Physiologically Based Pharmacokinetic Modeling of a Novel HER3-Targeted Antibody-Drug Conjugate, Patritumab Deruxtecan, to Assess Pharmacokinetic Drug-Drug Interaction

Poster # 029

Akiko Watanabe[1], Brian Chapron[2], Katherine L. Gill[2], Hikari Araki[1], Li Li[3], Tushar Garimella[3], Malaz Abutarif[3], Masakatsu Kotsuma[1]

[1] Quantitative Clinical Pharmacology Department, Daiichi Sankyo Co., Ltd. Tokyo, Japan; [2] Certara UK Ltd., Sheffield, UK, [3] Quantitative Clinical Pharmacology Department, Daiichi Sankyo, Inc., Basking Ridge, NJ, USA

BACKGROUND

- Patritumab deruxtecan (HER3-DXd) is a novel HER3-targeted antibody-drug conjugate (ADC) composed of a humanized monoclonal antibody specifically targeting HER3, a cleavable tetrapeptide-based linker, and a potent topoisomerase I inhibitor payload (DXd).[1]
- HER3-DXd is under investigation for the treatment of various cancers including non-small cell lung cancer[2].
- DXd is eliminated by hepatic uptake via OATP1B1/1B3, metabolism by CYP3A, biliary excretion via P-gp and BCRP, and urinary excretion[3].
- In a clinical drug-drug interaction (DDI) study between trastuzumab deruxtecan (T-DXd), which has the same linker and payload as HER3-DXd, and ritonavir (a strong CYP3A/OATP1B inhibitor) or itraconazole (a strong CYP3A inhibitor), an approximately 1.2-fold increase in DXd AUC was observed[4].
- The aim of this study is to inform the DDI risk of HER3-DXd by physiologically based pharmacokinetic modeling (PBPK) leveraging the available non-clinical and clinical information.

METHODS

A PBPK model linking intact ADC (T-DXd or HER3-DXd), reported in terms of antibody-conjugated DXd, to unconjugated DXd was developed in Simcyp Population-Based Simulator Version 21. The small molecule model within the Simcyp Simulator was used to develop both antibody-conjugated DXd and unconjugated DXd models. These two models were combined to treat antibody-conjugated DXd as a parent drug leading to the formation of unconjugated DXd as a metabolite. It was assumed that catabolism of each molecule of ADC led to instantaneous release of all of payload. The Sim-Japanese or Sim-Cancer population file was used for the simulation.
In addition, the sensitivity analysis of ADC clearance, reflecting the release of DXd from HER3-DXd, was conducted to determine if the impact of inhibitors on the PK of unconjugated DXd varies depending on the differences in DXd release rate among ADCs sharing the same DXd payload.

Model development

T-DXd
- Clinical PK data (Phase I study[5] and control arms of DDI study[4])

HER3-DXd
- Clinical PK data (Phase I study[2])

DXd
- Nonclinical in vitro & in vivo data
- Intrinsic CLs for each elimination pathway were included
 - Biliary excretion
 - Passive diffusion
 - OATP1Bs
 - P-gp & BCRP
 - Metabolism
 - CYP3A4
 - Urinary excretion

Model validation
- Clinical DDI study of T-DXd
 - T-DXd: 5.4 mg/kg, Q3W
 - Inhibitor
 - Ritonavir (a strong CYP3A/OATP1B inhibitor)
 - Itraconazole (a strong CYP3A inhibitor)

Model application
- DDI prediction of HER3-DXd
 - HER3-DXd: 5.6 mg/kg, Q3W
 - Inhibitor
 - Ritonavir
 - Itraconazole

Figure 1. Model development, validation and application for DDI prediction of HER3-DXd

RESULTS

Model development

The developed model reasonably described the PK profiles of antibody-conjugated and unconjugated DXd after intravenous administration of HER3-DXd.

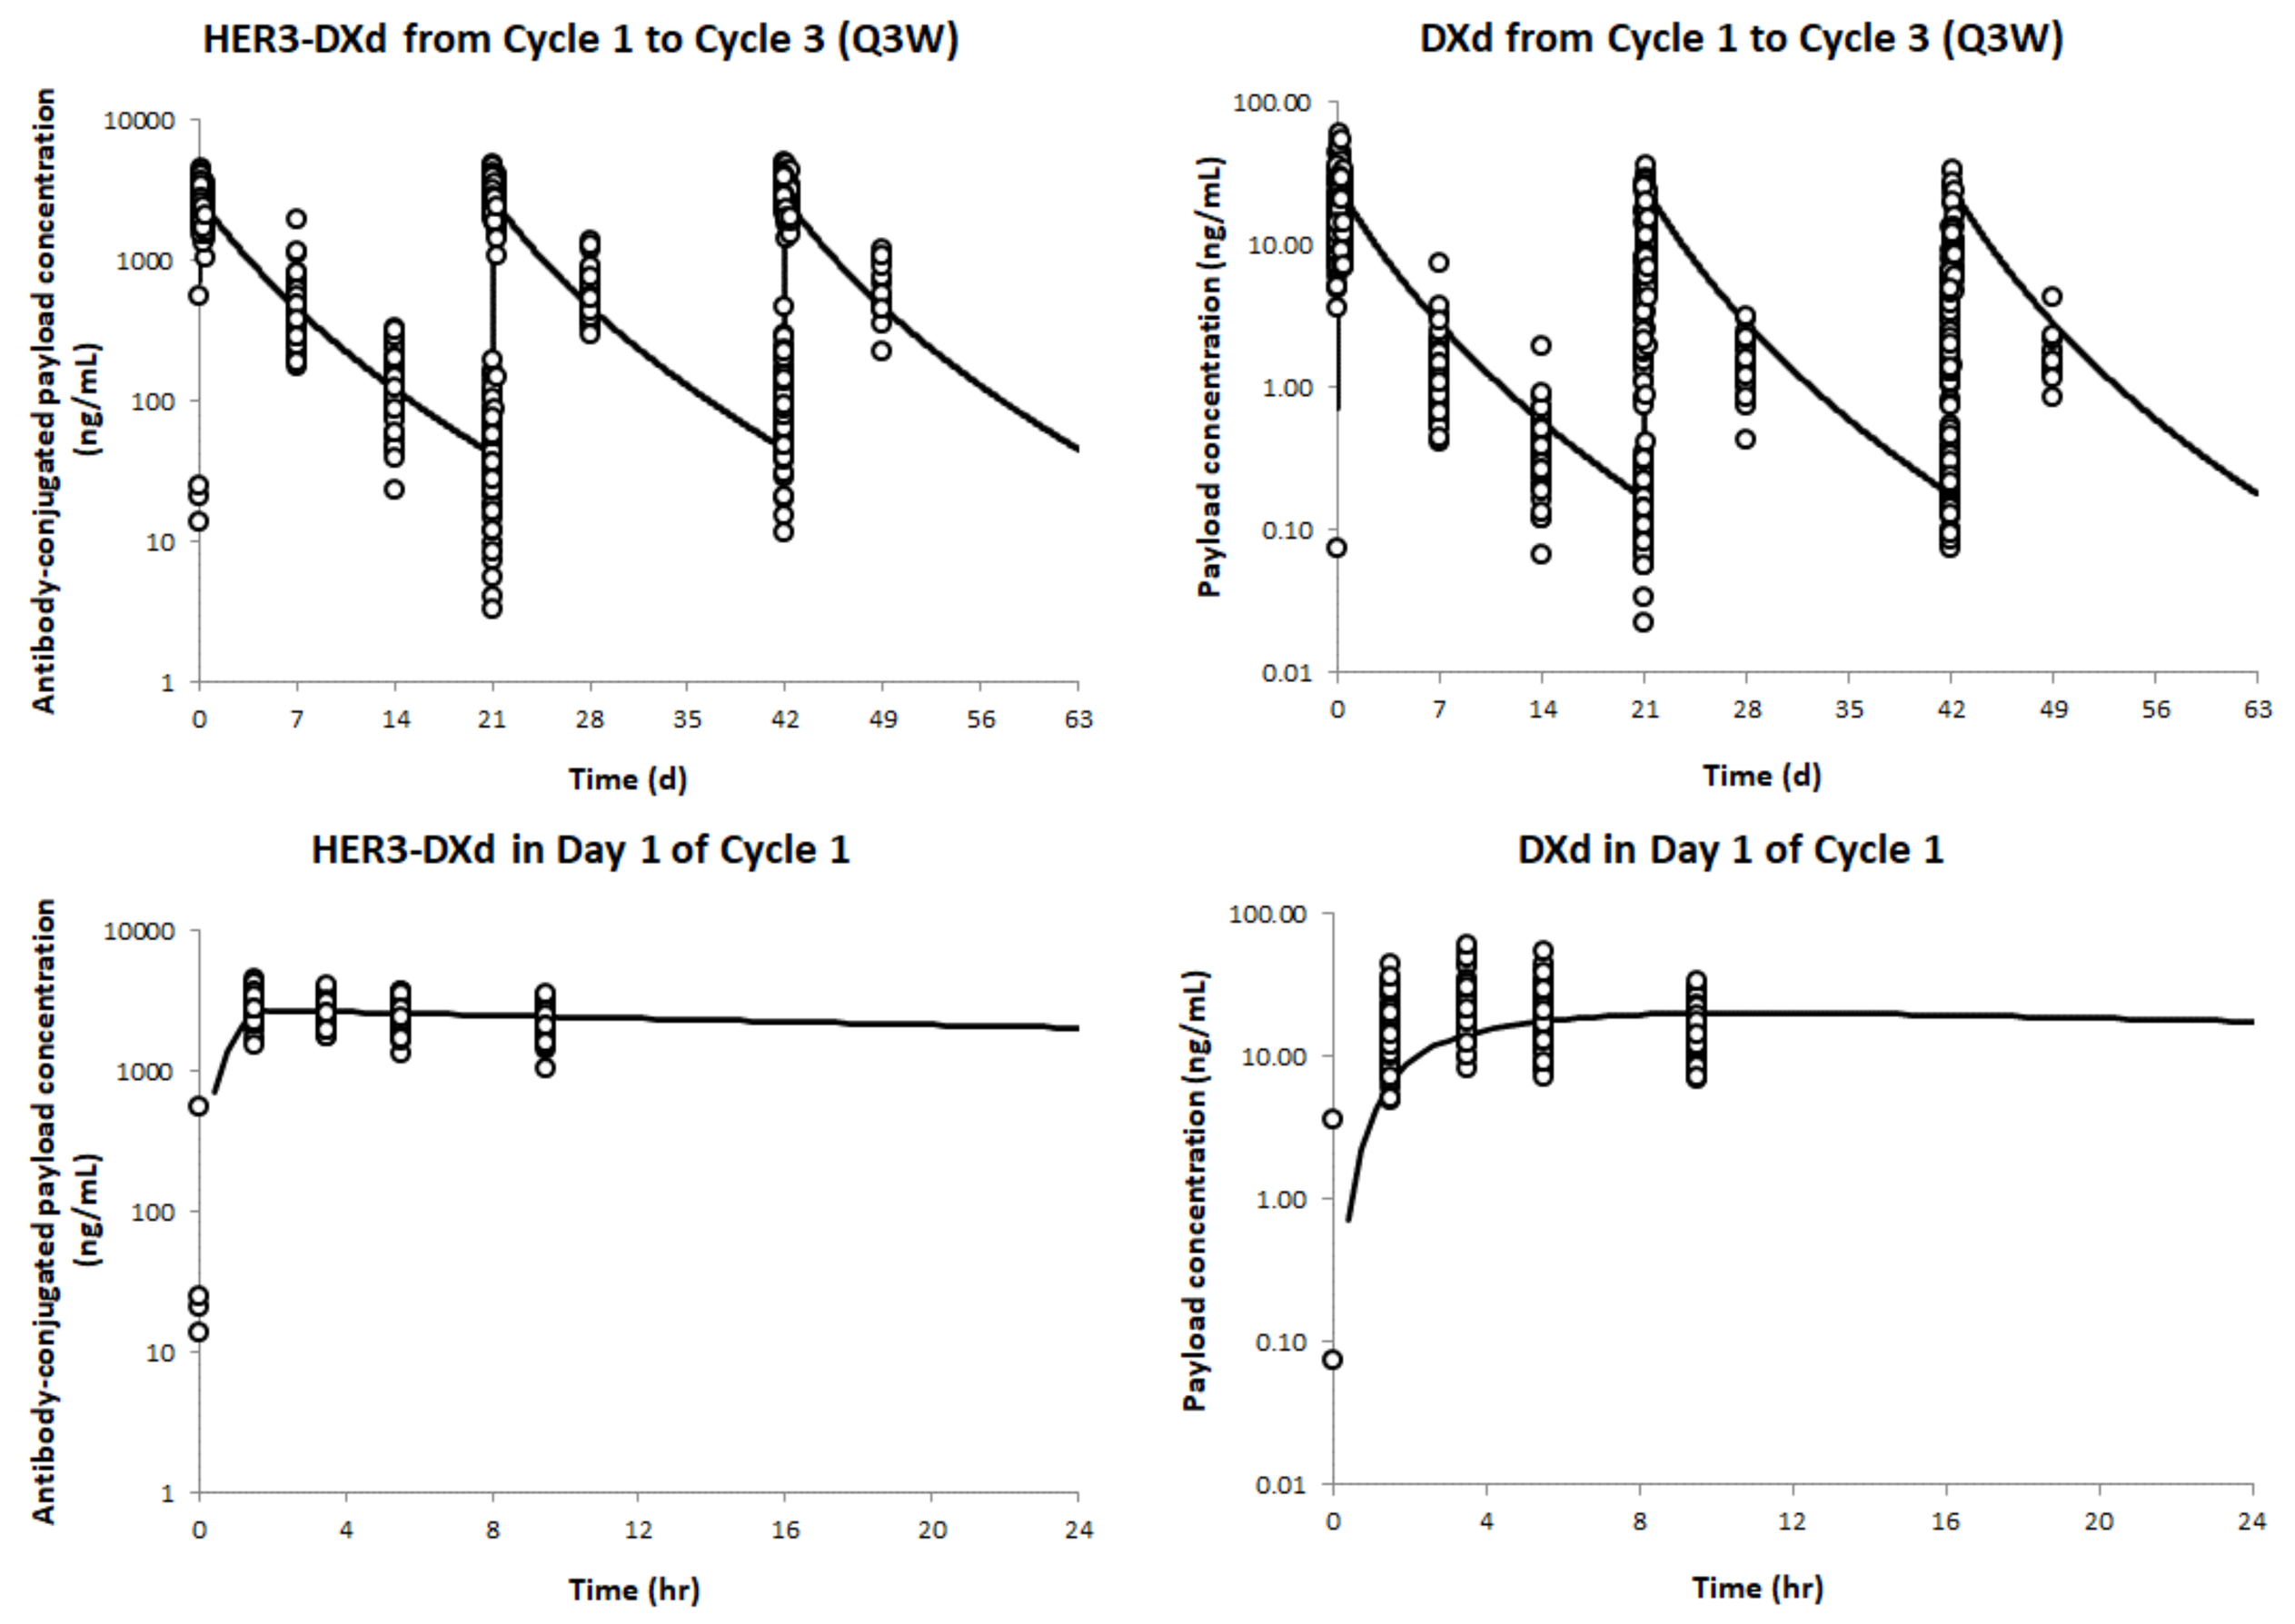
HER3-DXd from Cycle 1 to Cycle 3 (Q3W)
DXd from Cycle 1 to Cycle 3 (Q3W)
HER3-DXd in Day 1 of Cycle 1
DXd in Day 1 of Cycle 1
Antibody-conjugated payload concentration (ng/mL)
Payload concentration (ng/mL)
Time (d)
Time (hr)

Figure 2. Simulated and observed plasma concentrations of antibody-conjugated and unconjugated DXd after IV infusion of 5.6 mg/kg HER3-DXd

HER3-DXd was dosed every 3 weeks (Q3W). The simulated population consisted of 10 trials of 10 cancer patients ages 29 – 80, proportion of females = 0.635. The solid lines represent the simulated mean. The symbols represent the observed clinical data.

Model validation and application

The DXd model linked to the T-DXd model was validated by comparing the observed and simulated Cmax and AUC ratios in the presence or absence of ritonavir or itraconazole[6]. The validated DXd model was linked to HER3-DXd model to simulate the DDI effects after the HER3-DXd dosing.
The simulated DDI effects for DXd when HER3-DXd was co-administered with ritonavir or itraconazole were comparable to those when T-DXd was co-administered with these inhibitors, indicating that the impact of these DXd exposure changes is not clinical meaningful.

Table 1. Cmax and AUC ratios of DXd after 5.4 mg/kg dose of T-DXd (observed and simulated) or 5.6 mg/kg dose of HER3-DXd (simulated) in the presence and absence of ritonavir or itraconazole

With ritonavir			Cmax ratio of DXd (90% CI)	AUC ratio of DXd (90% CI)
T-DXd at 5.4 mg/kg	Observed	Asian	0.99 (0.85-1.14)	1.22 (1.08-1.37)
	Simulated	Japanese	1.24 (1.22-1.26)	1.24 (1.22-1.26)
		Cancer	1.34 (1.32-1.36)	1.34 (1.32-1.36)
HER3-DXd at 5.6 mg/kg	Simulated	Cancer	1.30 (1.28-1.32)	1.33 (1.31-1.35)

With itraconazole			Cmax ratio of DXd (90% CI)	AUC ratio of DXd (90% CI)
T-DXd at 5.4 mg/kg	Observed	Asian	1.04 (0.917-1.18)	1.18 (1.11-1.25)
	Simulated	Japanese	1.20 (1.18-1.21)	1.21 (1.19-1.23)
		Cancer	1.21 (1.19-1.23)	1.22 (1.19-1.24)
HER3-DXd at 5.6 mg/kg	Simulated	Cancer	1.18 (1.16-1.19)	1.19 (1.17-1.21)

Geometric mean (90% CI).
5.4 mg/kg of T-DXd or 5.6 mg/kg of HER3-DXd was dosed Q3W for three cycles. Itraconazole (200mg BD on day 17 of cycle 2 then 200mg QD) or ritonavir (200mg BD from day 17 of cycle 2) was dosed orally. The PK parameters were compared between cycle 2 (control) and cycle 3 (with inhibitor). The simulated populations consisted of 10 trials of 12 Japanese or cancer patients, age 48 – 80, proportion of females = 0.67 for the ritonavir cohort and 10 trials of 14 Japanese or cancer patients, age 31 – 69, proportion of females = 0.286 for the itraconazole cohort based on the design of the clinical DDI study of T-DXd.

Sensitivity analysis of ADC clearance

The sensitivity analysis of the ADC clearance for release of DXd from HER3-DXd showed that 10-fold changes had minimal impact on the predicted DDI (i.e., AUC and Cmax ratios) with ritonavir or itraconazole, although they caused large changes in the PK profiles of both conjugated and unconjugated DXd. This result suggested that the impact of these inhibitors on the DXd PK remained minimal even though the DXd PK was affected by the change in DXd release rate.

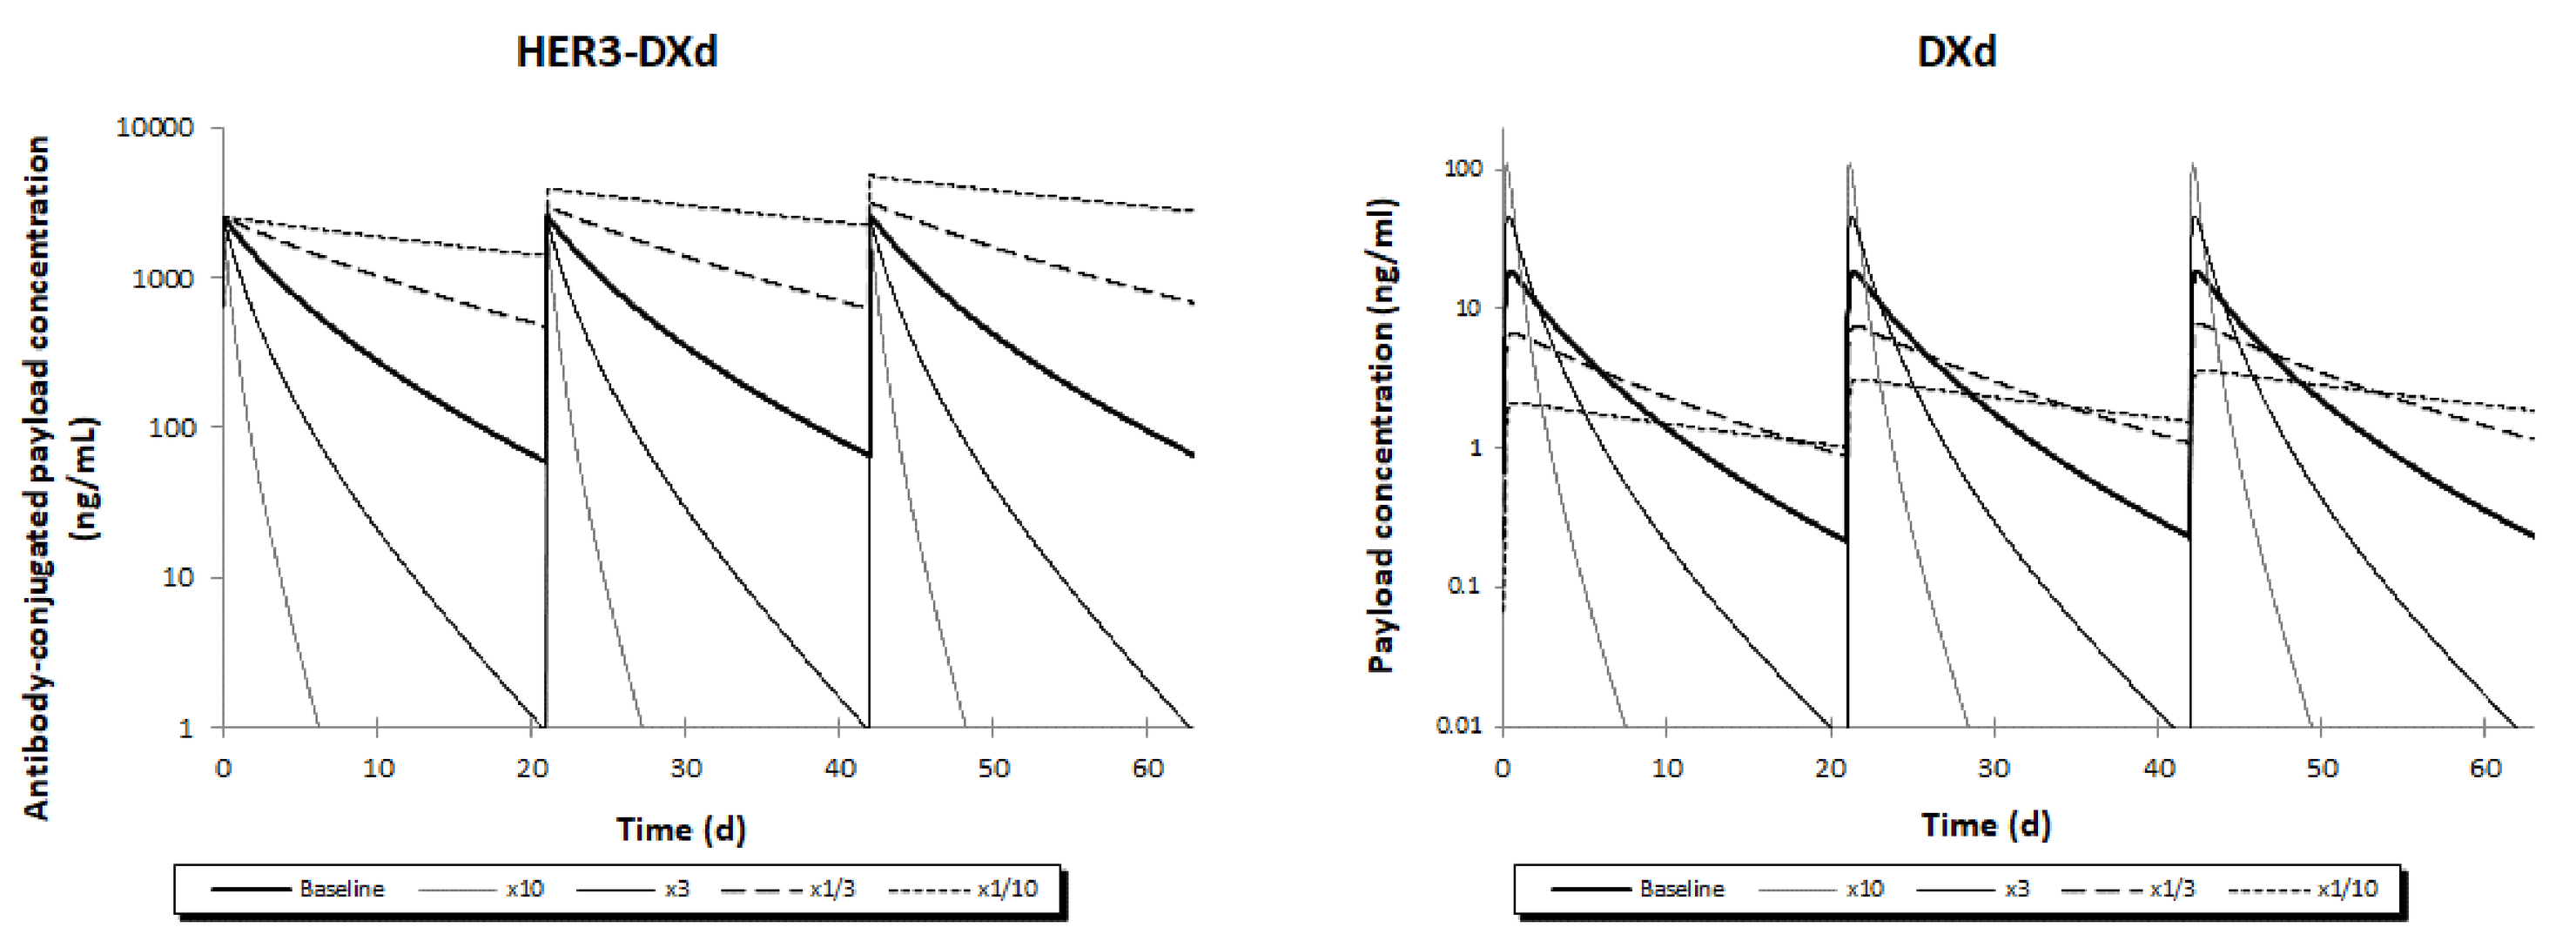
HER3-DXd
DXd
Antibody-conjugated payload concentration (ng/mL)
Payload concentration (ng/ml)
Time (d)
Baseline
x10
x3
x1/3
x1/10

Figure 3. Simulated PK profiles of HER3-DXd and DXd after IV infusion of 5.6 mg/kg HER3-DXd when changing ADC clearance

The ADC clearance was changed 10-, 3-, 1/3- or 1/10-fold from the value in the baseline model.
5.6 mg/kg of HER3-DXd was dosed Q3W for three cycles. The simulated populations consisted of 10 trials of 14 cancer patients, age 31 – 69, proportion of females = 0.286

Table 2. Simulated PK parameters of DXd and DDI effects on DXd with ritonavir or itraconazole after IV infusion of 5.6 mg/kg HER3-DXd across varying ADC clearance values for DXd release

Fold-change in ADC clearance	No inhibitor		DDI with ritonavir		DDI with itraconazole	
	C_{max} (ng/mL)	AUC (ng.day/mL)	C_{max} ratio	AUC ratio	C_{max} ratio	AUC ratio
10-fold decrease	3.30	52.1	1.60	1.58	1.43	1.43
3-fold decrease	7.06	66.7	1.37	1.37	1.23	1.24
Baseline model	16.4	68.4	1.30	1.33	1.18	1.19
3-fold increase	41.7	68.3	1.27	1.33	1.16	1.18
10-fold increase	104	68.2	1.23	1.33	1.14	1.19

The ADC clearance was changed 10-, 3-, 1/3- or 1/10-fold from the value in the baseline model.
5.6 mg/kg of HER3-DXd was dosed Q3W for three cycles. PK parameters were obtained in cycle 3 of HER3-DXd dosing. Ratios were calculated using cycle 3 (day1-17) / cycle 2 (day 1-17). The dosing schedules of inhibitors and the simulated populations for the DDI prediction were the same as shown in Table 1.

CONCLUSION

The HER3-DXd PBPK model predicted strong CYP3A and OATP1B inhibitors would have minimal impact on DXd after HER3-DXd dosing, similar to the case of T-DXd. In addition, the sensitivity analysis of the ADC clearance demonstrated that even if the DXd PK was affected by the change in DXd release rate, the impact of inhibitors on the DXd PK would remain minimal. This finding may be applicable to the DDI prediction in other DXd ADCs with different DXd release rates.
This study demonstrates the utility of ADC PBPK modeling and simulation to predict DDI risk of ADCs sharing the same payload.

REFERENCES

1. Hashimoto et. al., Clin Cancer Res 2019; 25: 7151-7161
2. Jänne et. al., Cancer Discov 2022; 12(1): 74-89
3. Nagai et. al., Xenobiotica 2019; 49: 1086-1096
4. Takahashi et. al., Clin Cancer Res 2021; 27: 5771-5780
5. Doi et. al., Lancet Oncol. 2017; 18: 1512-1522
6. Watanabe et. al., Poster presentation in ACoP12 2021

DISCLOSURES

AW, HA and MK are employees of Daiichi Sankyo Co., Ltd. LL, TG and MA are employees of Daiichi Sankyo, Inc. BC and KG are employees of Certara UK Ltd.

Copies of this poster obtained through this QR code and/or https://bit.ly/DSACCP2024 are for personal use only and may not be reproduced without written permission of the authors.

Presented at the 2024 ACCP Annual Meeting; Sep 8-10, 2024