First-in-Human Evaluation of Pharmacokinetics and Safety of Tilpisertib Fosmecarbil, an Oral Prodrug of a Tumor Progression Locus 2 Inhibitor, in Healthy Volunteers

Elijah J Weber[1], Xin Qi[1], Ann Qin[1], Frank Hong[1], Priyanka A Madia[1], Ingrid J Chang[1], Matthew McKevitt[1], and Ahmed A Othman[1]

[1]Gilead Sciences, Foster City, CA, USA

Poster #061

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Conclusions

- Single (up to 1500 mg) or multiple (up to 900 mg) oral doses of TIP were generally well tolerated in healthy participants
- TIP had a favorable pharmacokinetic profile that is supportive of once-daily dosing
- The pharmacokinetic and safety profiles of TIP in healthy volunteers have informed dose selection for an ongoing phase 2 trial of TIP in patients with UC (ClinicalTrials.gov: NCT06029972)

Plain Language Summary

- Inflammatory bowel disease may be caused in part by an overreaction to microbes in the bowels. A drug candidate called tilpisertib fosmecarbil (TIP) may be able to treat inflammatory bowel disease by blocking a protein called TPL2 that may be involved in the overreaction
- Varying doses of TIP were given to healthy volunteers either once, or every day for 10 days, to see how tolerable it is, and how long it stays in the body
- TIP was generally well tolerated at all doses given, and once-daily dosing resulted in levels of TIP in the body that may be high enough to block TPL2
- These results are helping us to decide how much TIP to give participants in future clinical trials that will test if this drug can be used to treat patients with inflammatory bowel disease

Introduction

- Inflammatory bowel disease (IBD), including ulcerative colitis (UC) and Crohn's disease, is a chronic and relapsing condition thought to be caused in part by inappropriate inflammatory responses to commensal microbiota[1]
- Tumor progression locus 2 (TPL2) is a cytoplasmic serine/threonine kinase and the primary regulator of extracellular signal-regulated kinase-mediated gene expression downstream of multiple proinflammatory stimuli including bacterial products, TNFa, and IL-1B[2]
- The prodrug tilpisertib fosmecarbil (TIP) is metabolized to GS-4875, a potent first-in-class TPL2 inhibitor, and is being investigated as a potential treatment for UC

Objectives

- This first-in-human study aimed to evaluate the pharmacokinetics, safety, and tolerability of escalating single and multiple oral doses of TIP in healthy participants

Methods

Study Design

- This was a double-blind, randomized, placebo-controlled, parallel-group, single and multiple ascending dose (SAD and MAD) study comprising nine cohorts (**Table 1**)
 - — Participants received TIP or placebo to match (3:1 ratio) under fasting conditions in both the SAD (cohorts 1–4 [75–1500 mg]) and MAD (cohorts 5–7 [75–900 mg QD], 8 [600 mg BID], and 9 [600 mg QD]) cohorts
- Cohort 9 consisted of participants homozygous for the *UGT1A1*28* allele, and was included to evaluate the potential additive effects of TIP on UGT1A1 inhibition and consequent hyperbilirubinemia in this population, who have a higher baseline risk of hyperbilirubinemia owing to reduced activity of UGT1A1
- Intensive pharmacokinetic sampling was conducted up to 120 hours post dose
- Safety and tolerability were assessed in both study parts

Table 1. Study Design

Cohort	Dose (mg)	Frequency[a]	N randomized
SAD study part			
1	75	Single dose	8
2	300	Single dose	8
3	900	Single dose	8
4	1500	Single dose	8
MAD study part			
5	75	QD for 10 days	8
6	300	QD for 10 days	8
7	900	QD for 10 days	8
8	600	BID for 10 days	8
9	600	QD for 10 days	9[b]

[a]All participants received TIP or placebo to match in a 3:1 ratio (TIP: 6, placebo: 2) under fasting conditions.
[b]One participant was enrolled and randomly assigned twice into this cohort under two different participant identification numbers, and was therefore included twice.
BID, twice daily; MAD, multiple ascending dose; QD, once daily; SAD, single ascending dose; TIP, tilpisertib fosmecarbil.

Analyses

- Plasma concentrations of the prodrug TIP and the parent drug GS-4875 were quantified using validated high-performance liquid chromatography-tandem mass spectrometry bioanalytical methods
- Dose proportionality assessment was conducted using a power model

Results

Participant Flow and Characteristics

- Of 73 participants randomly assigned, 71 (97.3%) received all doses of the study drug and 70 (95.9%) completed the study
- The median (range) participant age was 32 (18–48) years; 49.3% were women
- The mean (SD) body mass index was 25.9 (2.97) kg/m^2
- The majority of participants were white (53.4%) or black/African American (39.7%)

Pharmacokinetics of GS-4875

- Concentrations of TIP were not quantifiable in the majority of participants (72.9%), indicating rapid conversion to the active metabolite, GS-4875
- Following single oral doses of TIP (300–1500 mg), GS-4875 plasma exposure (C_{max} and AUC) increased in a less than dose-proportional manner (**Table 2**, **Figure 1**)
 - — Mean (90% CI) slopes (day 1; cohorts 2–4) were 0.65 (0.41–0.88) for AUC_{inf} and 0.53 (0.32–0.73) for C_{max}
- Following once-daily oral doses of TIP (75–900 mg), GS-4875 steady state plasma exposure increased in a less than dose-proportional manner (**Table 3**, **Figure 2**)
 - — Mean (90% CI) slopes (day 10; cohorts 5–7 and 9) were 0.72 (0.59–0.85) for AUC_{tau}, 0.64 (0.52–0.75) for C_{max}, and 0.86 (0.70–1.03) for C_{tau}

Table 2. Plasma Pharmacokinetic Parameters of GS-4875 Following Single Doses of TIP (SAD Cohorts)

Pharmacokinetic Parameter (Mean[a] [%CV])	Cohort 2 300 mg (N = 6)	Cohort 3 900 mg (N = 6)	Cohort 4 1500 mg (N = 6)
C_{max} (µg/mL)	1.50 (37.6)	3.09 (23.3)	3.36 (39.7)
T_{max} (h)[b]	4.00 (1.50, 4.00)	3.00 (2.00, 4.00)	3.00 (1.50, 6.00)
AUC_{last} (h·µg/mL)	26.0 (39.3)	56.1 (26.0)	72.2 (52.7)
AUC_{inf} (h·µg/mL)	27.1 (40.6)	58.8 (27.1)	78.0 (62.6)
$t_{1/2}$ (h)[b]	23.1 (12.8, 36.0)	28.9 (19.7, 30.4)	22.4 (16.4, 47.9)
AUC_{inf}/D (h·µg/mL/mg)[c]	0.0905 (40.6)	0.0654 (27.1)	0.0520 (62.6)
C_{max}/D (µg/mL/mg)[c]	0.005 (37.6)	0.0034 (23.3)	0.0022 (39.7)

[a]Means presented are unadjusted arithmetic means.
[b]Values are presented as median (minimum, maximum).
[c]Dose-normalized to TIP dose.
Pharmacokinetic parameters were estimated by noncompartmental analyses using Phoenix WinNonlin version 8.2. Cohort 1 pharmacokinetic data were deemed not evaluable and hence not included.
%CV, percentage coefficient of variation; AUC_{inf}, area under the concentration versus time curve from time zero to infinite time; AUC_{last}, area under the concentration versus time curve from time zero to the last quantifiable concentration; C_{max}, maximum observed concentration; D, dose; SAD, single ascending dose; $t_{1/2}$, terminal elimination half-life; TIP, tilpisertib fosmecarbil; T_{max}, time to maximum concentration.

Results (cont.)

Figure 1. Plasma GS-4875 Concentration Over Time Following Single Doses of TIP (SAD Cohorts 2–4)

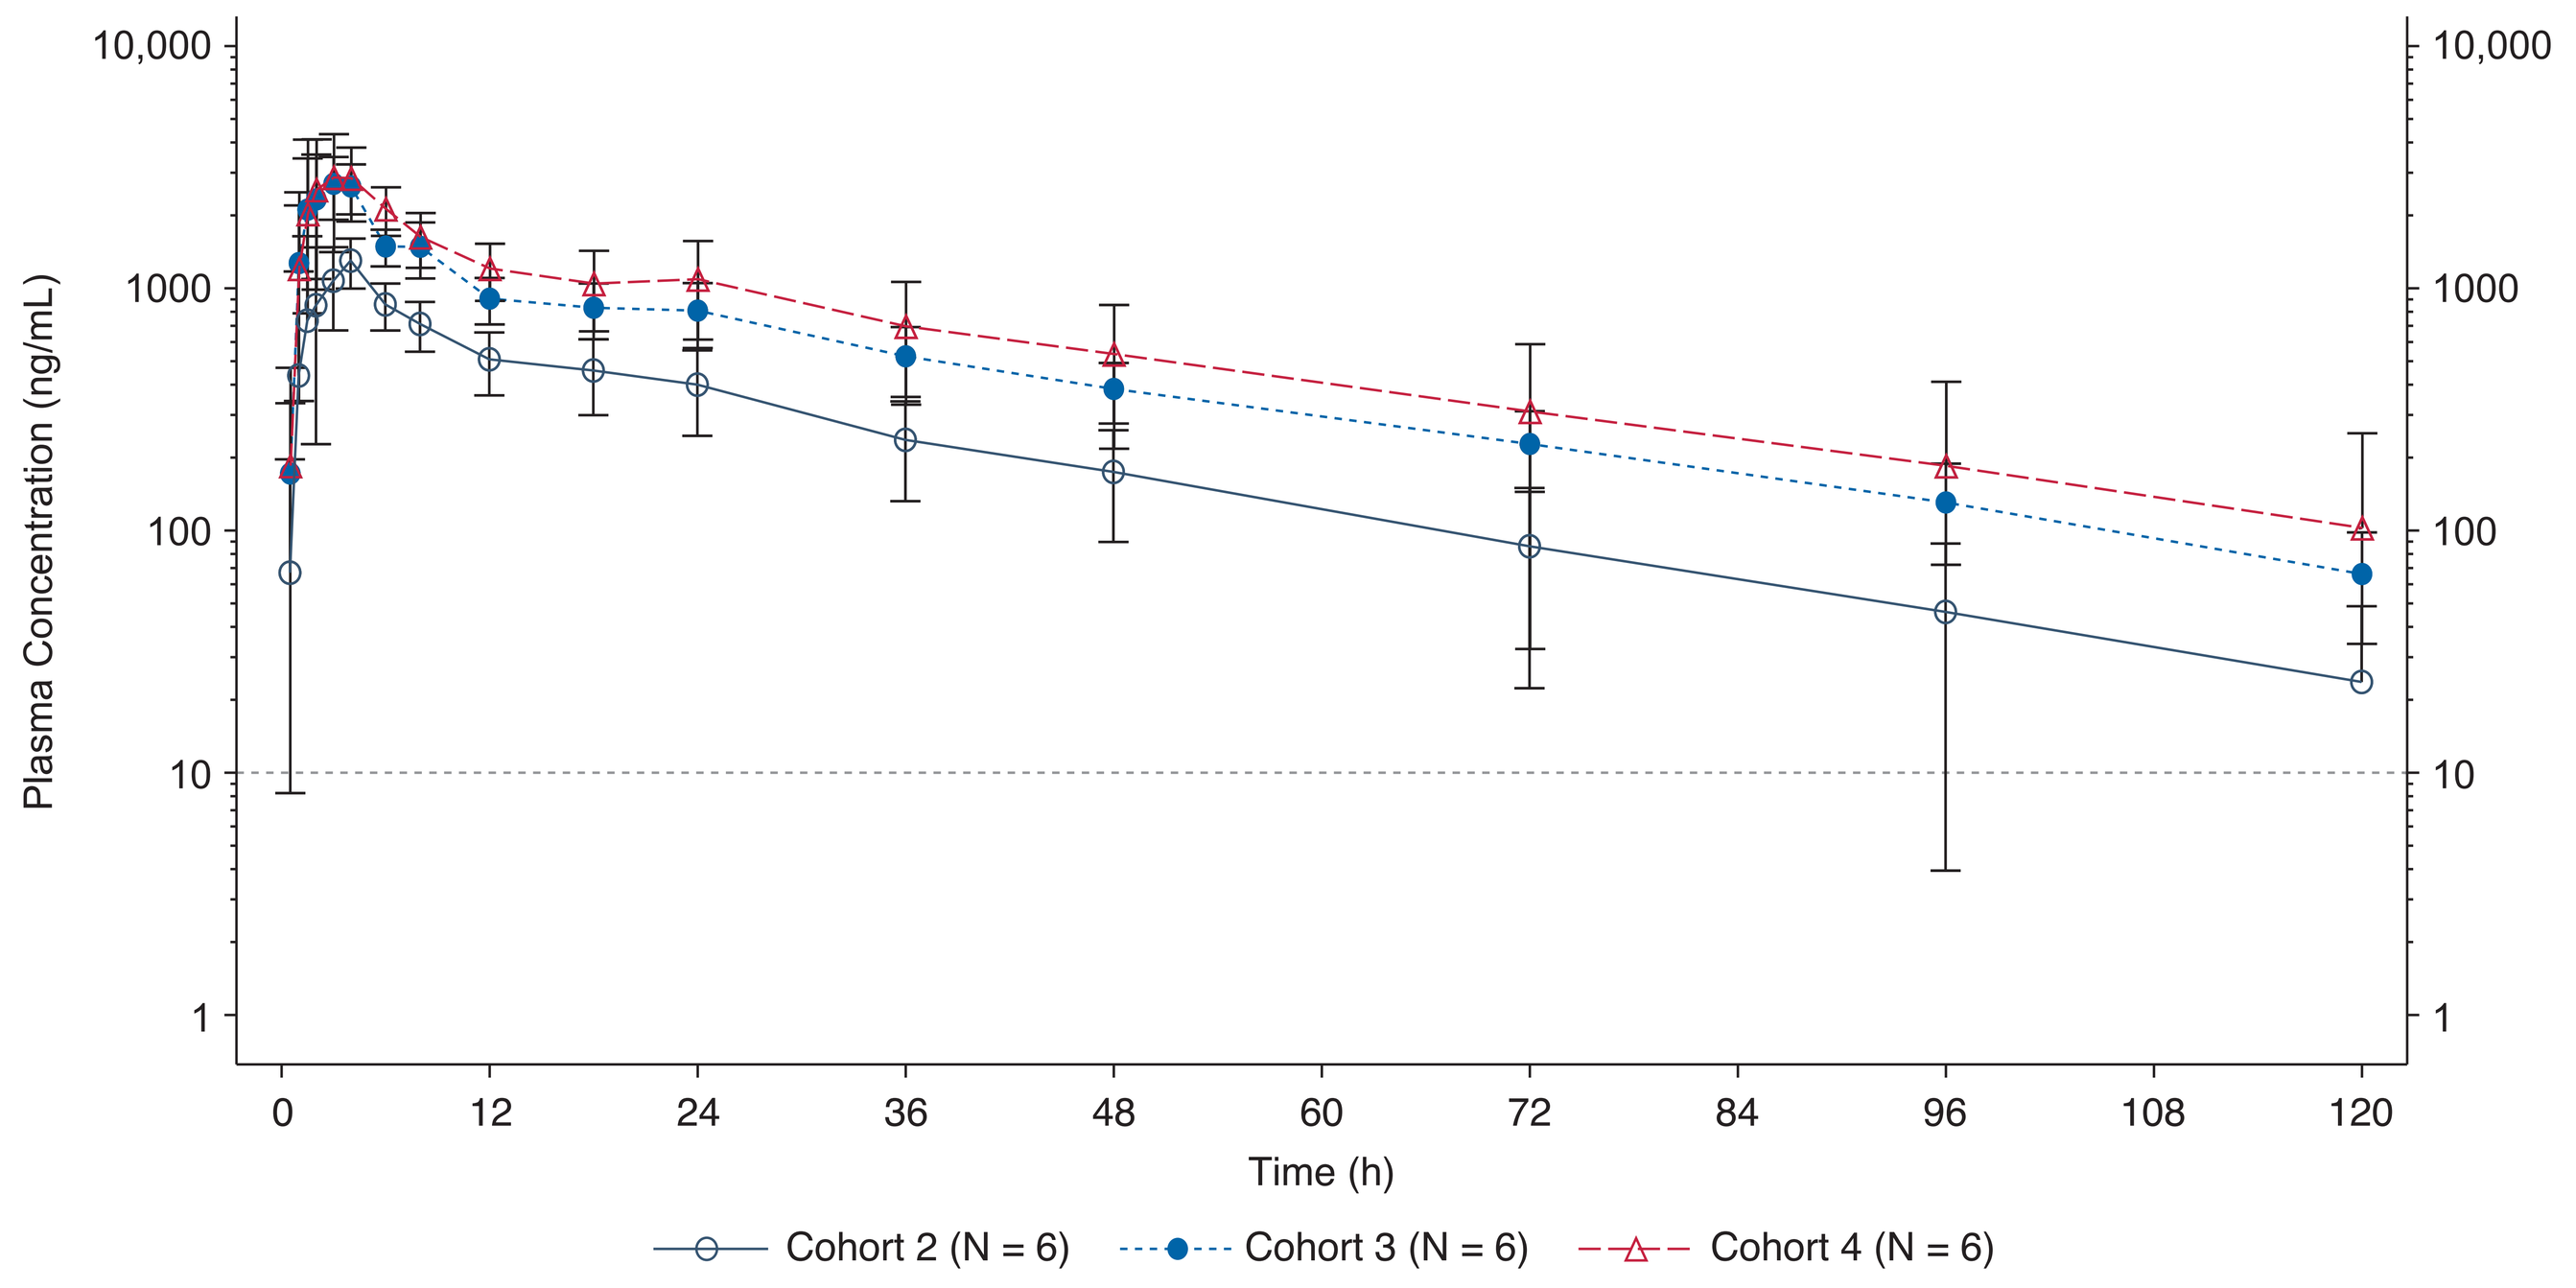
Plasma Concentration (ng/mL)
10,000
1000
100
10
1
Time (h)
0 12 24 36 48 60 72 84 96 108 120
Cohort 2 (N = 6)
Cohort 3 (N = 6)
Cohort 4 (N = 6)

Participants received a single dose of TIP under fasting conditions and blood sampling was performed up to 120 h post dose. The horizontal dashed line represents the lower limit of quantification. Data points are mean values. Error bars show standard deviation. Data are shown on a semi-log scale.
SAD, single ascending dose; TIP, tilpisertib fosmecarbil.

Figure 2. Plasma GS-4875 Concentration Over Time Following Multiple Doses of TIP (MAD Cohorts 5–9)

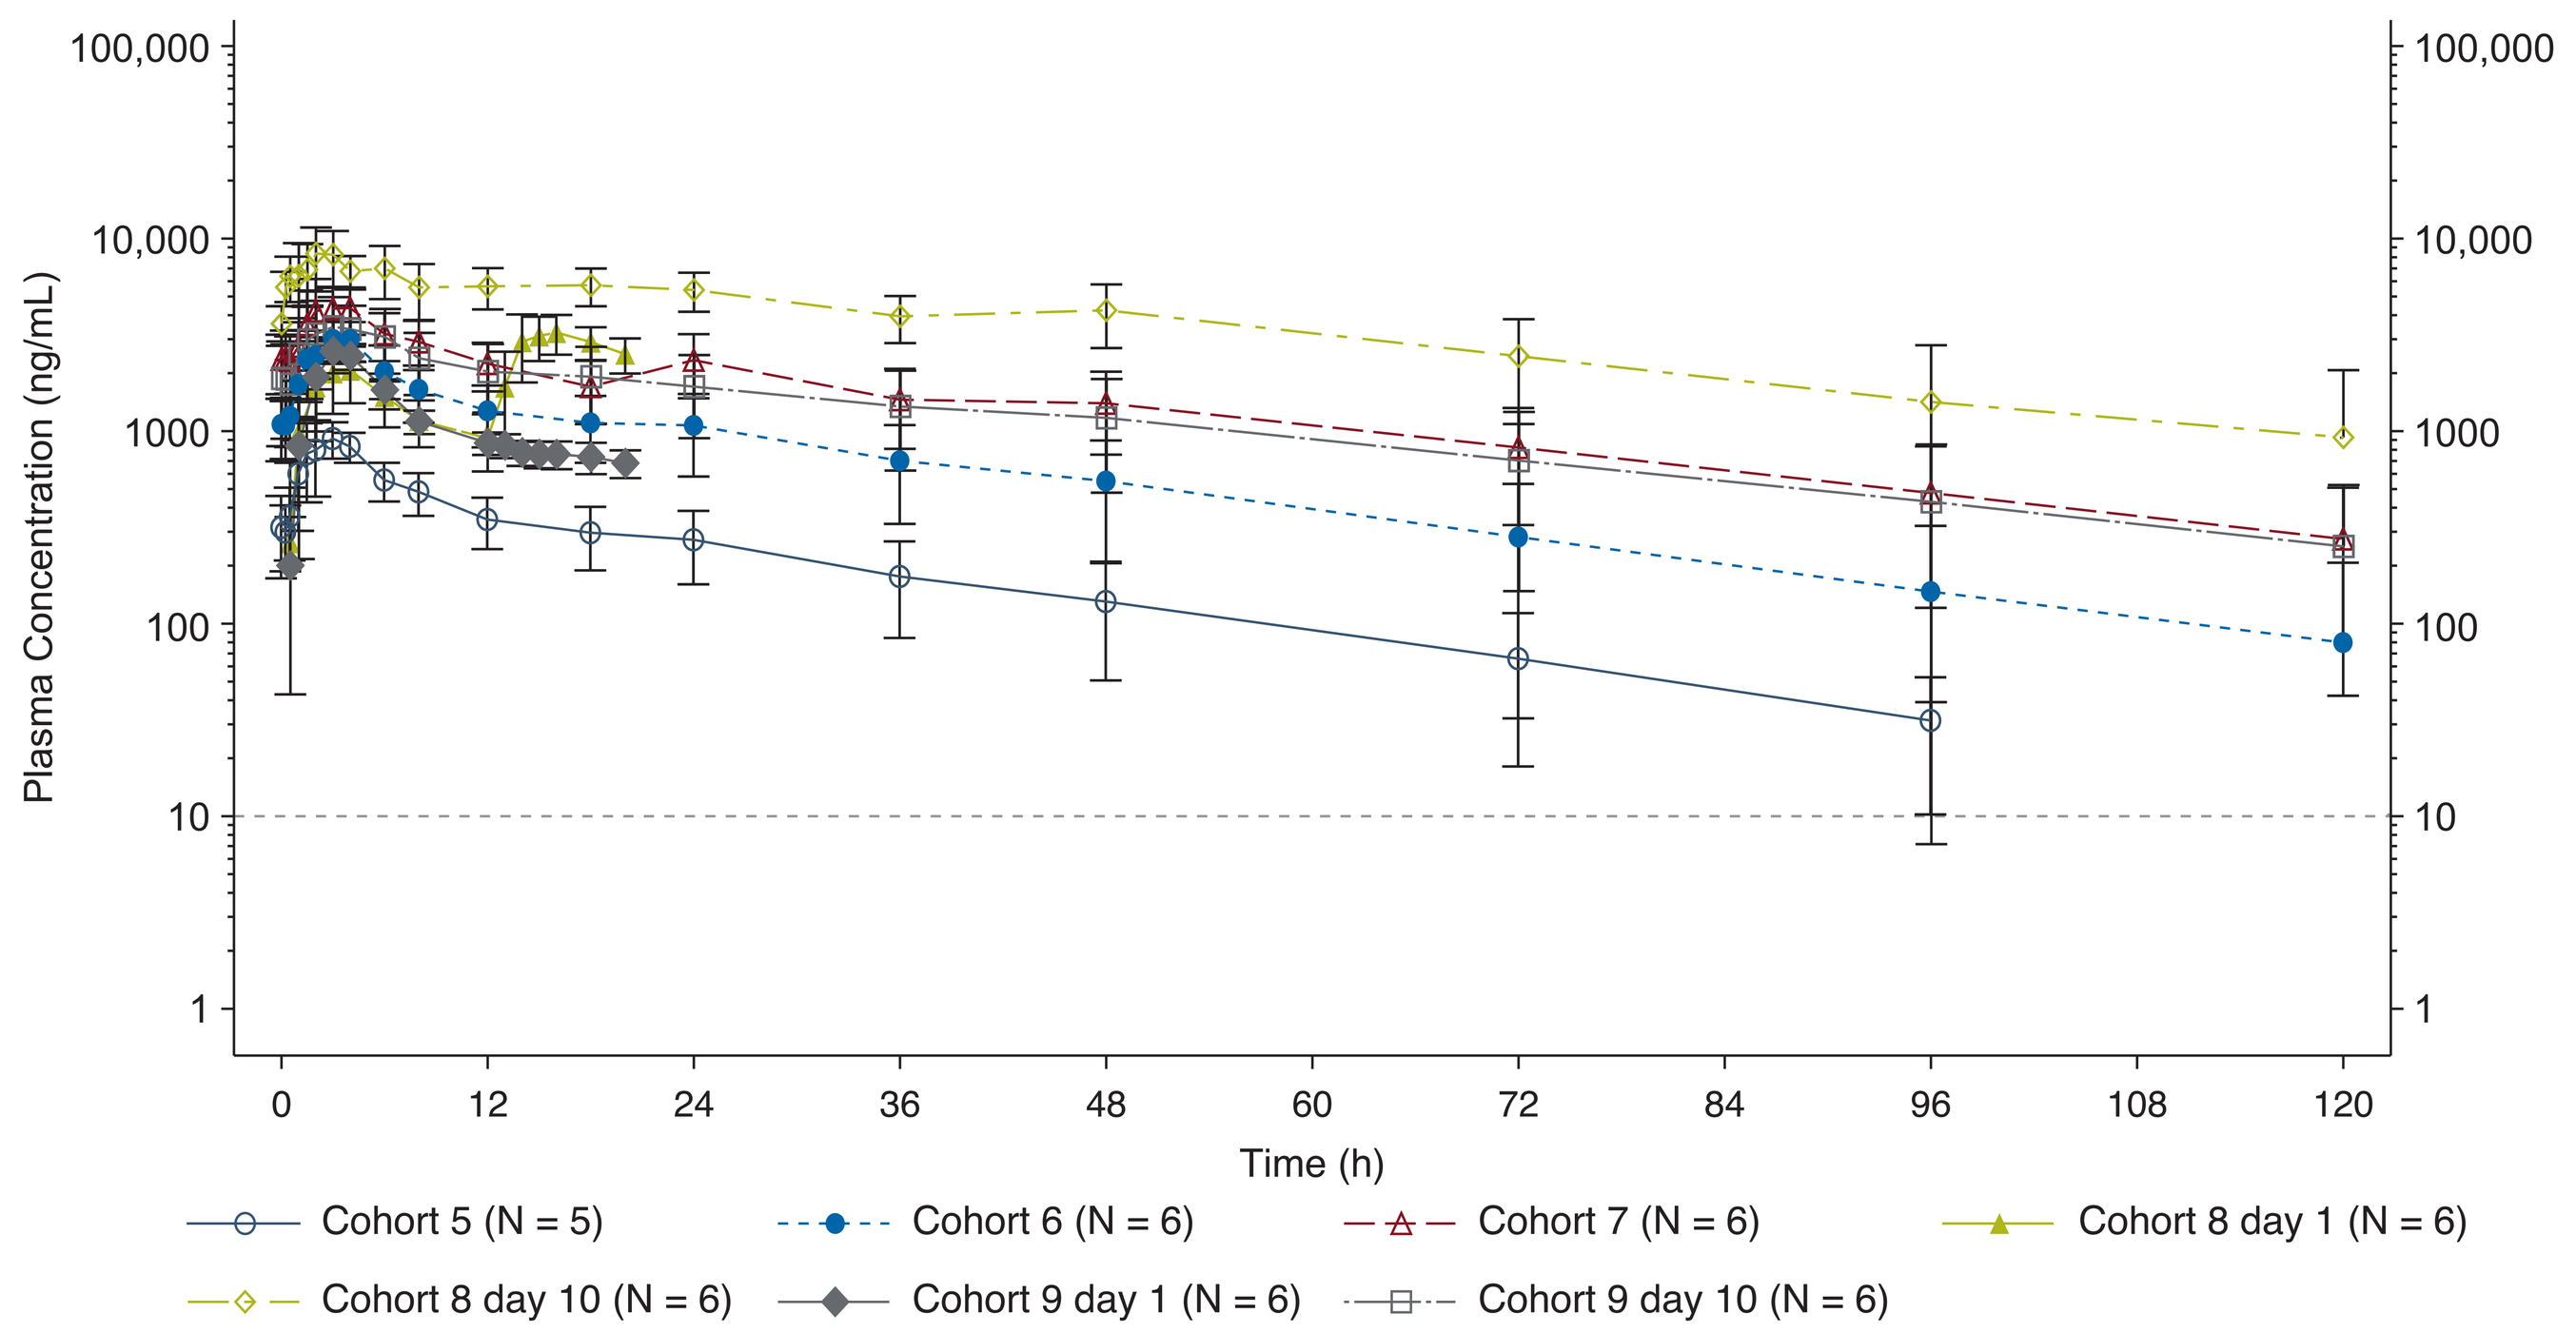
Plasma Concentration (ng/mL)
100,000
10,000
1000
100
10
1
Time (h)
0 12 24 36 48 60 72 84 96 108 120
Cohort 5 (N = 5)
Cohort 6 (N = 6)
Cohort 7 (N = 6)
Cohort 8 day 1 (N = 6)
Cohort 8 day 10 (N = 6)
Cohort 9 day 1 (N = 6)
Cohort 9 day 10 (N = 6)

Participants received TIP (QD or BID) over 10 days under fasting conditions. Blood sampling was performed up to 120 h after the last dose on day 1 (cohorts 8 and 9) or day 10 (cohorts 5–9). The horizontal dashed line represents the lower limit of quantification. Data points are mean values. Error bars show standard deviation. Data are shown on a semi-log scale.
BID, twice daily; MAD, multiple ascending dose; QD, once daily; TIP, tilpisertib fosmecarbil.

Table 3. Plasma Pharmacokinetic Parameters of GS-4875 Following Multiple Doses of TIP (MAD cohorts)

GS-4875 Pharmacokinetic Parameter (Mean[a] [%CV])	Cohort 5 75 mg QD (N = 5)	Cohort 6 300 mg QD (N = 6)	Cohort 7 900 mg QD (N = 6)	Cohort 8 600 mg BID (N = 6)	Cohort 9 *UGT1A1 *28/*28* 600 mg QD (N = 6)
Day 1					
C_{max} (µg/mL)[b]	–	–	–	2.48 (20.4)	2.76 (18.2)
T_{max} (h)[c]	–	–	–	2.50 (1.12, 4.00)	3.00 (2.00, 4.00)
AUC_{tau} (h·µg/mL)[b]	–	–	–	15.9 (18.9)	25.6 (15.2)
C_{tau} (µg/mL)[b]	–	–	–	0.972 (33.3)	0.507 (34.9)
Day 10					
C_{max} (µg/mL)[b]	0.980 (27.6)	3.17 (22.6)	4.96 (32.0)	8.64 (33.0)	3.82 (33.0)
T_{max} (h)[c]	3.00 (2.00, 3.10)	4.00 (1.50, 4.00)	3.01 (2.00, 4.00)	2.00 (2.00, 6.00)	3.00 (2.00, 6.00)
AUC_{tau} (h·µg/mL)[b]	10.5 (27.2)	37.4 (30.7)	62.2 (29.6)	77.5 (29.1)	55.1 (39.4)
C_{tau} (µg/mL)[b]	0.273 (41.3)	1.07 (45.7)	2.33 (36.8)	5.65 (24.1)	1.70 (46.0)
$t_{1/2}$ (h)[c]	21.8 (20.3, 31.8)	19.0 (13.9, 40.0)	28.5 (20.1, 39.1)	21.8 (13.0, 73.4)	28.4 (20.3, 43.7)
AUC_{tau}/D (h·µg/mL/mg)[b,d]	0.141 (27.2)	0.125 (30.7)	0.0691 (29.6)	0.129 (29.1)	0.0918 (39.4)
C_{max}/D (µg/mL/mg)[b,d]	0.0131 (27.6)	0.0106 (22.6)	0.0055 (32.0)	0.0144 (33.0)	0.0064 (33.0)

[a]Means presented are unadjusted arithmetic means.
[b]Values are for the morning dose in cohort 8.
[c]Values are presented as median (minimum, maximum).
[d]Dose-normalized to TIP dose.
Pharmacokinetic parameters were estimated by noncompartmental analyses using Phoenix WinNonlin version 8.2.
%CV, percentage coefficient of variation; AUC_{tau}, area under the concentration versus time curve over the dosing interval; BID, twice daily; C_{max}, maximum observed concentration; C_{tau}, observed drug concentration at the end of the dosing interval; D, dose; MAD, multiple ascending dose; QD, once daily; $t_{1/2}$, terminal elimination half-life; TIP, tilpisertib fosmecarbil; T_{max}, time to maximum concentration; UGT1A1, uridine diphosphate glucuronosyltransferase 1A1.

- Maximum plasma concentrations (median T_{max}) of GS-4875 were achieved 2.0–4.0 hours following TIP administration
- GS-4875 showed low-to-moderate interindividual variability (15–63 %CV) and a median half-life ($t_{1/2}$) of 19.0–28.9 hours across the SAD and MAD cohorts
- Consistent with the GS-4875 elimination $t_{1/2}$, GS-4875 exposure accumulated approximately 2-fold and 5-fold at steady state following once or twice daily dosing of TIP 600 mg, respectively
 - — Steady state was achieved by day 7 in all MAD cohorts

Safety

- Thirteen (24.1%) of the 54 participants who received TIP (across all cohorts) and five (26.3%) of the 19 participants who received placebo experienced at least one adverse event (AE)
- The most common AEs in participants receiving TIP (reported in at least two participants) were SARS-CoV-2 test positive (n = 4; 7.4%), and dizziness, dysgeusia, and nausea (each n = 2; 3.7%)
 - — Of these AEs, only SARS-CoV-2 test positive was also reported in participants receiving placebo (n = 1, 5.3%)
- Six participants (8.2%) experienced AEs deemed related to treatment, the most common being dysgeusia (TIP 600 mg BID, n = 2), headache (TIP 600 mg QD, n = 1; MAD pooled placebo, n = 1), and nausea (TIP 600 mg BID, n = 1; TIP 600 mg QD, n = 1)
- All AEs were mild (grade 1) in severity and no serious adverse events or deaths were reported
- No dose-limiting toxicities, or clinically relevant electrocardiogram or vital sign abnormalities were reported
- Three participants homozygous for the *UGT1A1*28* allele (cohort 9) had grade 3 laboratory abnormalities of blood bilirubin increase that were primarily mediated by rises in indirect bilirubin, but all events decreased to grade 0/grade 1 level by day 15
 - — No jaundice, icterus, or elevations of key liver enzymes were reported in any cohort 9 participants

References: **1.** Khor B, *et al. Nature.* 2011;474:307–17. **2.** Gantke T, *et al. Cell Res.* 2011;21:131–45.

Acknowledgments: Medical writing support was provided by Jennifer Hung PhD and Michael Molloy-Bland PhD of Oxford PharmaGenesis, Melbourne, Australia, and funded by Gilead Sciences, Inc. in accordance with Good Publication Practice 2022 (GPP2022) guidelines (https://www.ismpp.org/gpp-2022).
Disclosures: **XQ**, **AQ**, **FH**, **PM**, and **IC** are employees of, and may own stock in, Gilead Sciences, Inc. **EW, MM,** and **AO** were employed by Gilead at the time the study was conducted, and may own stock in Gilead Sciences, Inc.

Presented at the 2024 American College of Clinical Pharmacology (ACCP) Annual Meeting, September 8–10, 2024, Bethesda, MD. This poster is intended for healthcare professionals only.