	58.4 (49.2-67)	67.4 (58.4-75.2)
SAE	22.8 (20.3-25.5)	38.4 (32.9-43.4)	30.4 (25-36.5)	35.2 (29.4-41.6)	38.4 (30.5-47.9)	44.8 (36.5-54.5)
Anemia (laboratory-based), grade ≥3	9.2 (7.5-11.4)	29.6 (24.6-34.5)	9.4 (6.5-13)	10.8 (7.2-14.8)	2.5 (0.7-6.7)	2.8 (0.7-7.5)
Neutropenia (laboratory-based), grade ≥3	17.8 (15.6-20.5)	33.7 (29-38.6)	13.8 (10.3-17.7)	17.3 (13.1-22.2)	10 (5.6-15.3)	14.7 (9.1-21.3)
Thrombocytopenia (laboratory-based), grade ≥3	6.8 (5.3-8.6)	9.4 (6.6-12.6)	3.4 (1.7-5.9)	5 (2.5-8.3)	1.5 (0.3-3.8)	2.2 (0.5-5.7)
ILD any grade at day 180	6.7 (5.5-8)	8.8 (6.1-12.4)	14.5 (10.2-19.6)	15.4 (11.3-20.3)	15.6 (8.7-24.9)	14.3 (8.3-21.5)
ILD grade ≥3 at day 180	1.6 (1.1-2.4)	1.4 (0.5-3.5)	3.3 (1.6-6)	4.5 (2.1-7.9)	4.5 (1.5-9.7)	6 (2.1-12.5)

Based on posterior predictive checks, observed incidences of safety endpoints by tumor type were captured by the 95% prediction intervals of posterior simulations.

- Findings for ILD and other safety endpoints were generally consistent with those of previous model-predicted rates of AEs[4,5]
- Model-predicted AE rates were generally comparable between patients with *HER2*-mutant and HER2-overexpressing NSCLC at the same dose and were lower (up to a rate of 9%) with T-DXd 5.4 mg/kg Q3W compared with T-DXd 6.4 mg/kg Q3W
- Model-predicted rates of safety endpoints in patients with *HER2*-mutant NSCLC receiving T-DXd 5.4 mg/kg Q3W were comparable (±4%) to T-DXd 5.4 mg/kg in BC, except for about 8% higher SAE and any-grade ILD events in *HER2*-mutant NSCLC versus BC
- Patients with higher body weight (>95th percentile; >89 kg) had higher T-DXd and DXd exposures (31% to 37% higher AUC) compared with patients within normal body weight range based on PK analysis; however, despite the higher T-DXd or DXd exposures, no apparent or consistent trend was noted when comparing the model-estimated probability of safety endpoints in patients with higher body weight (75th to 90th percentile or >90th percentile) with those in the middle 50% (25th to 75th percentile) of body weight distribution for all evaluated safety endpoints (**Figure S3**)

Abbreviations

AE, adverse event; AFT, accelerated failure time; AST, aspartate aminotransferase; AUC, area under the serum concentration-time curve; AUC_{ss}, area under the serum concentration-time curve at steady state; BC, breast cancer; C_{avg}, average serum concentration; $C_{avg,cycle\,2}$, average serum concentration through the end of cycle 2; $C_{avg\text{-}TOE}$, average serum concentration to the time of safety event; C_{max}, maximum serum concentration; $C_{max,ss}$, maximum serum concentration at steady state; CDK4/6i, cyclin-dependent kinase 4/6 inhibitor; CrCL, creatinine clearance; CrI, credible interval; DXd, deruxtecan; E_{max}, maximum effect; ECHO/MUGA, echocardiogram/multigated acquisition; ECOG, Eastern Cooperative Oncology Group; E-R, exposure-response; GC, gastric cancer; GEJ, gastroesophageal junction; HER2, human epidermal growth factor receptor 2; IA, interim analysis; ILD, interstitial lung disease; NSCLC, non–small cell lung cancer; ORR, objective response rate; P, probability (or incidence) of modeled safety endpoint; PA, primary analysis; PK, pharmacokinetic; Q3W, every 3 weeks; R, randomization; SAE, serious adverse event; T-DXd, trastuzumab deruxtecan; TTE, time to event.

References

1. Nakada T et al. *Chem Pharm Bull (Tokyo).* 2019;67(3):173-185.
2. Ogitani Y et al. *Clin Cancer Res.* 2016;22(20):5097-5108.
3. Enhertu (fam-trastuzumab deruxtecan-nxki). Prescribing information. Daiichi Sankyo, Inc; 2024.
4. Yin O et al. *J Clin Pharm Ther.* 2021;110:986-996.
5. Li C et al. Presented at: San Antonio Breast Cancer Symposium; December 5-9, 2023; San Antonio, TX. PO3-04-01.
6. Yoshihara K et al. *J Clin Pharm.* 2023;63(11):1232-1243.
7. Li BT et al. *N Engl J Med.* 2022;386:241-251.
8. Goto K et al. *J Clin Oncol.* 2023;41:4852-4863.

Acknowledgments

We thank the patients participating in this study, as well as their families and caregivers. This study was sponsored by Daiichi Sankyo, in collaboration with AstraZeneca. In 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for T-DXd (DS-8201). Under the guidance of authors, editorial support was provided by Andre Wang, PharmD, and Jill Shults, PhD, of ApotheCom, funded by Daiichi Sankyo, Inc.

Disclosures

A. Khatri, M. Abutarif, and T. Garimella disclose employment by Daiichi Sankyo. D. Polhamus, R. Garcia, and T. Yoder disclose employment by Metrum Research Group.