075

Population Pharmacokinetics of Trastuzumab Deruxtecan in Subjects With *HER2*-Mutant and HER2-Overexpressing Non–Small Cell Lung Cancer

Amit Khatri,[1] Daniel Polhamus,[2] Malaz Abutarif,[1] Tushar Garimella[1]

[1]Quantitative Clinical Pharmacology, Daiichi Sankyo, Inc, Basking Ridge, NJ, USA;
[2]Metrum Research Group, Tariffville, CT, USA.

Objectives

- To update the previously developed population pharmacokinetics (PopPK) model for the antibody-drug conjugate trastuzumab deruxtecan (T-DXd) and its topoisomerase I inhibitor payload (DXd) with pharmacokinetic (PK) data from patients with human epidermal growth factor receptor 2 (*HER2*)-mutant or HER2-overexpressing non–small cell lung cancer (NSCLC) in the DESTINY-Lung01 and DESTINY-Lung02 studies:
 - To evaluate the impact of potential covariates on the PK of T-DXd and DXd
 - To estimate individual post hoc PK parameters used to derive exposure in subsequent exposure-response analyses
 - To compare exposures among subpopulations of interest, including by HER2 status, hepatic function, renal function, race-country, and country for the population with *HER2*-mutant NSCLC

Conclusions

- This updated PopPK analysis, with data from patients with NSCLC, supports T-DXd 5.4 mg/kg dosing in *HER2*-mutant NSCLC from a PK perspective
 - T-DXd and DXd exposures were similar across patients with breast cancer and *HER2*-mutant NSCLC or HER2-overexpressing NSCLC at each dose of T-DXd (5.4 and 6.4 mg/kg every 3 weeks [Q3W])
 - T-DXd and DXd exposures were similar in patients with NSCLC regardless of sex, region, race-country, and baseline hepatic and renal function
- Furthermore, the effects of covariates of PK on T-DXd and DXd were consistent with previous analyses,[1,2] and no new significant covariates were identified in this update

Supplementary content available
Copies of materials obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ACCP and the author of this poster.

This information is the intellectual property of the authors/presenters.
Please contact Amit Khatri at amit.khatri@daiichisankyo.com for permission to reprint and/or redistribute.
Presented at the 2024 American College of Clinical Pharmacology (ACCP) Annual Meeting;
September 8-10, 2024; Bethesda, Maryland

Background

- T-DXd is an antibody-drug conjugate composed of 3 components: a humanized immunoglobulin G1 monoclonal antibody targeting HER2, a tetrapeptide-based cleavable linker, and a topoisomerase I inhibitor payload[3-5]
- T-DXd is approved for the treatment of adult patients with unresectable or metastatic HER2-positive or HER2-low breast cancer, *HER2*-mutant NSCLC, HER2-positive gastric or gastroesophageal junction adenocarcinoma, and HER2-positive (immunohistochemistry 3+) solid tumors with no other treatment options, based on phase 2 or phase 3 trials[6]
 - The recommended dose of T-DXd is 5.4 mg/kg Q3W in all approved indications, except for patients with HER2-positive gastric or gastroesophageal junction adenocarcinoma, for whom the recommended dose is 6.4 mg/kg Q3W[6]
 - PopPK analyses of T-DXd in patients with HER2-positive or HER2-low breast cancer and other solid tumors from phase 1 to 3 clinical trials have been published[1,2]
 - T-DXd was evaluated at doses of 5.4 and 6.4 mg/kg Q3W in patients with *HER2*-mutant NSCLC in phase 1 and 2 trials, including DESTINY-Lung01 and DESTINY-Lung-02[7,8]

Methods

- Analysis was performed using PK data from patients with HER2-expressing breast cancer or other solid tumors from 4 phase 1 studies (NCT02564900, NCT03366428, NCT03368196, NCT03383692), 5 phase 2 studies (DESTINY-Gastric01 [NCT03329690], DESTINY-Breast01 [NCT03248492], DESTINY-Lung01 [NCT03505710], DESTINY-Lung02 [NCT04644237], DESTINYGastric02 [NCT04014075], and 2 phase 3 studies (DESTINY-Breast03 [NCT03529110], DESTINY-Breast04 [NCT03734029])
- In this updated analysis, data from patients with *HER2*-mutant or HER2-overexpressing NSCLC treated with T-DXd 5.4 mg/kg Q3W or 6.4 mg/kg Q3W in DESTINY-Lung01 and DESTINY-Lung02 and in a small cohort (n = 18) from a phase 1 study (NCT02564900) were included
- Sparse sampling for serum T-DXd and DXd concentrations was collected from patients before and after the end of the infusion for up to 8 cycles across phase 2 and 3 studies. Intensive PK sampling was included in earlier phase 1 and 2 studies[1]
- All patients who received at least 1 dose of T-DXd and had 1 evaluable postdose concentration of T-DXd or DXd were eligible for inclusion in the PopPK analysis
- The PopPK analysis was performed with a nonlinear mixed-effects modeling approach using NONMEM (version 7.4.4), with sequential fitting of T-DXd and DXd data. The structural PK model is shown in **Figure 1**
- Covariate effects from the previous PopPK analysis were retained in the model as prespecified covariates unless diagnostic plots showed any indication for modification. NSCLC *HER2*-mutant and HER2-overexpressing cancer types were also included under a full modeling approach
- Additional candidate covariates evaluated were race, race-country (Asian from Japan, Asian not from Japan, non-Asian), sex, presence of antidrug antibodies (yes/no), creatinine clearance (CrCl), hepatic impairment (National Cancer Institute criteria),[9] and Eastern Cooperative Oncology Group performance status
- The effect of covariates was evaluated by use of both univariate graphical analyses and multivariate analyses on steady-state exposure of T-DXd and DXd
- Individual exposures, such as maximum serum concentration (C_{max}), minimum serum concentration (C_{min} or C_{trough}), and area under the serum time-concentration curve (AUC) at cycle 1 and steady-state (AUC_{ss}; cycle 9), for both T-DXd and DXd were calculated using the updated PopPK model and the individual post hoc PK parameters

Figure 1. Structural PopPK model for T-DXd and DXd

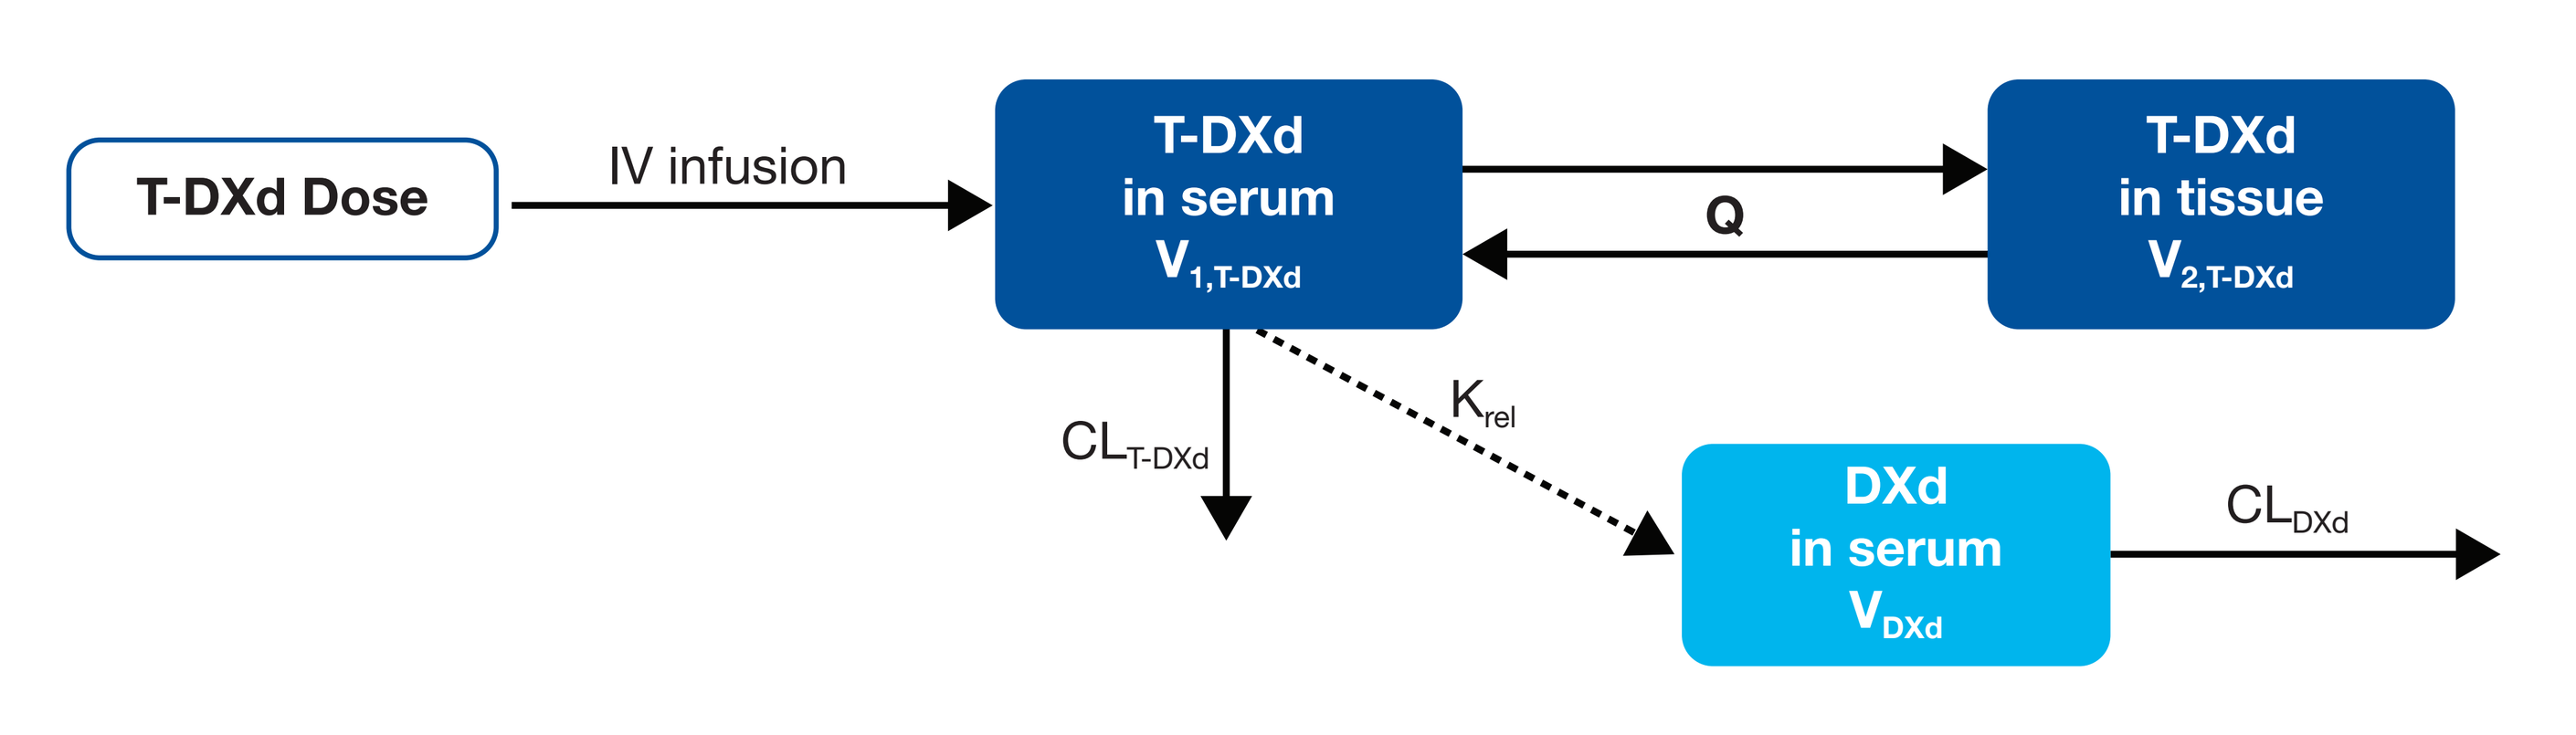
T-DXd Dose
IV infusion
T-DXd in serum $V_{1,T\text{-}DXd}$
Q
T-DXd in tissue $V_{2,T\text{-}DXd}$
$CL_{T\text{-}DXd}$
K_{rel}
DXd in serum V_{DXd}
CL_{DXd}

Adapted with permission from Yin O et al. Population pharmacokinetics of trastuzumab deruxtecan in patients with HER2-positive breast cancer and other solid tumors. *Clin Pharmacol Ther.* 2021;109:1314-1325; published by Wiley Periodicals LLC on behalf of American Society for Clinical Pharmacology and Therapeutics.

Results

Patients

- Data from 1821 patients were included from the 11 studies, contributing 24,558 evaluable T-DXd concentrations and 24,507 evaluable DXd concentrations to the PopPK analysis
 - T-DXd dose ranged from 0.8 mg/kg to 8.0 mg/kg across the studies
 - 1128 patients (61.9%) had breast cancer, 293 patients (16.1%) had gastric cancer, 346 patients (19.0%) had NSCLC, and 55 patients (3.1%) had other cancer types
 - Among patients with NSCLC, 246 had *HER2*-mutant NSCLC and 100 had HER2-overexpressing NSCLC
 - Most patients in the pooled data set were female (n = 1425; 78.2%), and the median age was 58.0 years (range, 20.0-96.0 years)
 - Overall, 998 patients (54.9%) were Asian, 646 patients (35.5%) were White, 129 patients (7.1%) were other race, and 38 patients (2.1%) were African American

PopPK Final Models for T-DXd and DXd

- The results of the T-DXd and DXd PK analysis, including parameter estimates, were consistent with previous analyses[1,2]
- The effect of NSCLC (*HER2*-mutant and HER2-overexpressing) was not significant. No additional covariates were included in the T-DXd or DXd PopPK model compared with previous analyses
- Relationships between patient-specific covariates and T-DXd PK parameters are shown in **Equations 1-3,** and those for DXd PK parameters are shown in **Equations 4-6**
 - Clearance of T-DXd ($CL_{T\text{-}DXd}$) and central volume of T-DXd ($V_{1,T\text{-}DXd}$) and peripheral volume of T-DXd ($V_{2,T\text{-}DXd}$) were estimated at 0.409 L/d, 2.69 L, and 6.46 L, respectively. Interindividual variability (IIV) for $CL_{T\text{-}DXd}$ and $V_{1,T\text{-}DXd}$ and $V_{2,T\text{-}DXd}$ were 25%, 16%, and 81%, respectively
 - DXd elimination clearance was estimated at 19.6 L/h with IIV of 30%

Equation 1. Elimination Clearance of T-DXd

$$CL_{T\text{-}DXd} = 0.409\ \text{L/d} \times \left(\frac{Weight\ in\ kg}{57.8}\right)^{0.399} \times \left(\frac{Albumin\ in\ \frac{g}{L}}{40}\right)^{-0.472} \times \left(\frac{Tumor\ size\ in\ mm}{57}\right)^{0.0506}$$

(× 0.915, if Asian from Japan), (× 1.17, if GC), (× 1.02, if *HER2*-mutant NSCLC), (× 1.14, if HER2-overexpressing NSCLC), or (× 1.07, if other cancer)

Equation 2. T-DXd Central Volume of Distribution

$$V_{1,T\text{-}DXd} = 2.69\ \text{L} \times \left(\frac{Weight\ in\ kg}{57.8}\right)^{0.443}$$

(× 1.10, if GC), (× 0.93, if *HER2*-mutant NSCLC), (× 0.982, if HER2-overexpressing NSCLC), or (× 1.13, if male)

Equation 3. T-DXd Peripheral Volume of Distribution

$$V_{2,T\text{-}DXd} = 6.46\ \text{L}\ (\times\ 0.762,\ \text{if Asian from Japan})$$

Equation 4. Elimination Clearance of DXd

$$CL_{DXd} = 19.6\ \text{L/h} \times \left(\frac{Weight\ in\ kg}{57.8}\right)^{0.341} \times \left(\frac{Total\ bilirubin\ in\ \frac{\mu mol}{L}}{8}\right)^{-0.139} \times \left(\frac{AST\ in\ \frac{U}{L}}{30}\right)^{-0.195}$$

(× 0.891, if itraconazole), (× 0.878, if ritonavir)

Equation 5. DXd Volume of Distribution

$$V_{DXd} = 17.0\ \text{L/m}^2 \times \text{Body surface area in m}^2 \times \left(\frac{Age\ in\ y}{57}\right)^{0.59}$$

(× 1.58, if FL-DP2), (× 0.753, if NSCLC), (× 0.764, if non-Asian)

Equation 6. Release Rate Constant

$$K_{rel} = 0.0197\ \text{h}^{-1} \times \text{cycle}^{-0.155}\ (\times\ 0.732,\ \text{if cycle} > 1)$$

Covariate Effects of T-DXd and DXd on PK

- The covariate effects on T-DXd and DXd AUC_{ss} are shown in **Figure 2**
- For both T-DXd and DXd AUC_{ss}, most covariate effects were contained within the 0.8-1.25 exposure ratio interval relative to a reference patient, suggesting there is no clinically meaningful effect of the covariates on AUC_{ss} (**Figure 2**) with the following exceptions:
 - Patients with extreme values of body weight (90 kg; 95th percentile) had 31% and 37% higher T-DXd and DXd AUC_{ss}, respectively, relative to a typical patient with a weight of 58 kg; however, these differences in T-DXd or DXd exposures were not considered clinically meaningful based on exposure-response and multivariate analyses
- PK exposure estimates of T-DXd and DXd after a dosing regimen of T-DXd 5.4 or 6.4 mg/kg Q3W were:
 - Similar in patients with *HER2*-mutant NSCLC and HER2-overexpressing NSCLC (**Figure 3**)
 - Similar between patients with breast cancer and patients with *HER2*-mutant and HER2-overexpressing NSCLC
- T-DXd and DXd exposure estimates were similar in patients with NSCLC according to sex (**Figure 4**), region (**Figure 5**), and race-country (**Figure 6**)
- T-DXd and DXd exposure estimates were similar in patients with NSCLC according to baseline hepatic (**Figure 7**) and renal function status (**Figure 8**)
- Similar trends were observed for C_{max} and C_{min} at steady state (QR code)

Figure 2. Covariate Effects on AUC_{ss} of (A) T-DXd and (B) DXd

A

Covariate		Median (95% CI)
Baseline body weight (kg)	If 44 kg (5th percentile)	0.848 (0.834-0.860)
	If 52 kg (25th percentile)	0.938 (0.932-0.943)
	If 60 kg (50th percentile)	1.02 (1.02-1.03)
	If 70 kg (75th percentile)	1.12 (1.11-1.14)
	If 90 kg (95th percentile)	1.31 (1.28-1.34)
Baseline target tumor size (mm)	If 12 mm (5th percentile)	1.08 (1.05-1.11)
	If 31 mm (25th percentile)	1.03 (1.02-1.04)
	If 54 mm (50th percentile)	1.00 (1.00-1.00)
	If 87 mm (75th percentile)	0.979(0.973-0.986)
	If 155 mm (95th percentile)	0.952 (0.937-0.968)
Sex	If Male	1.00 (1.00-1.00)
Cancer type	If NSCLC overexpressing	0.882 (0.835-0.936)
	If NSCLC mutant	0.986 (0.953-1.02)
	If CRC or others	0.937 (0.868-1.01)
	If Gastric cancer	0.859 (0.827-0.892)
Race-country	If Asian from Japan	1.10 (1.07-1.13)
Baseline serum albumin (g/L)	If 31 g/L (5th percentile)	0.888 (0.868-0.910)
	If 37 g/L (25th percentile)	0.965 (0.958-0.972)
	If 40 g/L (50th percentile)	1.00 (1.00-1.00)
	If 43 g/L (75th percentile)	1.03 (1.03-1.04)
	If 46 g/L (95th percentile)	1.07 (1.05-1.08)

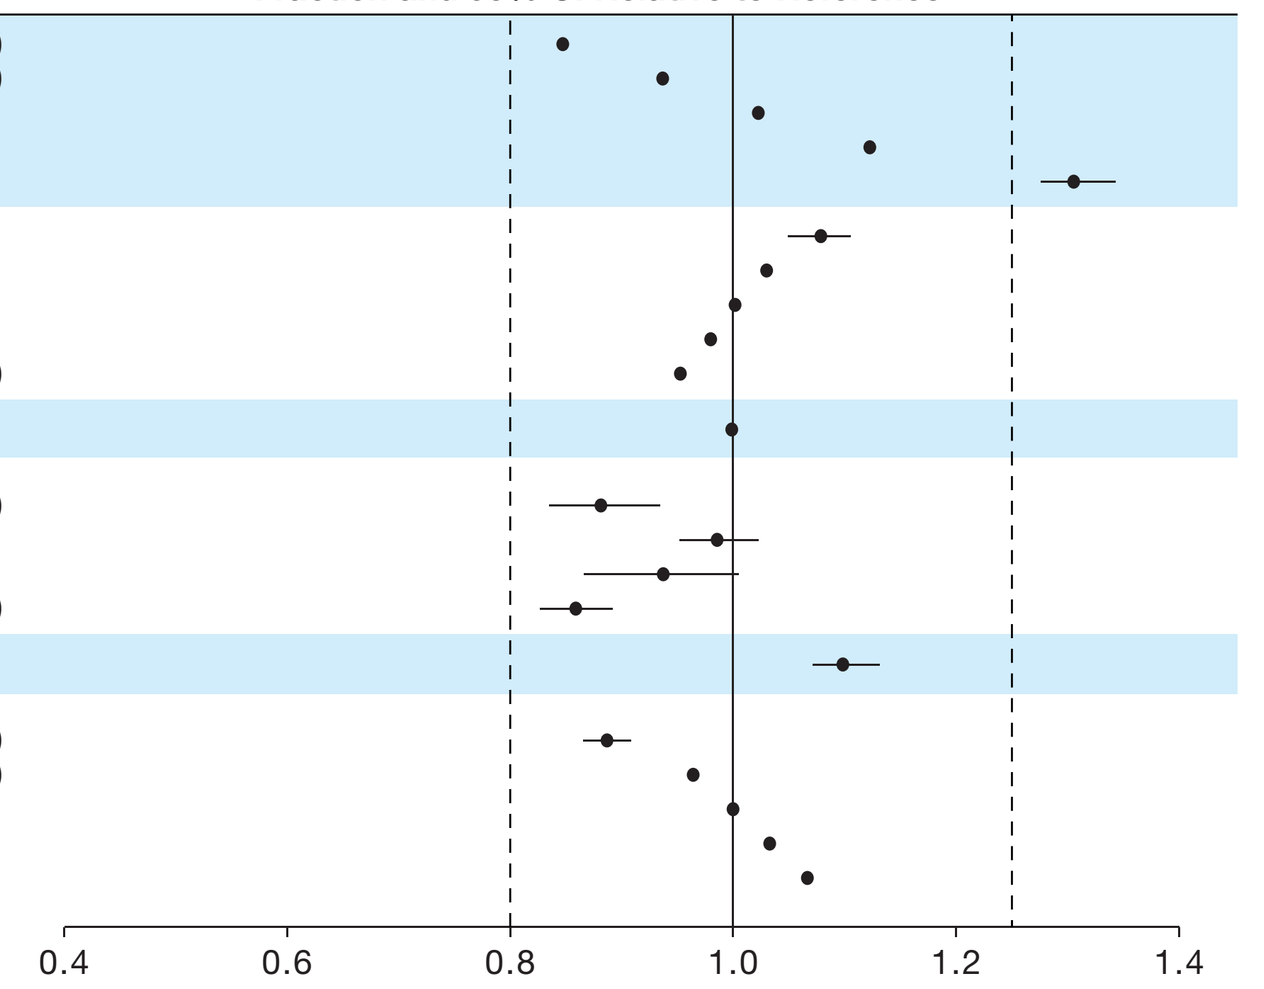
Fraction and 95% CI Relative to Reference
0.4
0.6
0.8
1.0
1.2
1.4

B

Covariate		Median (95% CI)
Baseline body weight (kg)	If 44 kg (5th percentile)	0.824 (0.798-0.850)
	If 52 kg (25th percentile)	0.928 (0.916-0.939)
	If 60 kg (50th percentile)	1.03 (1.02-1.03)
	If 70 kg (75th percentile)	1.15 (1.12-1.17)
	If 90 kg (95th percentile)	1.37 (1.30-1.44)
AST (U/L)	If 15 U/L (5th percentile)	0.873 (0.850-0.897)
	If 20 U/L (25th percentile)	0.924 (0.909-0.939)
	If 27 U/L (50th percentile)	0.980 (0.976-0.984)
	If 41 U/L (75th percentile)	1.06 (1.05-1.08)
	If 85 U/L (95th percentile)	1.23 (1.18-1.28)
Cancer type	If NSCLC overexpressing	0.864 (0.815-0.915)
	If NSCLC mutant	0.917 (0.888-0.947)
	If CRC or others	0.937 (0.869-1.01)
	If Gastric cancer	0.944 (0.910-0.983)
Baseline total bilirubin (μmol/L)	If 3.4 μmol/L (5th percentile)	0.888 (0.860-0.916)
	If 5.1 μmol/L (25th percentile)	0.939 (0.924-0.955)
	If 7.7 μmol/L (50th percentile)	0.995 (0.993-0.996)
	If 10.3 μmol/L (75th percentile)	1.04(1.03-1.05)
	If 17.1 μmol/L (95th percentile)	1.11 (1.08-1.14)

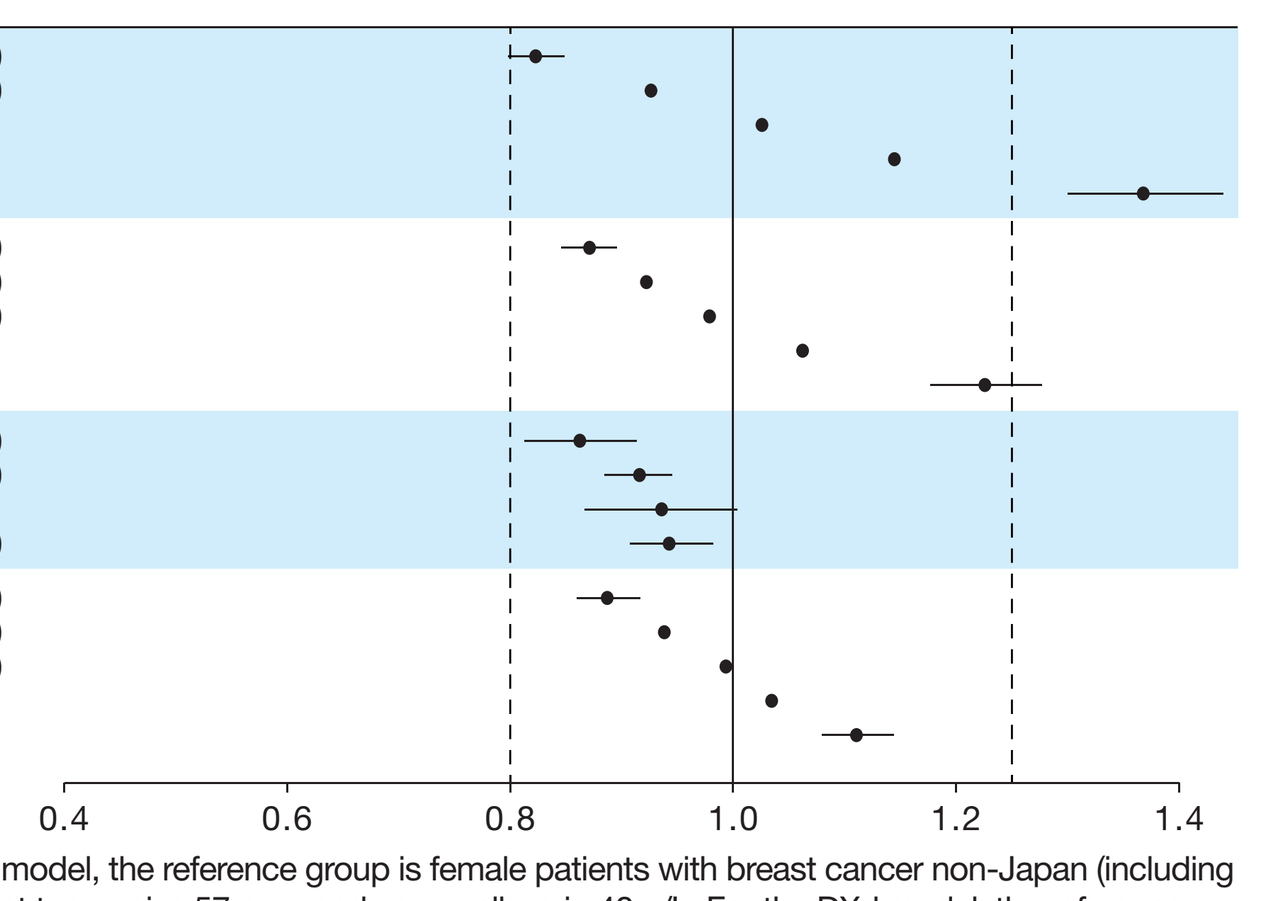
Fraction and 95% CI Relative to Reference
0.4
0.6
0.8
1.0
1.2
1.4

Points and whiskers represent the median and 90% CI, respectively. For the T-DXd model, the reference group is female patients with breast cancer non-Japan (including Asian not from Japan and non-Asian) with baseline mean body weight 57.8 kg, target tumor size 57 mm, and serum albumin 40 g/L. For the DXd model, the reference group is patients with breast cancer Asian (not from Japan) with baseline mean body weight 57.8 kg, aspartate aminotransferase 30 U/L, and total bilirubin 8 μmol/L.

Figure 3. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With Breast Cancer or NSCLC

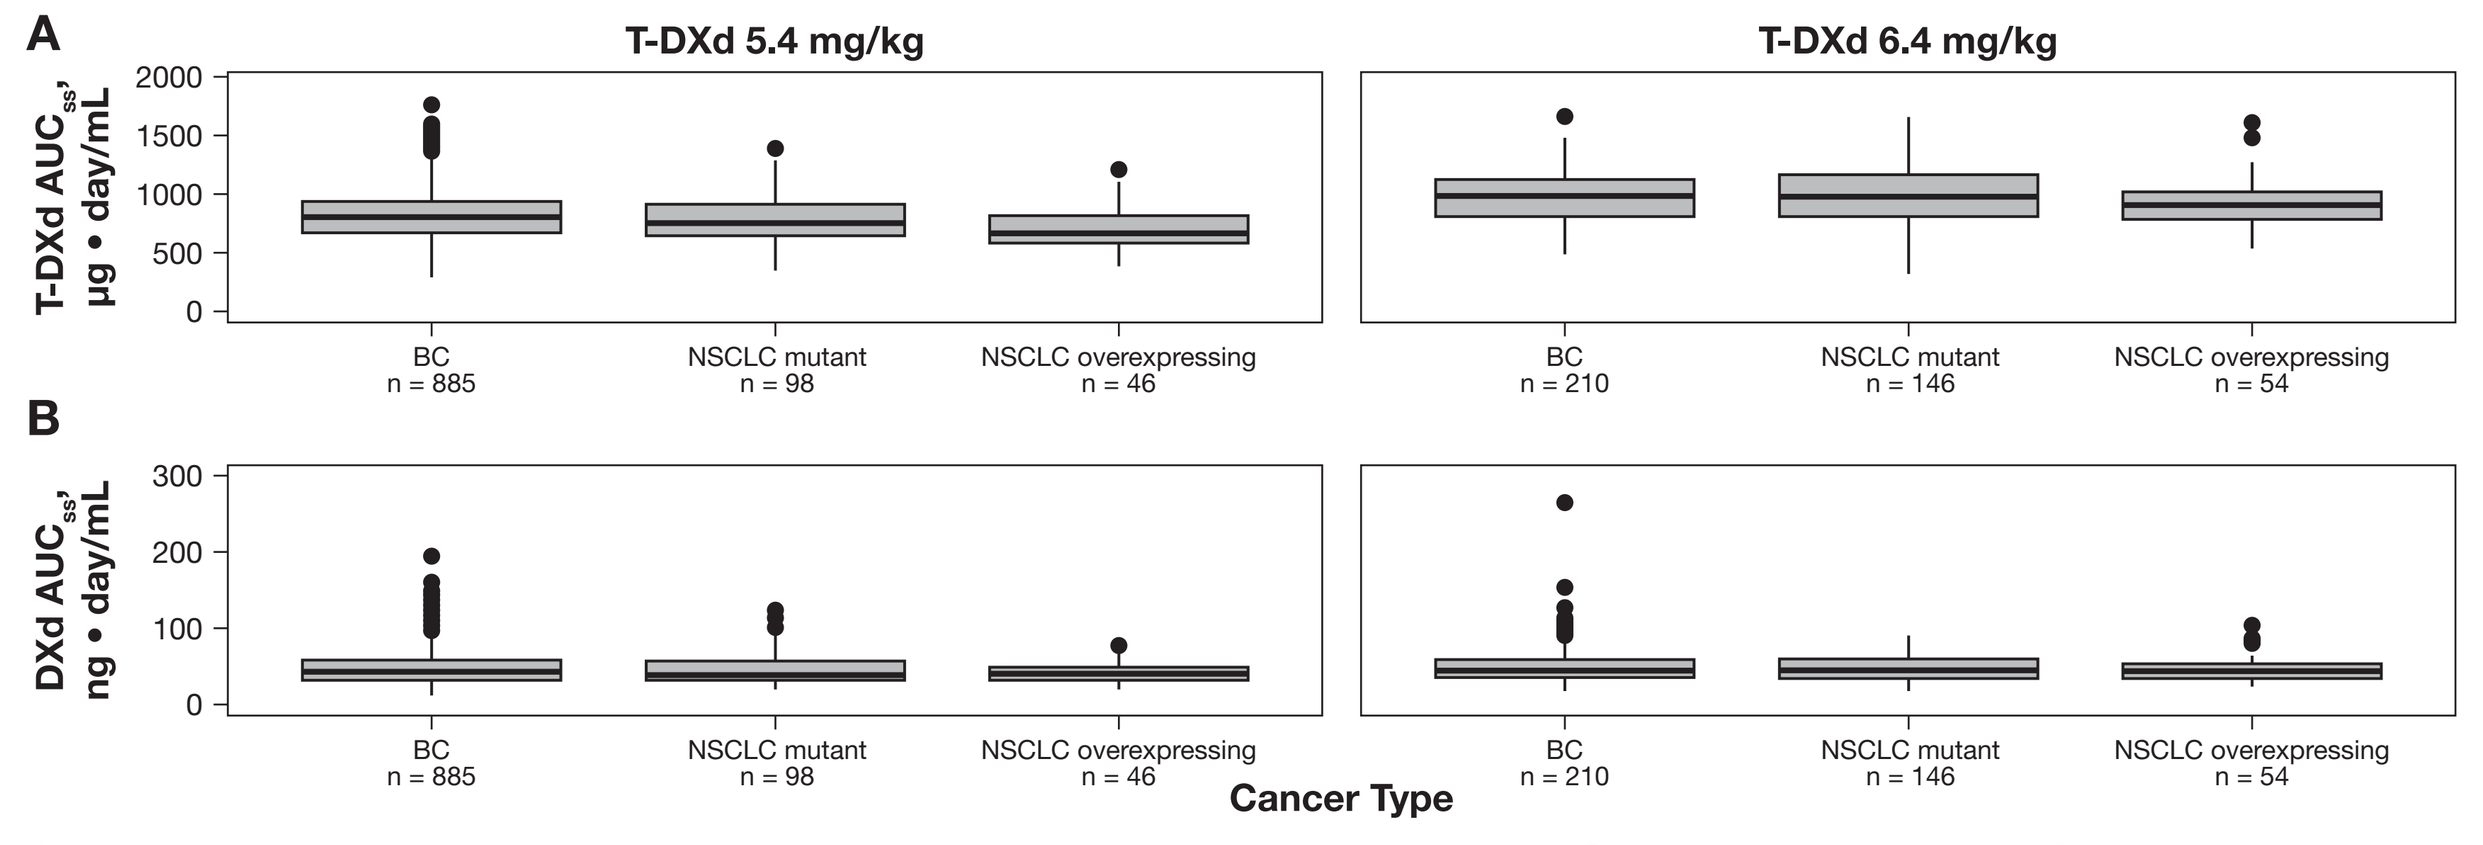
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
BC n = 885
NSCLC mutant n = 98
NSCLC overexpressing n = 46
BC n = 210
NSCLC mutant n = 146
NSCLC overexpressing n = 54
B
DXd AUCss, ng • day/mL
Cancer Type

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 times the interquartile range. Points represent data outside the whiskers.

Figure 4. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With NSCLC by Sex

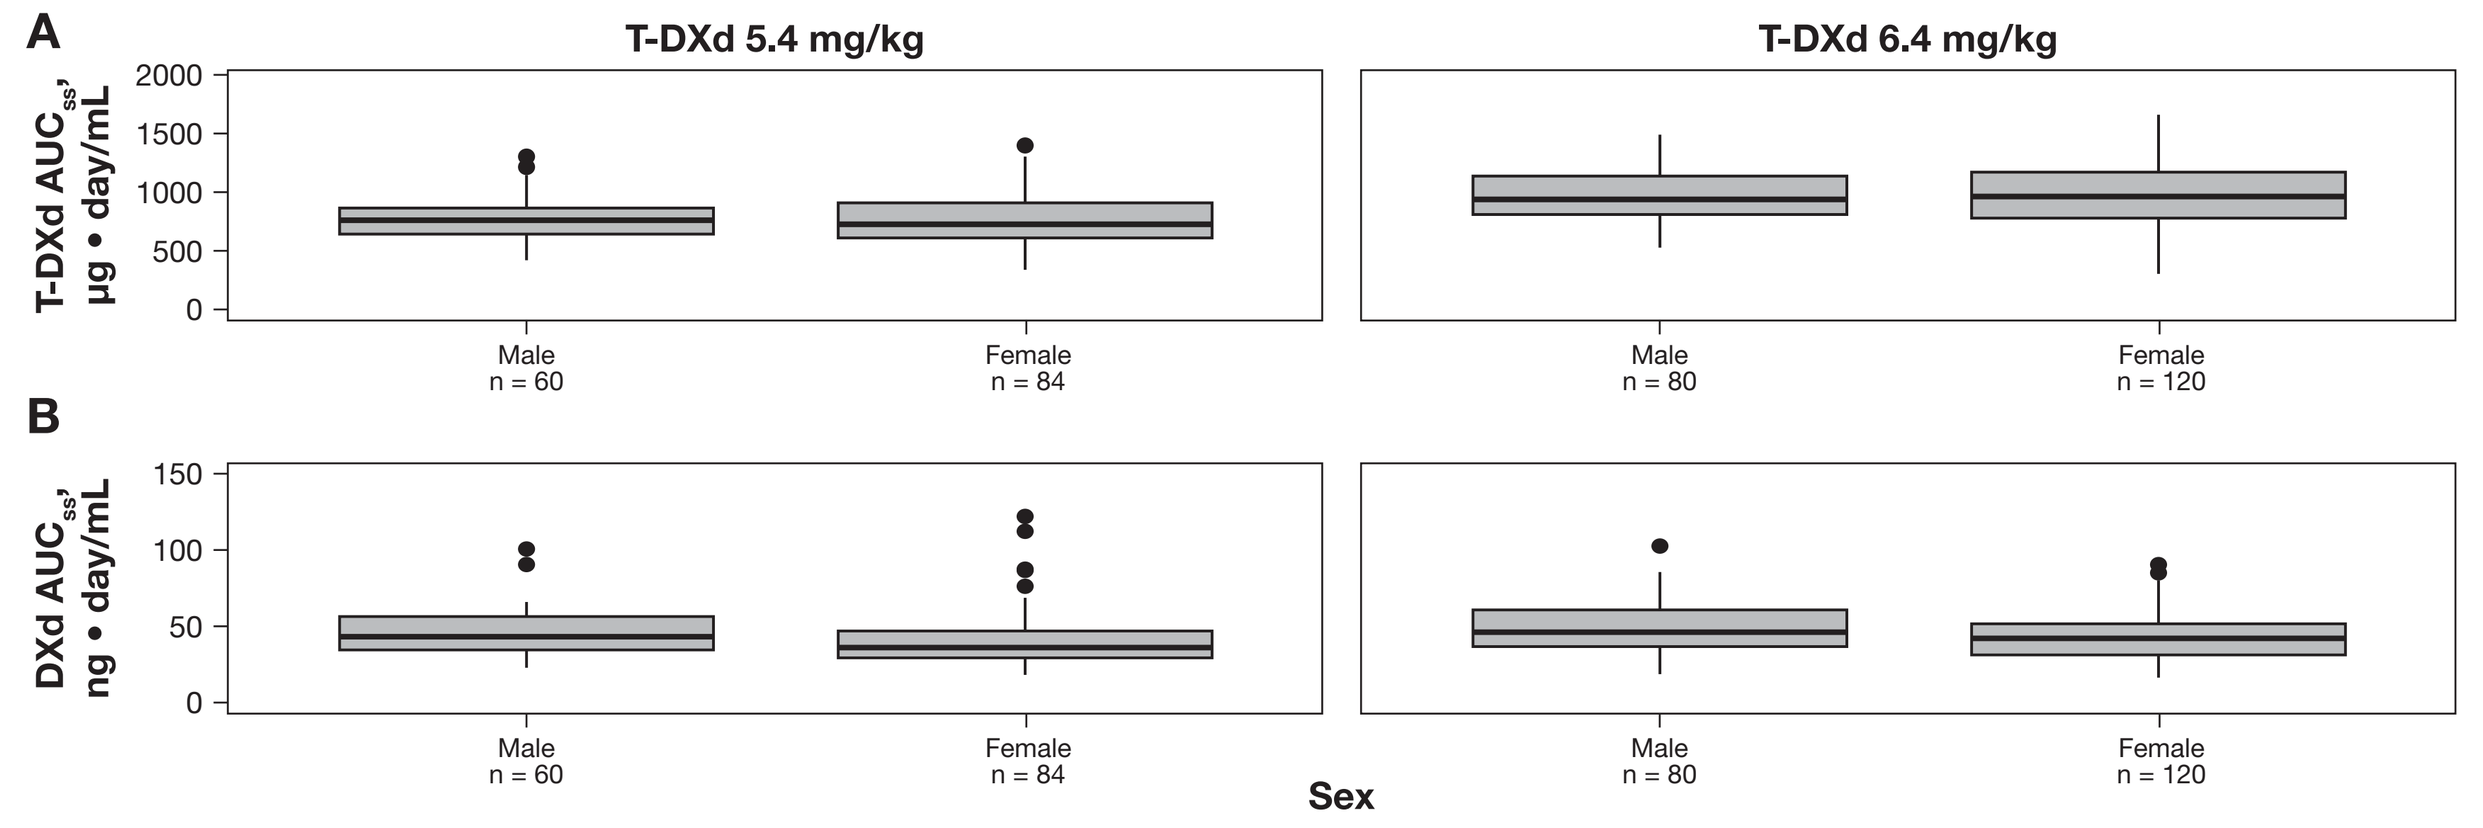
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
Male n = 60
Female n = 84
Male n = 80
Female n = 120
B
DXd AUCss, ng • day/mL
Sex

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 times the interquartile range. Points represent data outside the whiskers.

Figure 5. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With NSCLC by Region

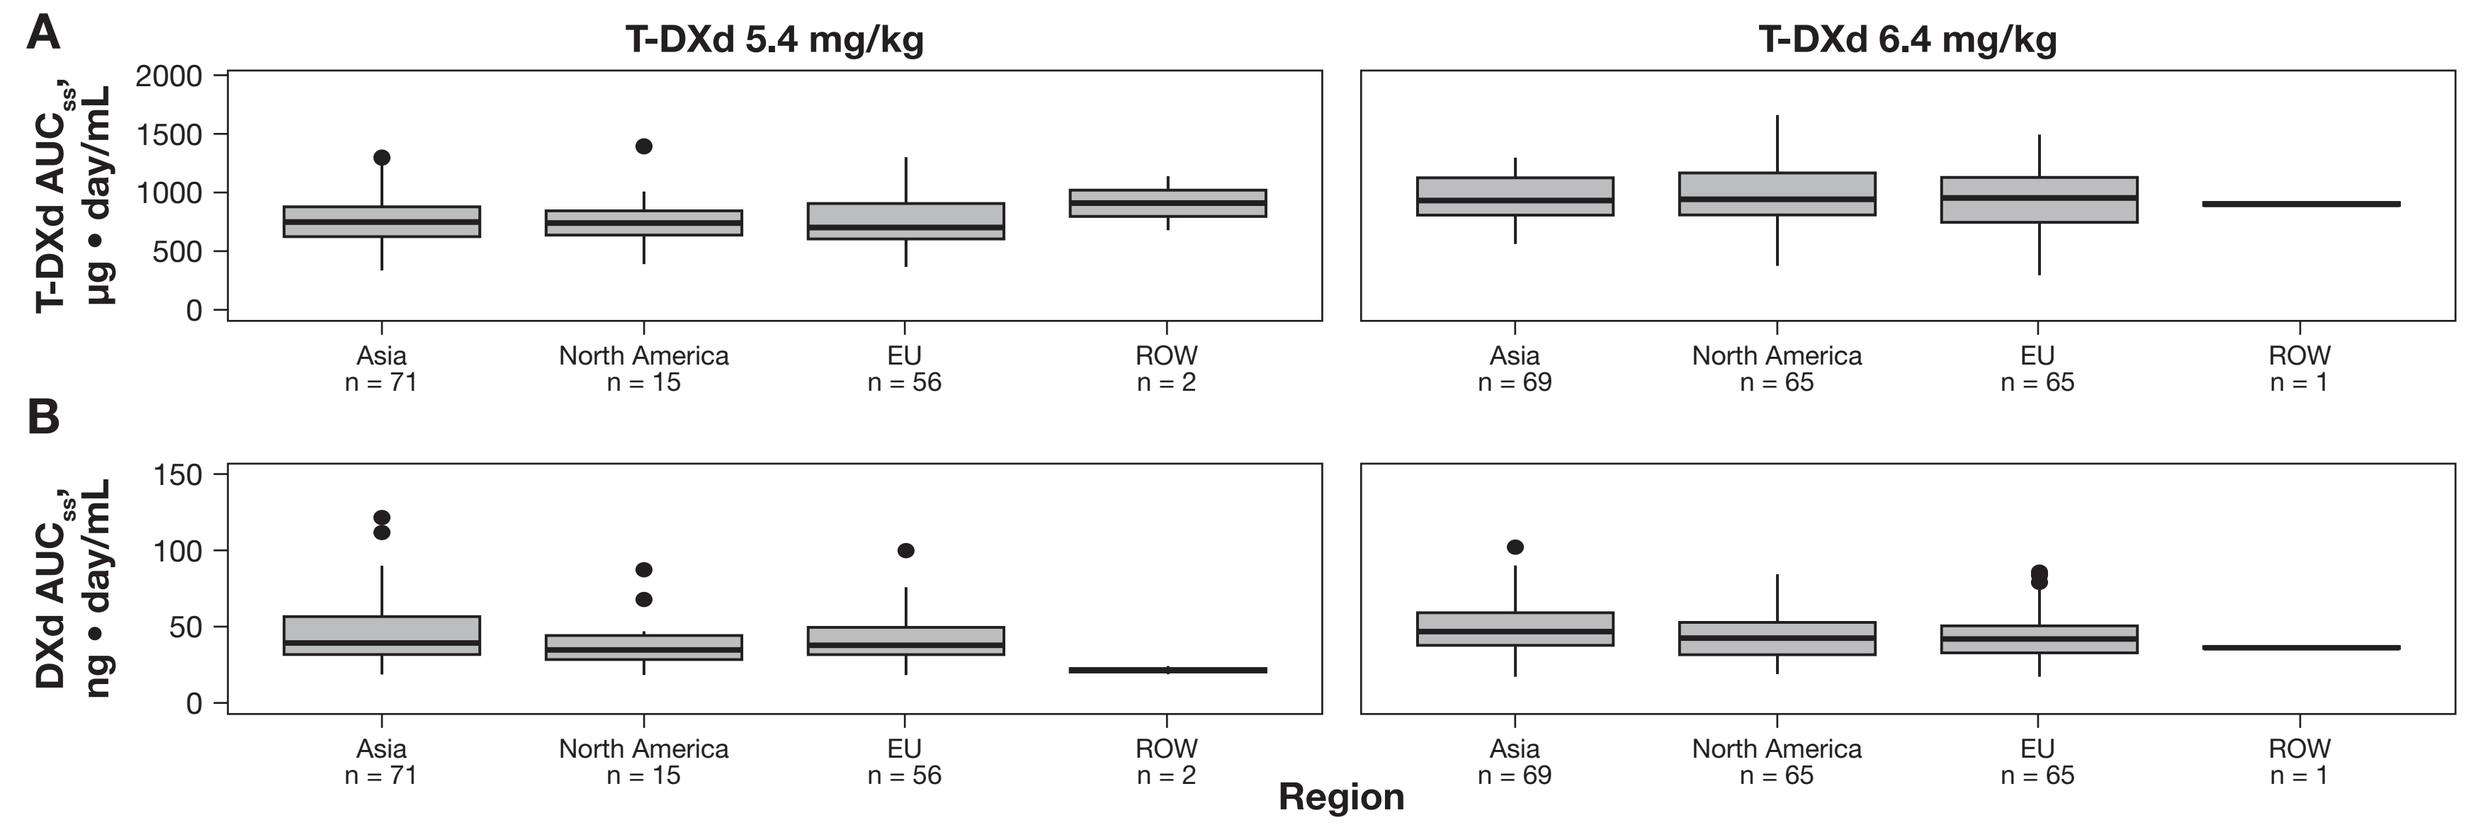
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
Asia n = 71
North America n = 15
EU n = 56
ROW n = 2
Asia n = 69
North America n = 65
EU n = 65
ROW n = 1
B
DXd AUCss, ng • day/mL
Region

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 times the interquartile range. Points represent data outside the whiskers.

Figure 6. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With NSCLC by Race-Country

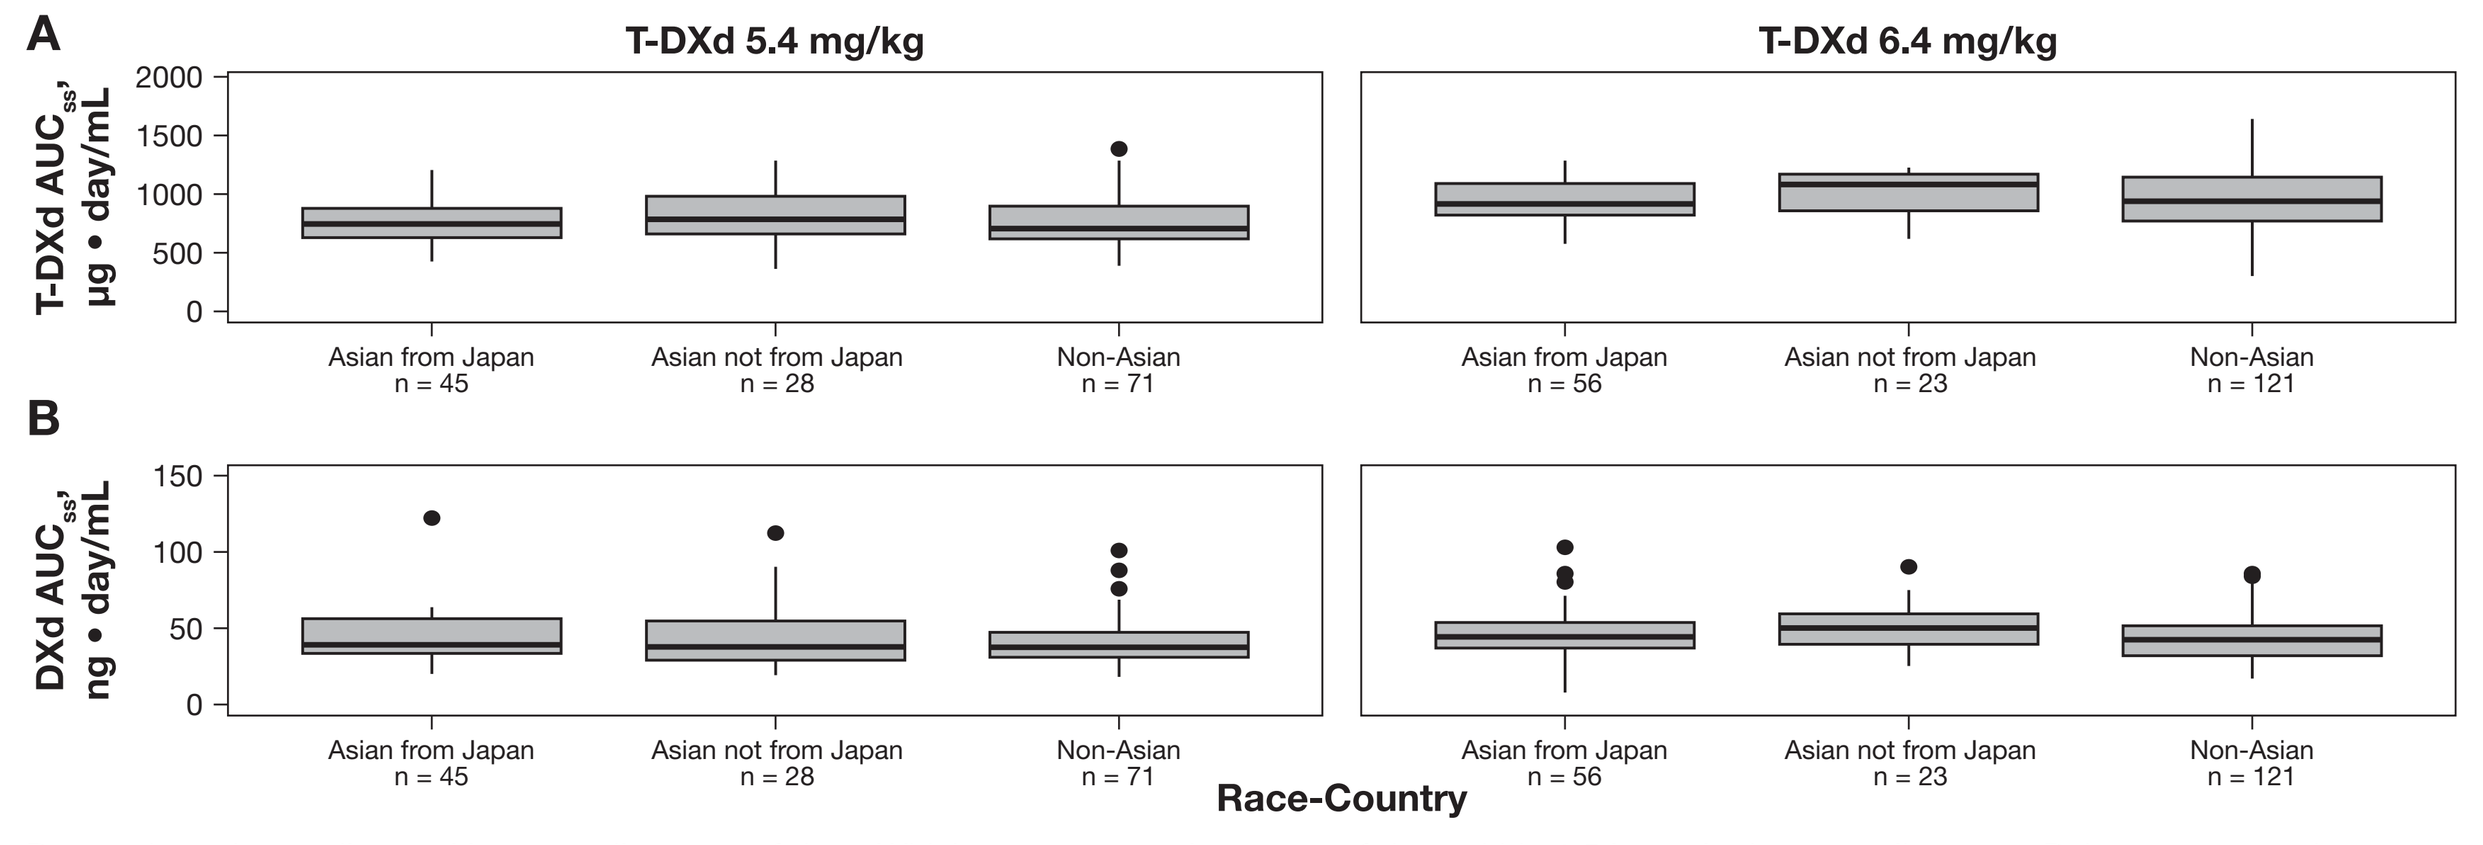
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
Asian from Japan n = 45
Asian not from Japan n = 28
Non-Asian n = 71
Asian from Japan n = 56
Asian not from Japan n = 23
Non-Asian n = 121
B
DXd AUCss, ng • day/mL
Race-Country

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 imes the interquartile range. Points represent data outside the whiskers.

Figure 7. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With NSCLC by Hepatic Function Status

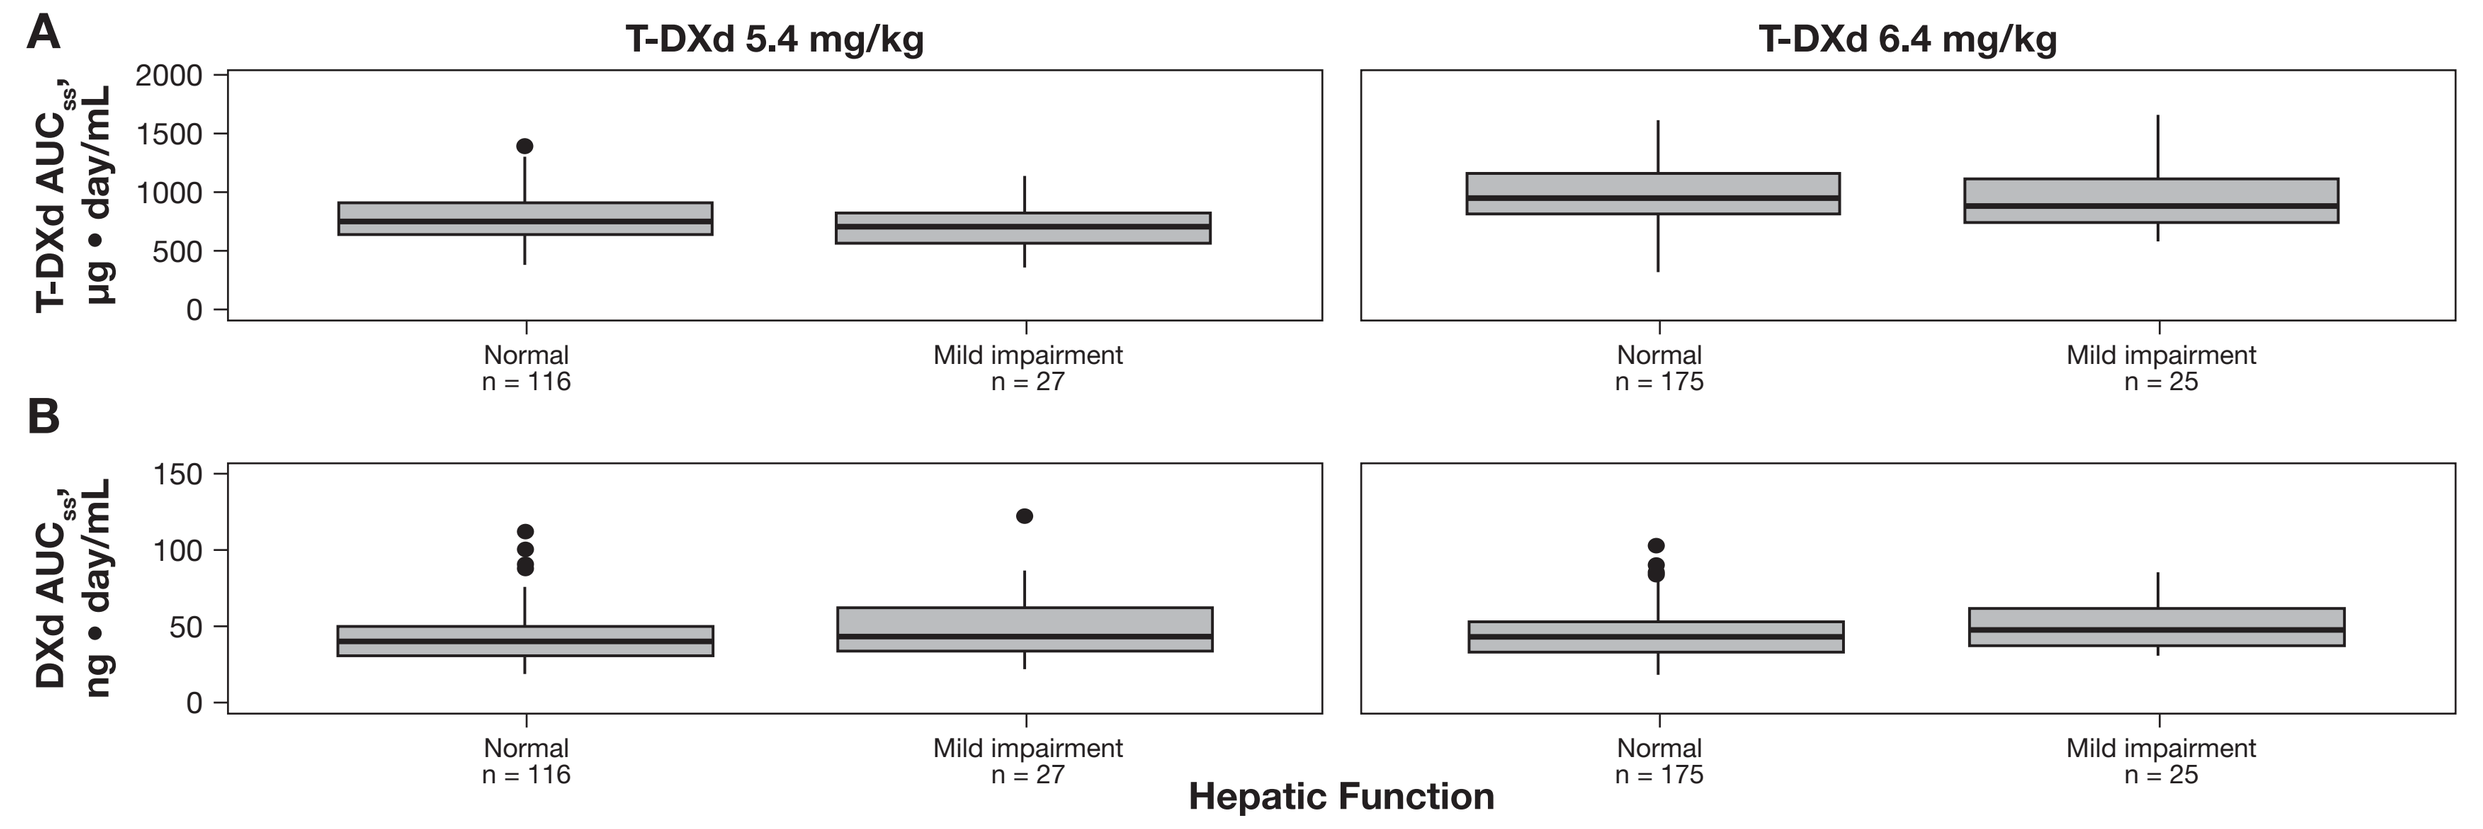
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
Normal n = 116
Mild impairment n = 27
Normal n = 175
Mild impairment n = 25
B
DXd AUCss, ng • day/mL
Hepatic Function

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 times the interquartile range. Points represent data outside the whiskers. Hepatic function was defined as normal or mild impairment according to NCI criteria.[9]

Figure 8. Steady State (A) T-DXd and (B) DXd Exposure After Administration of T-DXd 5.4 mg/kg or 6.4 mg/kg Q3W in Patients With NSCLC by Renal Function Status

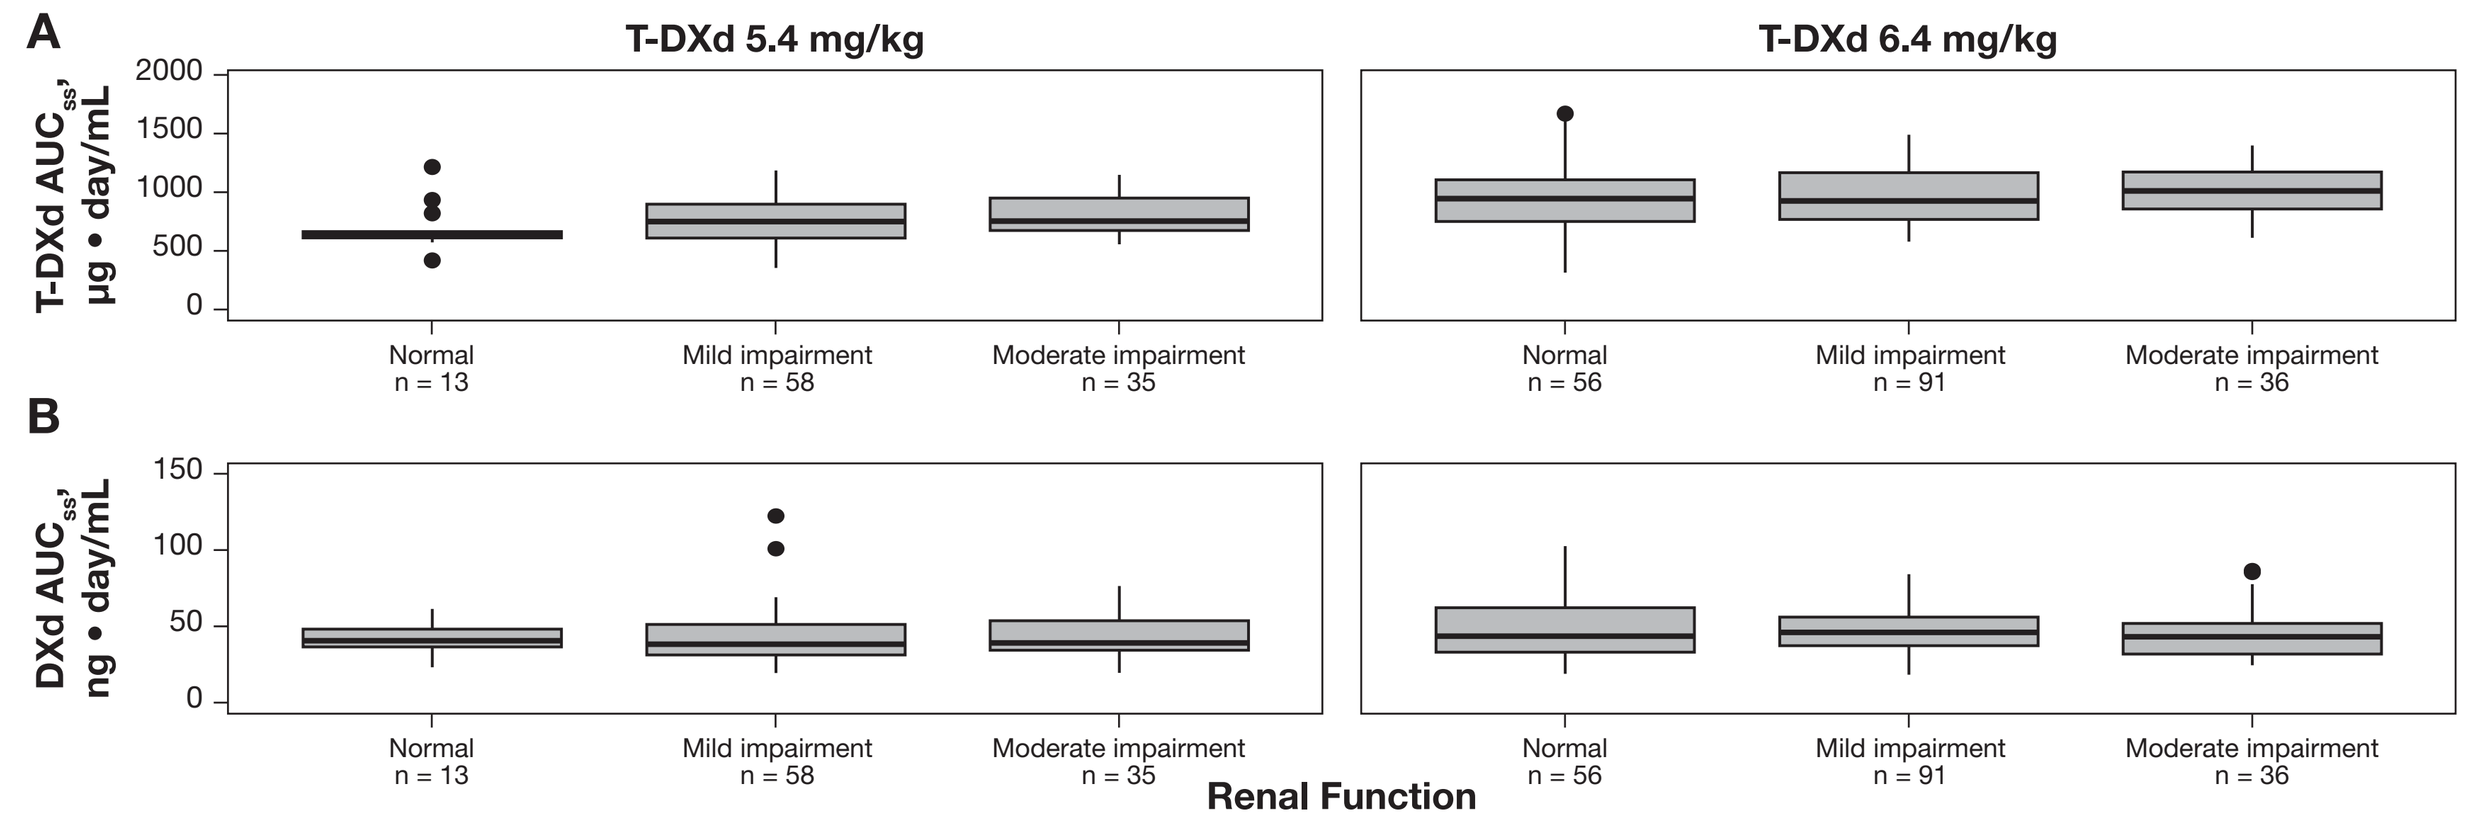
A
T-DXd 5.4 mg/kg
T-DXd 6.4 mg/kg
T-DXd AUCss, µg • day/mL
Normal n = 13
Mild impairment n = 58
Moderate impairment n = 35
Normal n = 56
Mild impairment n = 91
Moderate impairment n = 36
B
DXd AUCss, ng • day/mL
Renal Function

Boxes show the median and interquartile range of data. Whiskers represent the extent of data within 1.5 times the interquartile range. Points represent data outside the whiskers. Normal renal function or mild renal impairment were classified according to Cockcroft-Gault–calculated CrCl criteria.[10]

Abbreviations

AST, aspartate aminotransferase; AUC_{ss}, area under the serum time-concentration curve—steady state; BC, breast cancer; CL, clearance; CL_{DXd}, clearance of DXd; $CL_{T\text{-}DXd}$, clearance of T-DXd; C_{max}, maximum serum concentration; C_{min}, minimum serum concentration; CRC, colorectal cancer; CrCl, creatinine clearance; DXd, deruxtecan; EU, European Union; GC, gastric cancer; HER2, human epidermal growth factor receptor 2; IV, intravenous; K_{rel}, release rate constant; NCI, National Cancer Institute; NSCLC, non–small cell lung cancer; PK, pharmacokinetics; Pop, population; Q, distributional clearance for T-DXd; Q3W, every 3 weeks; ROW, rest of world; T-DXd, trastuzumab deruxtecan; $V_{1,T\text{-}DXd}$, central volume of T-DXd; $V_{2,T\text{-}DXd}$, peripheral volume of T-DXd; V_{DXd}, volume of DXd.

References

1. Yin O et al. *Clin Pharmacol Ther.* 2021;109:1314-1325.
2. Lu Z et al. Presented at ASCPT 2024 Annual Meeting; March 27-29, 2024; Colorado Springs, CO. PII-136.
3. Nakada T et al. *Chem Pharm Bull.* 2019;67:173-185.
4. Ogitani Y et al. *Clin Cancer Res.* 2016;22:5097-5108.
5. Trail P et al. *Pharmacol Ther.* 2018;181:126-142.
6. Enhertu (fam-trastuzumab deruxtecan-nxki). Prescribing information. Daiichi Sankyo, Inc., 2024.
7. Li BT et al. *N Engl J Med.* 2022;386:241-251.
8. Goto K et al. *J Clin Oncol.* 2023;41:4852-4863.
9. Han AN et al. *Curr Pharmacol Rep.* 2021;7:213-226.
10. Cockcroft DW, Gault MH. *Nephron.* 1976;16:31-41.

Acknowledgments

We thank the patients participating in this study, as well as their families and caregivers. This study was sponsored by Daiichi Sankyo, in collaboration with AstraZeneca. In 2019, AstraZeneca entered into a global development and commercialization collaboration agreement with Daiichi Sankyo for T-DXd (DS-8201). Under the guidance of authors, editorial support was provided by Sara Duggan, PhD, and Greg Town, BSc, of ApotheCom, funded by Daiichi Sankyo, Inc.

Disclosures

A. Khatri, M. Abutarif, and T. Garimella disclose employment by Daiichi Sankyo. E Cobbina is a former employee of Daiichi Sankyo.

DESTINY