Concentration-QTc interim analysis of I-DXd in subjects with advanced solid tumors from study IDeate-PanTumor01 (DS7300-A-J101)

P092

Brittany P. Tran,[1] Bill Poland,[2] Narasimha M. Midde,[1] Naoko Okamoto,[1] Jasmeet Singh,[1] Meng Qian,[1] Naoto Yoshizuka,[1] Soniya S. Vaidya,[1] Yvonne Lau[1]

[1]Daiichi Sankyo, Inc., Basking Ridge, NJ, USA; [2]Certara Inc., Princeton, NJ, USA

Copies of this poster obtained through this QR code and/or https://bit.ly/DSACCP2024 are for personal use only and may not be reproduced without permission of the authors.

PURPOSE

The objectives of this interim C-QTc analysis were to:

- Characterize the relationship between change from baseline QTc (ΔQTc) with: I-DXd plasma concentration; and DXd plasma concentration
- Predict concentration-related QTc prolongation (if any) with associated CI at the therapeutic doses of interest, 8 and 12 mg/kg

CONCLUSIONS

- 90% CI upper bounds of predicted mean ΔQTcF (Fridericia's correction) and mean ΔQTcP (population-specific correction) did not cross the 10 ms threshold and were negative for all doses, supporting a lack of a clinically meaningful effect on QTc interval for I-DXd and DXd in this population
- No demographic, electrolyte, or concomitant medication covariates had a significant relationship with QTc in this analysis
- Based on currently available clinical safety data and this interim C-QTc analysis, there are no findings that raise concerns of QT prolongation with I-DXd

INTRODUCTION

- I-DXd is a B7-H3–directed ADC that leverages clinically validated deruxtecan technology, with a plasma-stable linker and potent topoisomerase I inhibitor payload (a derivative of exatecan)[1,2]
- I-DXd is being evaluated in the ongoing Phase 1/2 dose-escalation and -expansion study IDeate-PanTumor01 (NCT04145622) in patients with advanced solid malignant tumors[3] (**Figure 1**)
- In vitro studies have shown that DXd did not inhibit the hERG channel current at concentrations up to 10 µmol/L (approximately 5000 ng/mL) in hERG-transfected Chinese hamster ovary cells[4]

Figure 1: Ongoing IDeate-PanTumor01 (DS7300-A-J101) study

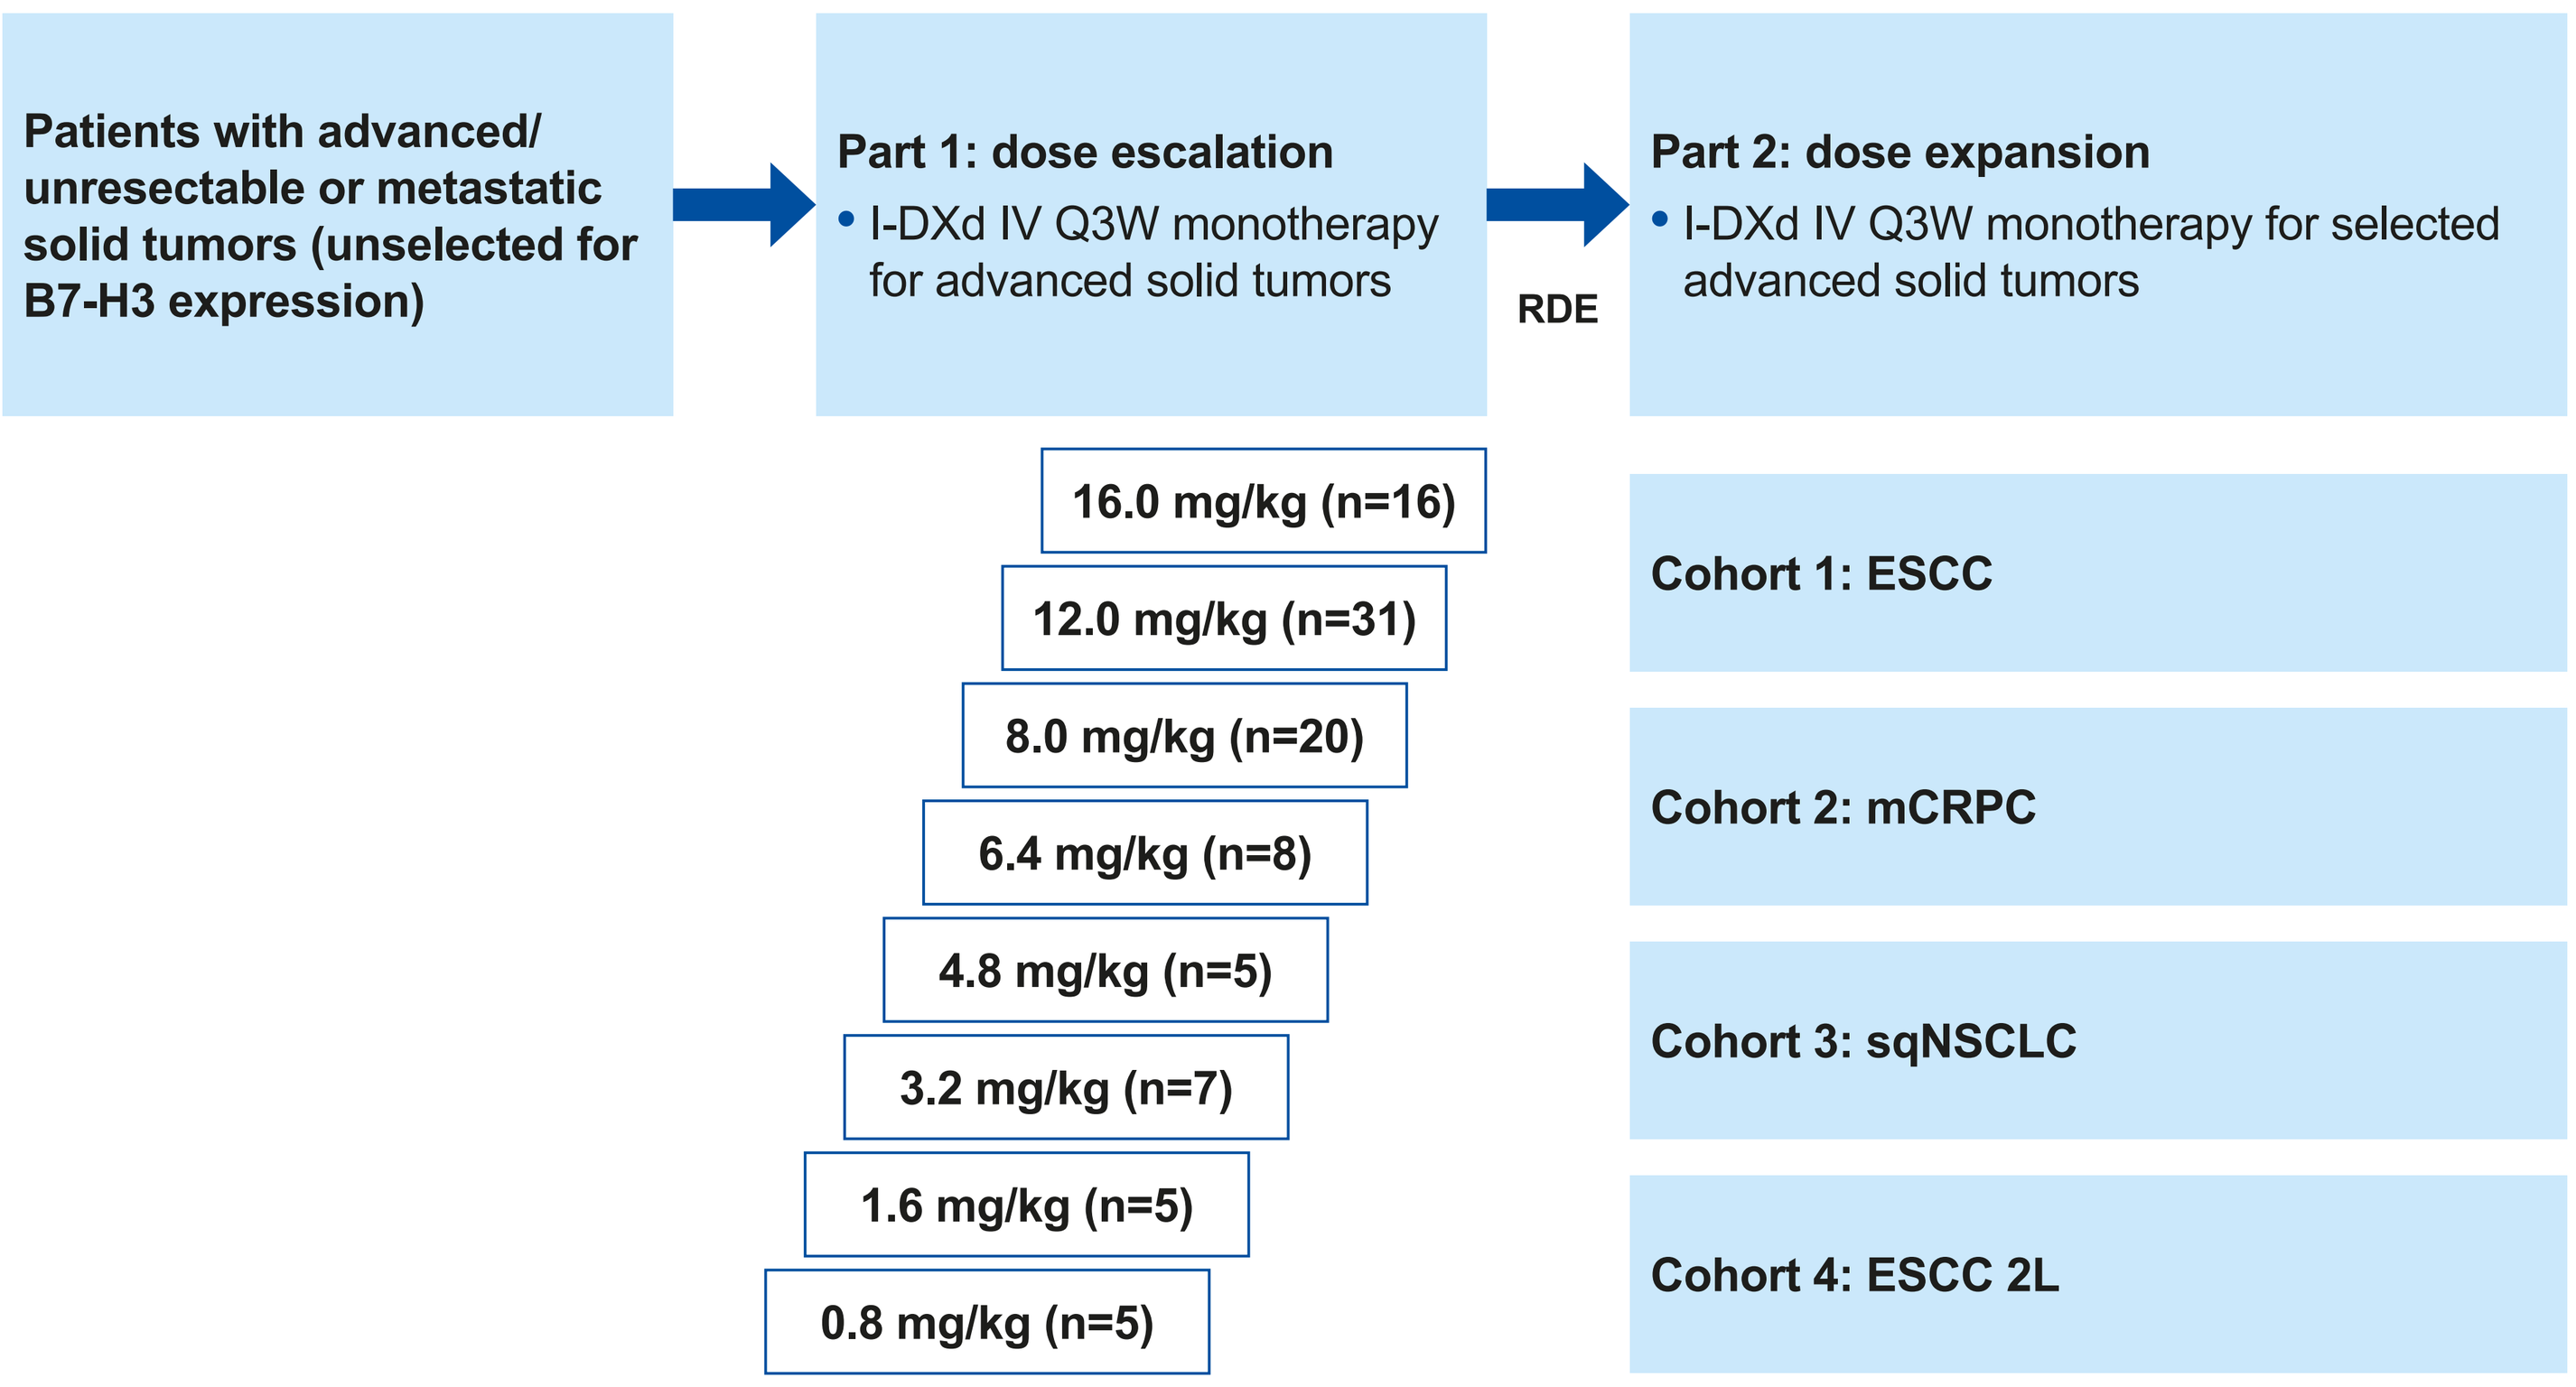
Patients with advanced/ unresectable or metastatic solid tumors (unselected for B7-H3 expression)
Part 1: dose escalation
• I-DXd IV Q3W monotherapy for advanced solid tumors
RDE
Part 2: dose expansion
• I-DXd IV Q3W monotherapy for selected advanced solid tumors
16.0 mg/kg (n=16)
12.0 mg/kg (n=31)
8.0 mg/kg (n=20)
6.4 mg/kg (n=8)
4.8 mg/kg (n=5)
3.2 mg/kg (n=7)
1.6 mg/kg (n=5)
0.8 mg/kg (n=5)
Cohort 1: ESCC
Cohort 2: mCRPC
Cohort 3: sqNSCLC
Cohort 4: ESCC 2L

154 subjects from Part 1 and Part 2 of the ongoing IDeate-PanTumor01 study were included in this analysis, out of a planned total of approximately 250 subjects in this study. All subjects in Part 1 were analyzed except for 1 subject at each of 12 and 16 mg/kg. In addition, 59 subjects dosed at I-DXd 12 mg/kg in Part 2 were analyzed (24 in Cohort 1, 30 in Cohort 2, and 5 in Cohort 3).

METHODS

- Triplicate 12-lead ECGs and time-matched PK data across a dose range of 0.8 to 16 mg/kg IV Q3W were analyzed as of January 26, 2023
- A prespecified linear mixed-effects model for ΔQTc (ΔQTcF or ΔQTcP) was used. The intercept and slope were modeled as population mean values with additive random between-subject variability. A term for baseline QTc effect by subject was included
- Model development began by confirming the significance of the baseline effect and the random effect on slope. Potential covariates, including demographic, electrolyte, and concomitant medications with known or potential QT prolongation risk were tested as intercept effects in the model. Stepwise forward selection–backward elimination (P<0.01 for both forward and backward) search strategy was used
- The final models were used to predict ΔQTc and the associated 90% CI versus concentration, particularly at the geometric mean of C_{max} at each dose, for comparison with a 10-ms threshold

$$\boxed{\Delta QTcF_{it}} = \underbrace{(\theta_{int} + \eta_{int,i})}_{\text{Intercept}} + \underbrace{(\theta_{slp} + \eta_{slp,i})}_{\text{Slope}} \boxed{Conc_{it}} + \boxed{\theta_{bl}} \underbrace{(QTcF_{i0} - QTcF_{0})}_{\text{(Baseline QTcF in subject } i\text{) – (Mean } QTcF_{i0} \text{ over all subjects)}} + \boxed{\varepsilon_{it}}$$

Observed change from baseline (Day 1 predose) QTcF in subject *i* at time *t*

Observed concentration in subject *i* at time *t*

Fixed effect for baseline

Residual error

RESULTS

Table 1: Baseline characteristics in analysis dataset

Characteristic		Overall (N=154)
Sex (%)	Male	81.2
	Female	18.8
Race (%)	White	53.9
	Asian	36.4
	Black	5.2
	Other	4.5
Age (years)	Mean (SD)	65.1 (9.12)
Weight (kg)	Mean (SD)	75.6 (19.1)
Calcium (mmol/L)	Mean (SD)	2.29 (0.15)
Potassium (mmol/L)	Mean (SD)	4.09 (0.36)
Concomitant medication (%)	Potential QT prolongation risk	36.8
	Known QT prolongation risk	3.8
QTcF (ms)	Mean (SD)	412 (20.9)
QTcP (ms)	Mean (SD)	418 (20.7)

Figure 2: Baseline QT, QTcF, and QTcP versus RR interval

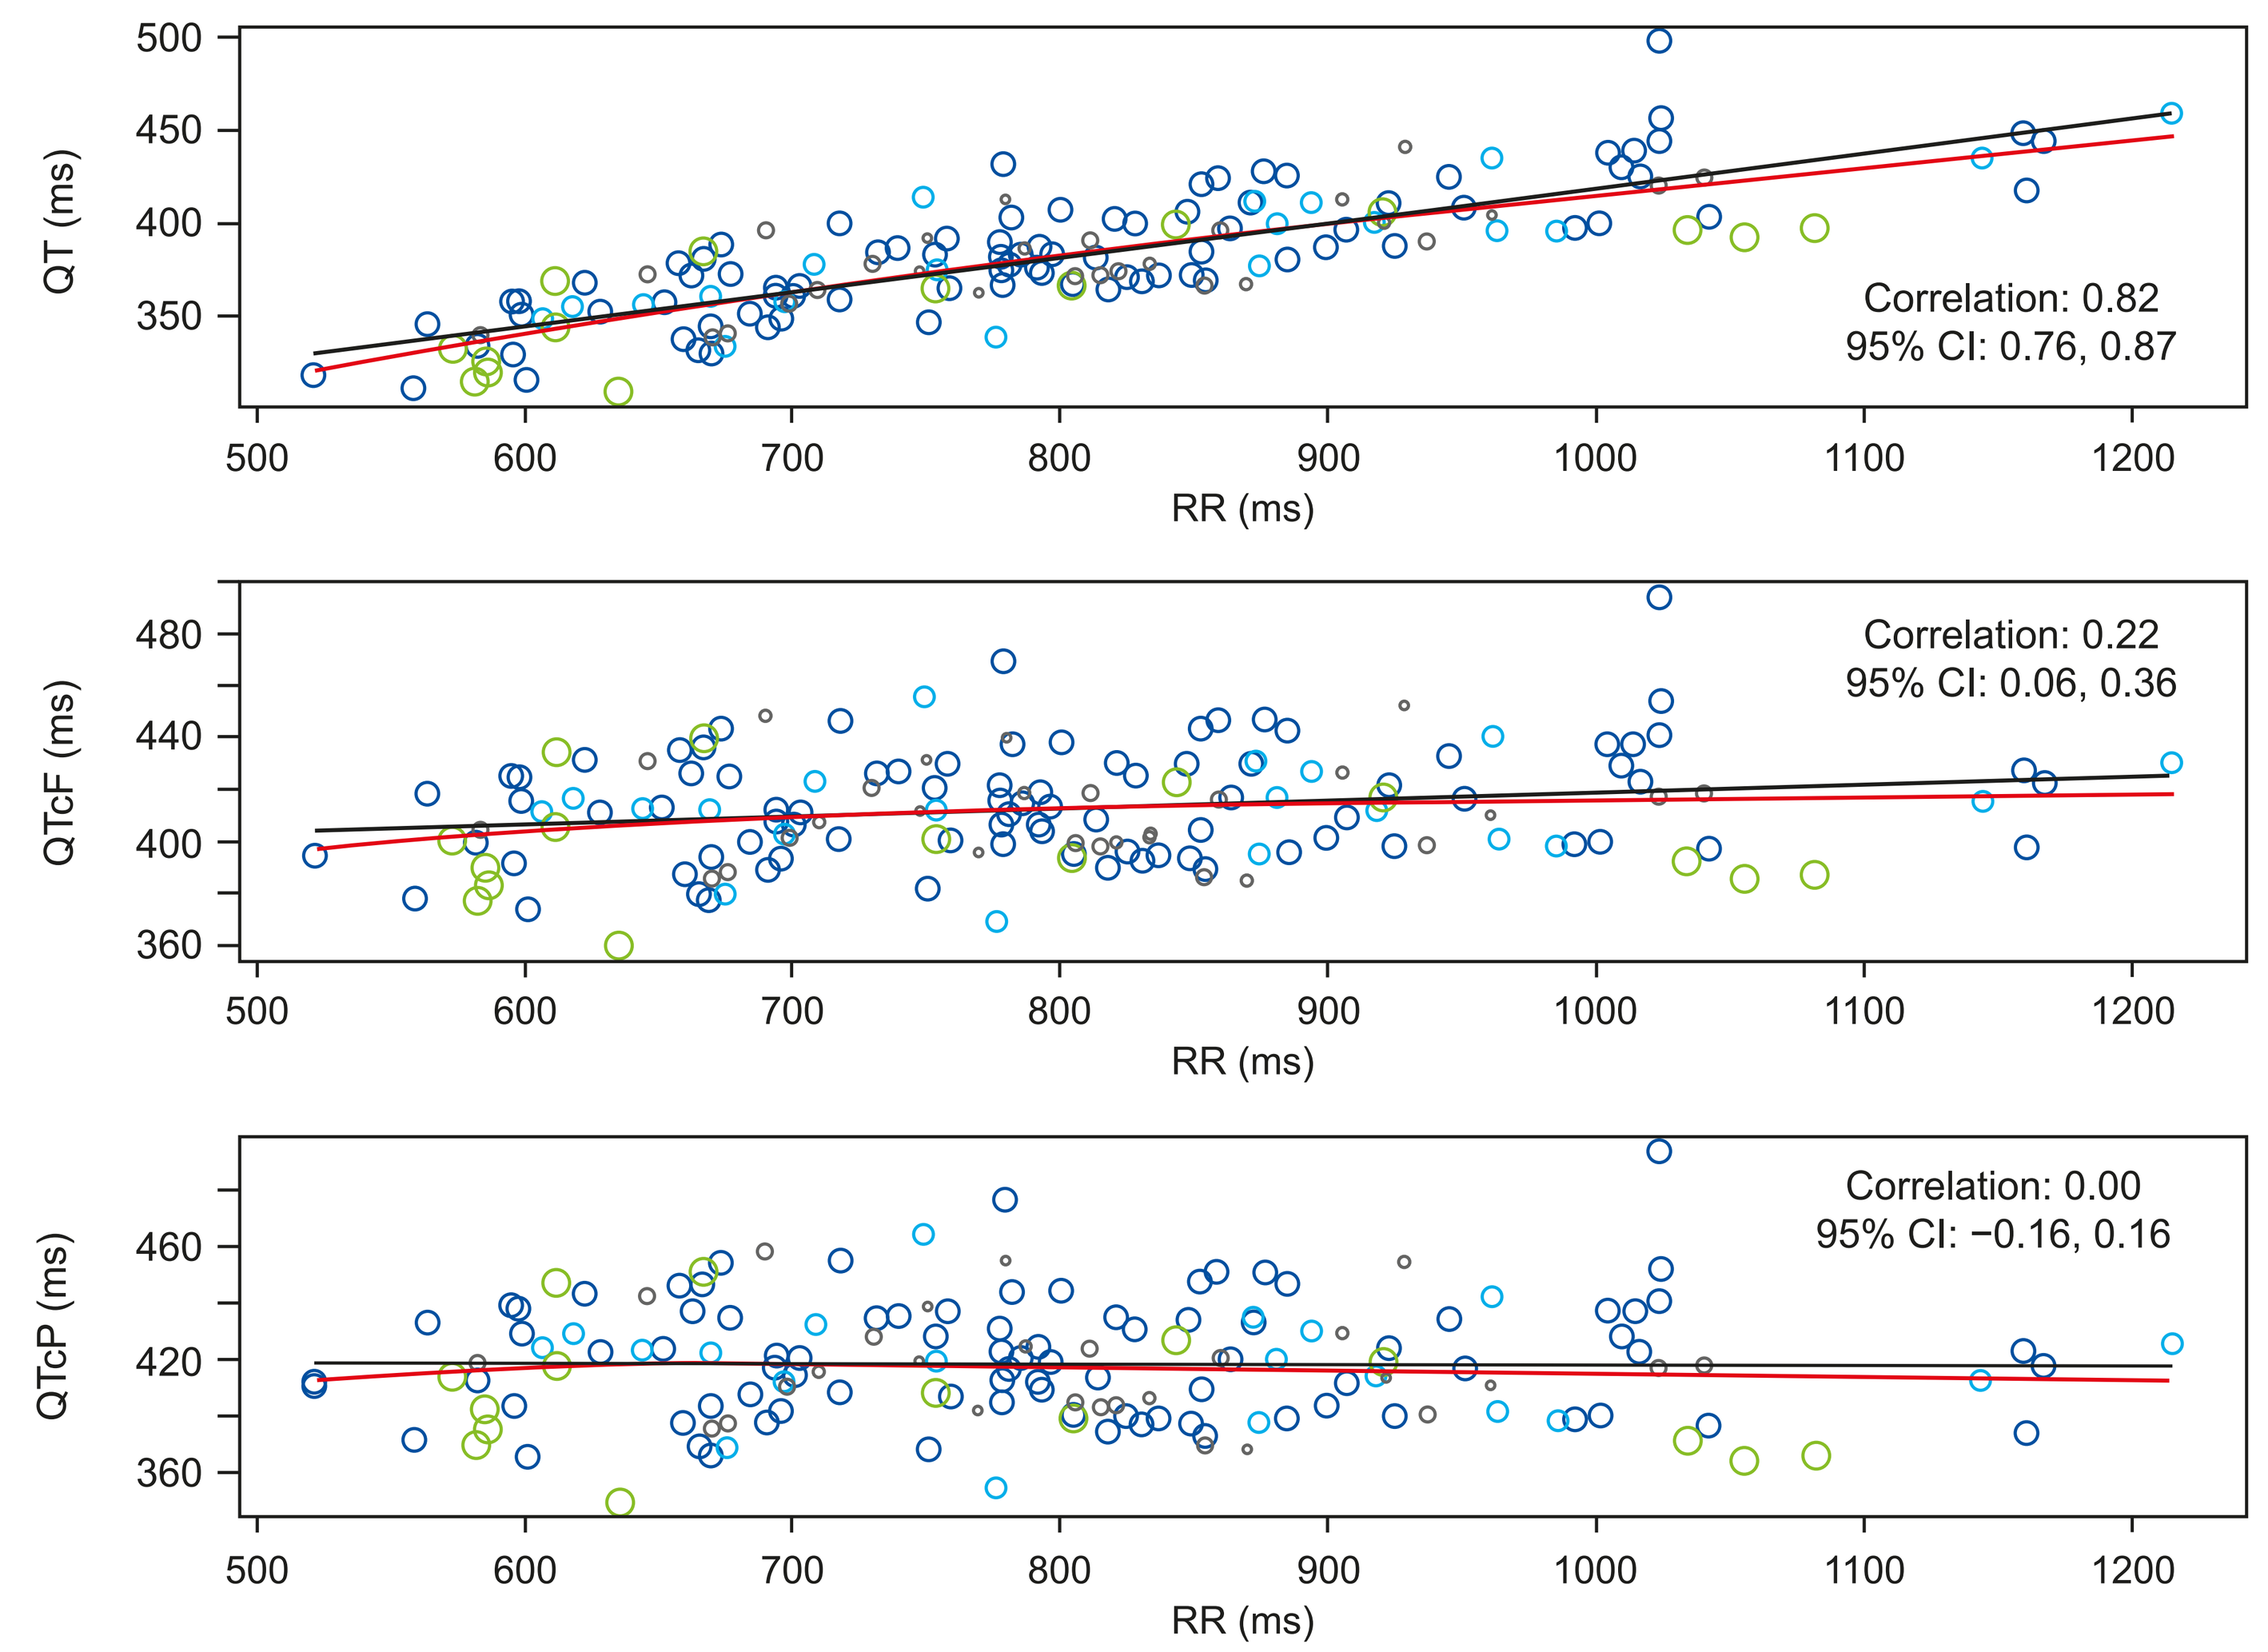
QT (ms)
RR (ms)
Correlation: 0.82
95% CI: 0.76, 0.87
QTcF (ms)
RR (ms)
Correlation: 0.22
95% CI: 0.06, 0.36
QTcP (ms)
RR (ms)
Correlation: 0.00
95% CI: −0.16, 0.16

Correlation: Pearson (bivariate) correlation coefficient. Red curves: locally estimated scatterplot smoothing; gray lines: linear regressions. Points (observations) are larger at higher doses: dark gray indicates <8 mg/kg; light blue, 8 mg/kg; dark blue, 12 mg/kg; green, 16 mg/kg.

- A population-specific correction was estimated to remove slope from the plot of QTc versus RR: QTcP = QT/RR^p (RR in seconds), where p was estimated at 0.393, as the Fridericia correction did not completely remove the slope (**Figure 2**)
- Base models for I-DXd and DXd were successfully estimated with a baseline QTcF effect and between-subject variability on the intercept and slope terms (with correlation). No covariate was statistically significant in the stepwise forward and backward elimination
- In all four models (ΔQTcF and ΔQTcP versus I-DXd and DXd concentration), the slope was negative, indicating no QT prolongation associated with either analyte. In the model of ΔQTcF versus DXd concentration, the slope was negative and statistically significant (P<0.05); however, the slope was not significant in the other three models
- Predictions of ΔQTcF and ΔQTcP from the final models at Cycle 3 GM C_{max} values at 8 and 12 mg/kg, estimated by non-compartmental analysis (**Table 2**), and as a function of concentration (**Figure 3**), showed that all predictions were negative

Table 2: Predicted ΔQTcF or ΔQTcP versus Cycle 3 C_{max} by dose

Dose (mg/kg)	Observed GM C_{max}	N	ΔQTcF Estimate (90% CI)	ΔQTcP Estimate (90% CI)
I-DXd				
8	197 µg/mL	13	−3.25 (−4.44, −2.06)	−3.28 (−4.48, −2.09)
12	304 µg/mL	60	−3.75 (−5.22, −2.29)	−3.71 (−5.18, −2.23)
DXd				
8	3.85 ng/mL	14	−3.06 (−4.25, −1.87)	−2.85 (−4.03, −1.67)
12	6.05 ng/mL	65	−4.00 (−5.43, −2.56)	−3.22 (−4.62, −1.82)

Figure 3: Concentration-ΔQTc relationships from final models

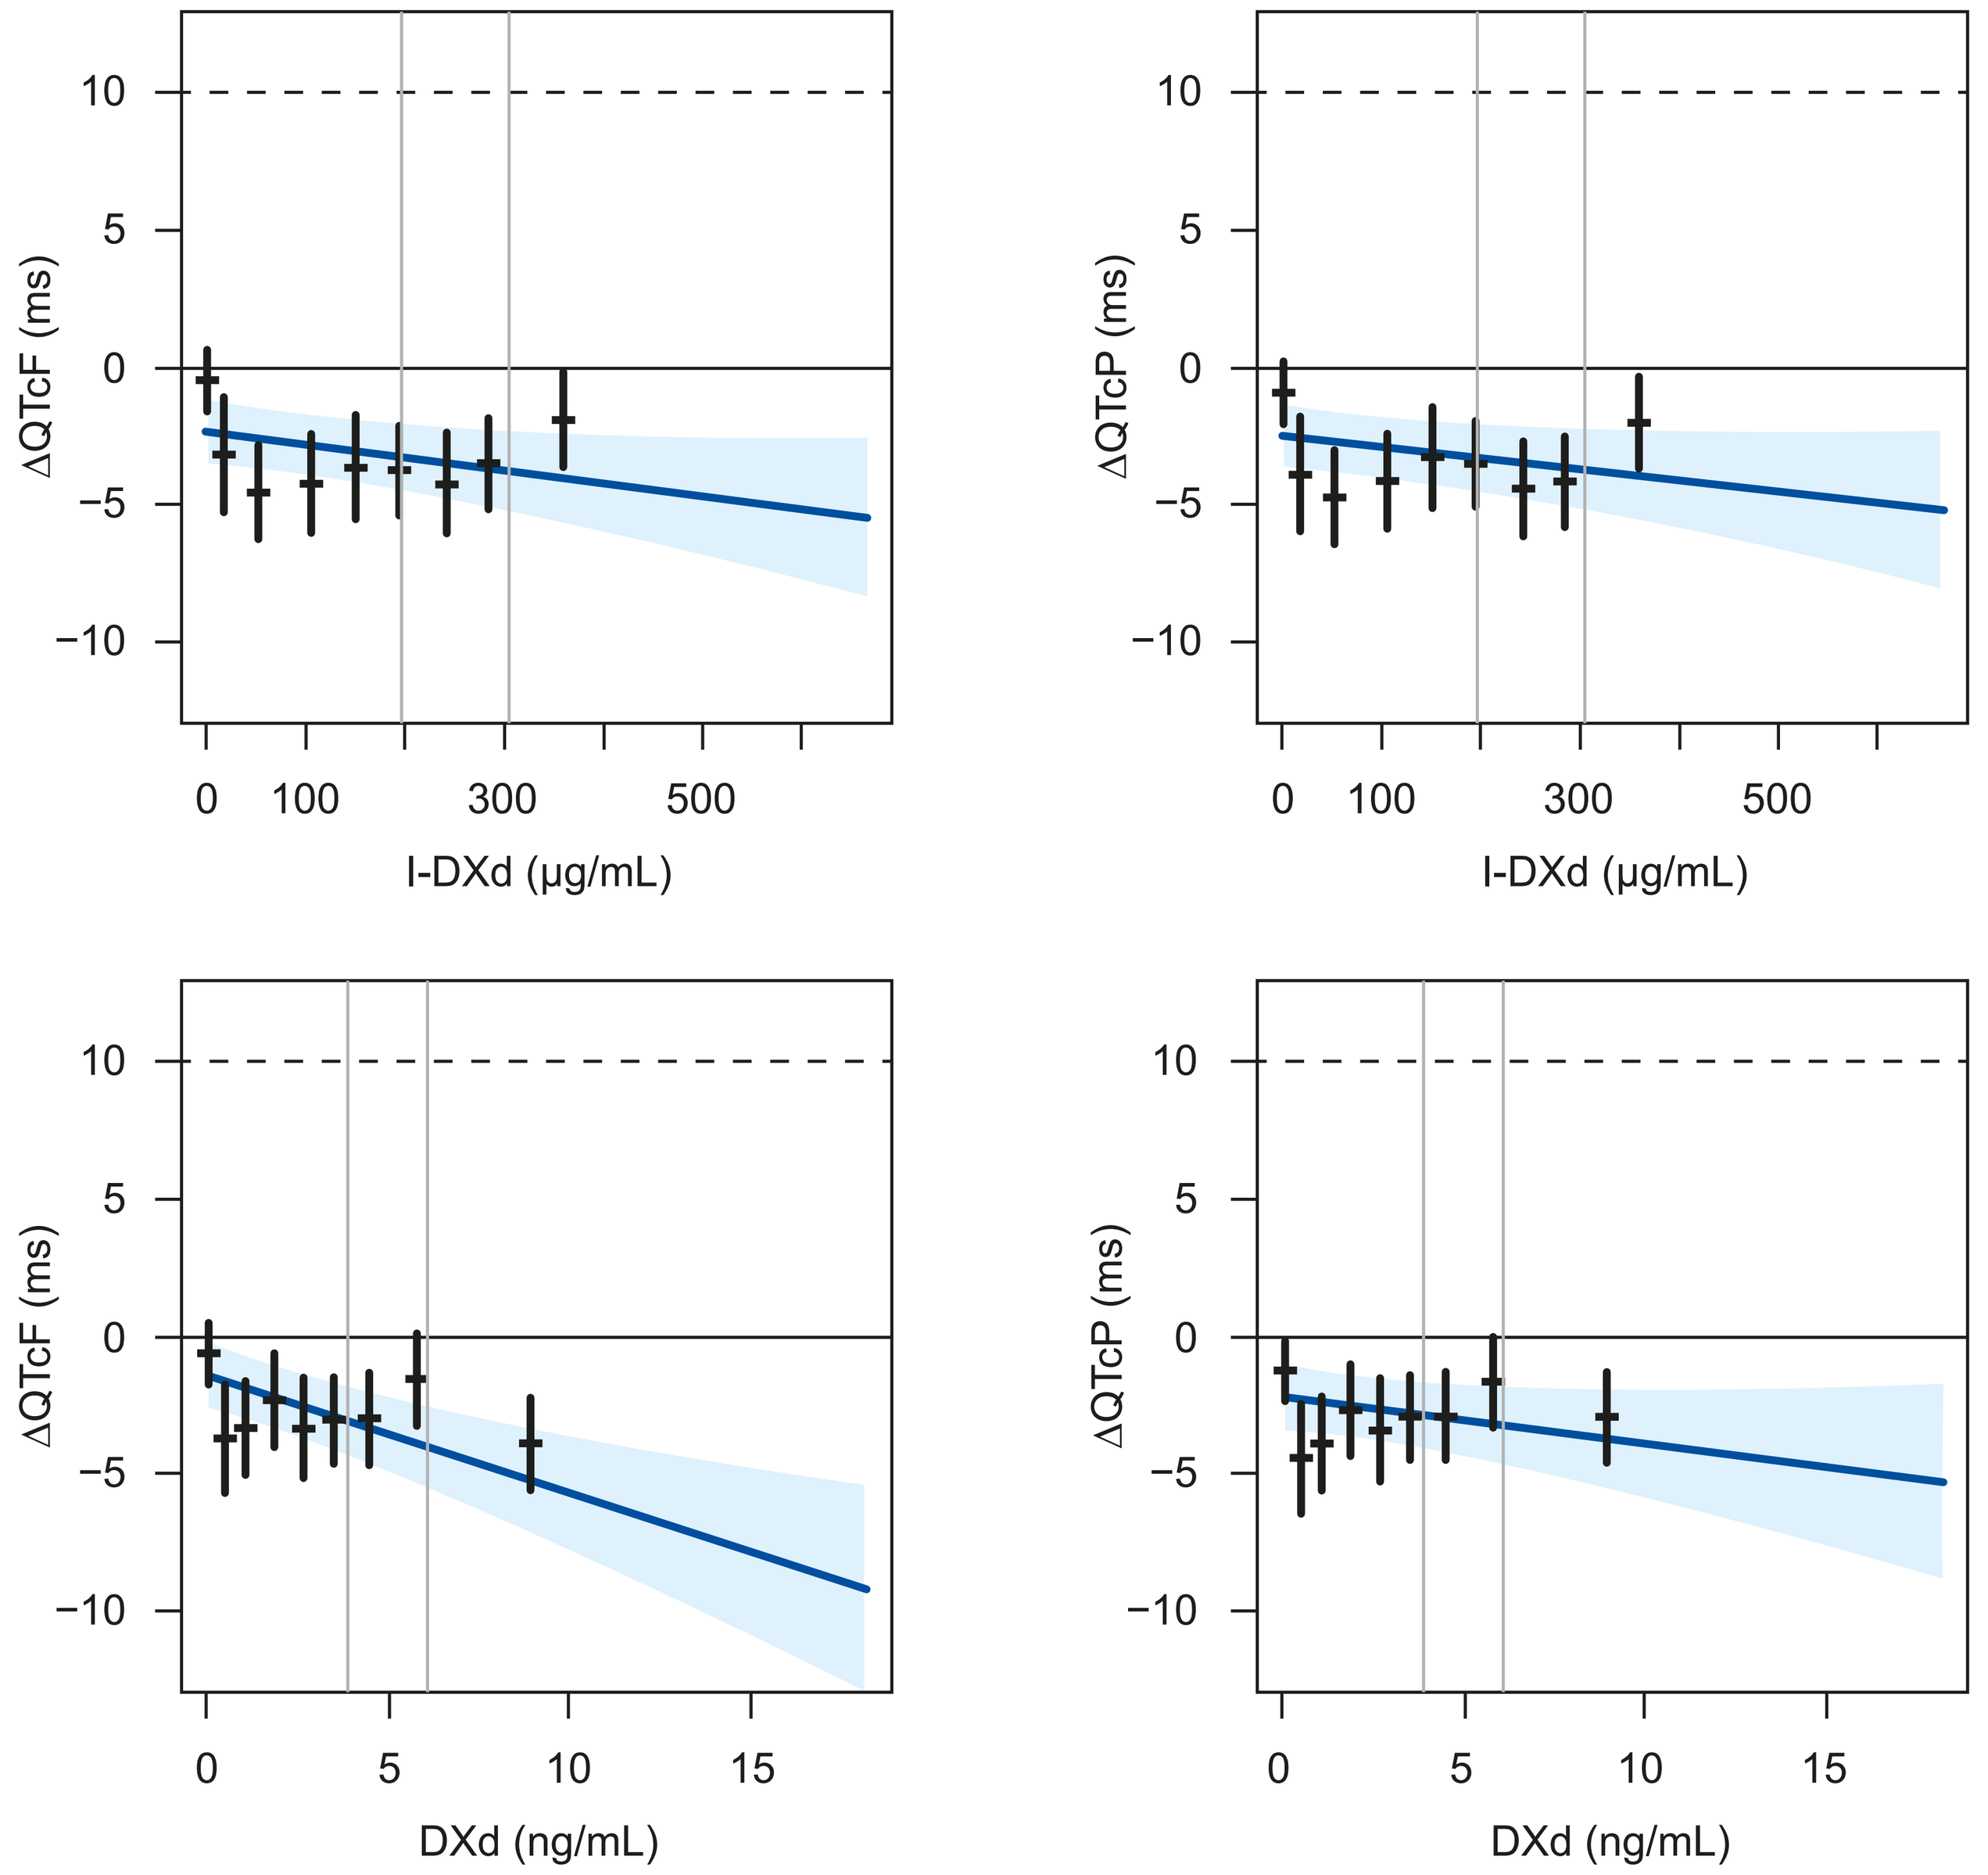
ΔQTcF (ms)
I-DXd (µg/mL)
ΔQTcP (ms)
I-DXd (µg/mL)
ΔQTcF (ms)
DXd (ng/mL)
ΔQTcP (ms)
DXd (ng/mL)

Center blue line: projected mean change in QTc (with covariates set to sample means); blue shaded area: its 2-sided 90% CI; black bars: mean and 90% CI of observed ΔQTc at the mean of each concentration decile; dotted horizontal lines: ΔQTc = 10 and 20 ms; grey vertical lines: geometric mean C_{max} at 8 mg/kg and 12 mg/kg.

ACKNOWLEDGMENTS

We thank the patients, their families, and their caregivers for their participation and study staff for their contributions.

DISCLOSURES

This study is sponsored by Daiichi Sankyo, Inc. In October 2023, Daiichi Sankyo entered into a global development and commercialization collaboration agreement with Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA for ifinatamab deruxtecan (I-DXd).

Graphics and copyediting support was provided by BOLDSCIENCE®, Inc., and was funded by Daiichi Sankyo, Inc., and Merck Sharp & Dohme LLC, a subsidiary of Merck & Co., Inc., Rahway, NJ, USA.

REFERENCES

1. Yamato M, et al. *Mol Cancer Ther*. 2022;21:635–646.
2. Katsumata L, et al. *Cancer Res*. 2023;83:(7 Suppl.):4891–4891.
3. Patel MR, et al. *Ann Oncol*. 2023;34(Suppl. 2):S481.
4. FDA Industry Systems. NDA/BLA multi-disciplinary review and evaluation: Enhertu (fam-trastuzumab deruxtecan-nxki). BLA 761139. https://www.accessdata.fda.gov/drugsatfda_docs/nda/2019/761139Orig1s000MultidisciplineR.pdf. Accessed August 16, 2024.

ABBREVIATIONS

2L, second-line; ACCP, American College of Clinical Pharmacology; ADC, antibody–drug conjugate; B7-H3, B7 homolog 3; CI, confidence interval; C_{max}, maximum plasma concentration; C-QTc, concentration QTc; DXd, deruxtecan; ECG, electrocardiogram; ESCC, esophageal squamous cell carcinoma; GM, geometric mean; hERG, human ether-a-go-go related gene; I-DXd, ifinatamab deruxtecan; IV, intravenous; mCRPC, metastatic castration-resistant prostate cancer; PK, pharmacokinetic; Q3W, every 3 weeks; QTc, heart rate–corrected QT interval; QTcF, Fridericia's correction; QTcP, population-specific correction; RDE, recommended dose for expansion; RR, the time elapsed between two successive R waves; SD, standard deviation; sqNSCLC, squamous non-small cell lung cancer.

Presented at the ACCP Annual Meeting. September 8–10, 2024; Bethesda, MD, USA.

brittany.tran@daiichisankyo.com